POS0509

Time to Diagnosis and Associated Comorbidity Burden in Axial Spondyloarthritis in General Practice in France: Results from the THIN® Database

C. Prati[1], A. Constantin[2], E. Dernis[3], S. Le Mouel[4], M. Rozenblat[5], J. Gottenberg[6], A. Vyncke[7], E. Arnee[8], M. Ducros[8], C. Tamberou[8], J. Gandrup Horan[9]

Affiliations: [1]Besancon University Hospital Department of Rheumatology Besancon, France, [2]Purpan Hospital and Paul Sabatier Toulouse III University, Rheumatology, Toulouse, France , [3]Centre Hospitalier du Mans, Rheumatology, Le Mans, France , [4]Les Chênes Medical Practice, Hinx, France , [5]Centre Coralis, Ozoir La Ferriere, France , [6]Hôpitaux Universitaires de Strasbourg, Université de Strasbourg Rheumatology Department Strasbourg, France, [7]UCB Biopharma, Colombes, France, [8]GERS DATA, Patient Data France, Boulogne-Billancourt France, GERS, [9]UCB Pharma, Slough, UK.

Objectives

- To describe the patient characteristics and comorbidity burden at the time of earliest recorded back pain in general practice and at the time of axial spondyloarthritis (axSpA) diagnosis
- To estimate the time to diagnosis of axSpA from earliest recorded back pain in general practice.

Background

- Delayed diagnosis of axSpA remains a challenge for multiple reasons, including lack of awareness in primary care and difficulty in distinguishing chronic inflammatory back pain from other, more prevalent back pain types.
- Longer diagnostic delays are associated with worse clinical, psychological, social and economic outcomes[1].
- Investigating the time to diagnosis and associated comorbidity burden will help inform targets for improvement and raise awareness.

Methods

Study Design

- This retrospective descriptive study utilized data from the French electronic health records THIN® database from a representative sample of 2,000 general practitioners (GPs) between January 2010 and August 2023 (**Figure 1**).

Patients

- Adult patients with axSpA and at least 3 years of continuous enrolment with their GP prior to axSpA diagnosis, with documented back pain in their medical history prior to axSpA diagnosis, were included. Patients diagnosed with rheumatoid arthritis and psoriatic arthritis within one year preceding axSpA diagnosis were excluded.

Study Assessment and Statistical Analyses

- Results were reported descriptively using summary statistics. Patient characteristics and musculoskeletal, extra-musculoskeletal manifestations and comorbidities were assessed at 1) the time of earliest recorded back pain by the GP and 2) the time of axSpA diagnosis. Time to diagnosis was defined as time between these two time points. Differences between genders for time to diagnosis was tested with a Student's t-test.

Results

- A total of 4,402 patients with axSpA were included, with a mean (standard deviation, SD) number of years with available data before axSpA diagnosis of 10.4 (4.9) years. 43% were male and the mean (SD) age was 40 (14) years at earliest back pain diagnosis and 47 (14) years at axSpA diagnosis.
- The burden of musculoskeletal, extra-musculoskeletal manifestations and comorbidities increased between the time of earliest recorded back pain and the time of axSpA diagnosis. The largest increases were seen for enthesitis, fatigue, depression and anxiety (**Figure 3**).
- The mean (SD) time in years between the earliest back pain diagnosis recorded by the GP and the first axSpA diagnosis was 6.3 (4.5) years, with 38% of patients experiencing a time to diagnosis longer than 7 years. No significant differences in time to diagnosis between men and women were observed (**Figure 2**). Patients had a median (Q1, Q3) of 3 (2, 7) documented back pain episodes recorded in their GP records prior to their axSpA diagnosis (**Figure 4**).

Summary

This analysis evaluated time to diagnosis in axSpA and associated comorbidity burden

Time to diagnosis was 6.3 years on average, and no significant difference was observed between men and women

Musculoskeletal, extra-musculoskeletal manifestations and comorbidities increased in the time between the earliest GP consultation for back pain and axSpA diagnosis

Figure 2 Distribution of diagnostic delay

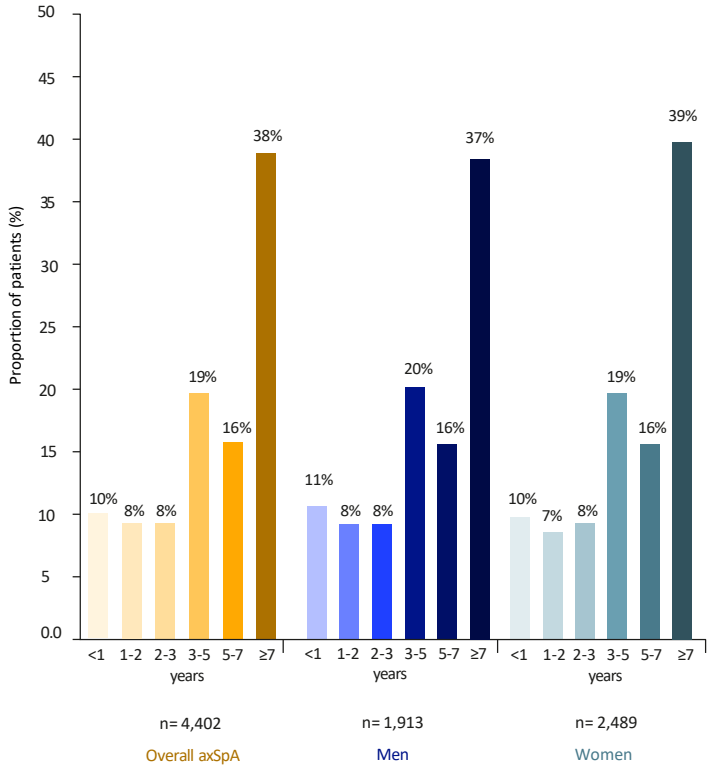
Proportion of patients (%)
10%
8%
8%
19%
16%
38%
11%
8%
8%
20%
16%
37%
10%
7%
8%
19%
16%
39%
<1 1-2 2-3 3-5 5-7 ≥7
years
n= 4,402
Overall axSpA
n= 1,913
Men
n= 2,489
Women

Figure 1 Study design

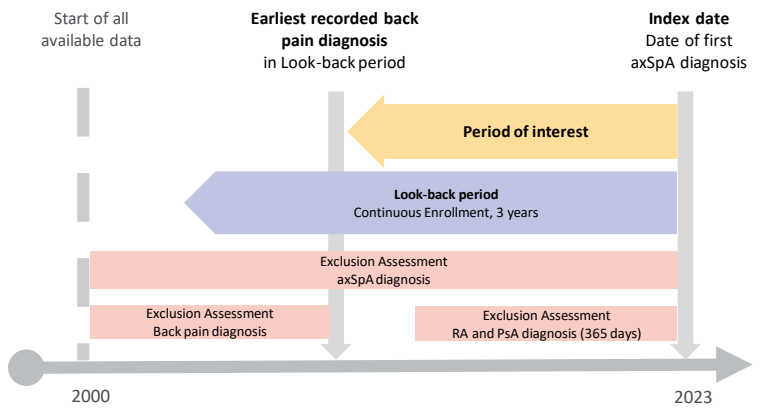
Start of all available data
Earliest recorded back pain diagnosis
in Look-back period
Index date
Date of first axSpA diagnosis
Period of interest
Look-back period
Continuous Enrollment, 3 years
Exclusion Assessment
axSpA diagnosis
Exclusion Assessment
Back pain diagnosis
Exclusion Assessment
RA and PsA diagnosis (365 days)
2000
2023

Figure 4 Distribution of number, location and type of back pain diagnosis between earliest back pain registered and axSpA diagnosis

A) Distribution of location and type of back pain diagnoses

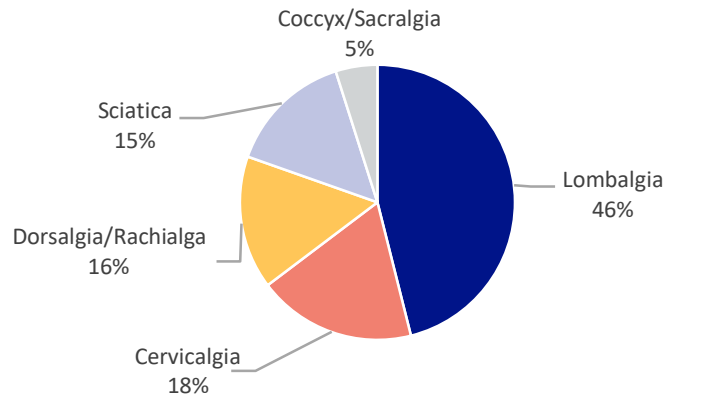
Coccyx/Sacralgia
5%
Sciatica
15%
Lombalgia
46%
Dorsalgia/Rachialga
16%
Cervicalgia
18%

B) Distribution of the number of back pain diagnosis codes registered with the GP

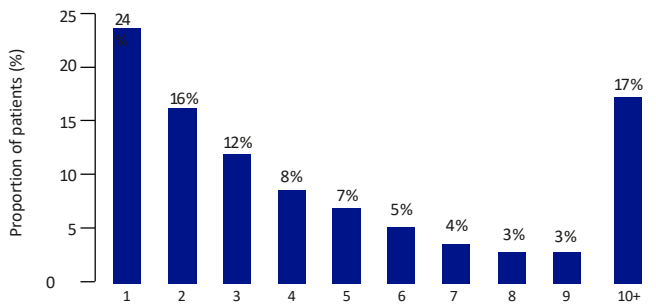
Proportion of patients (%)
24
16%
12%
8%
7%
5%
4%
3%
3%
17%
1 2 3 4 5 6 7 8 9 10+

Figure 3 Distribution of musculoskeletal, extra-musculoskeletal manifestations and comorbidities

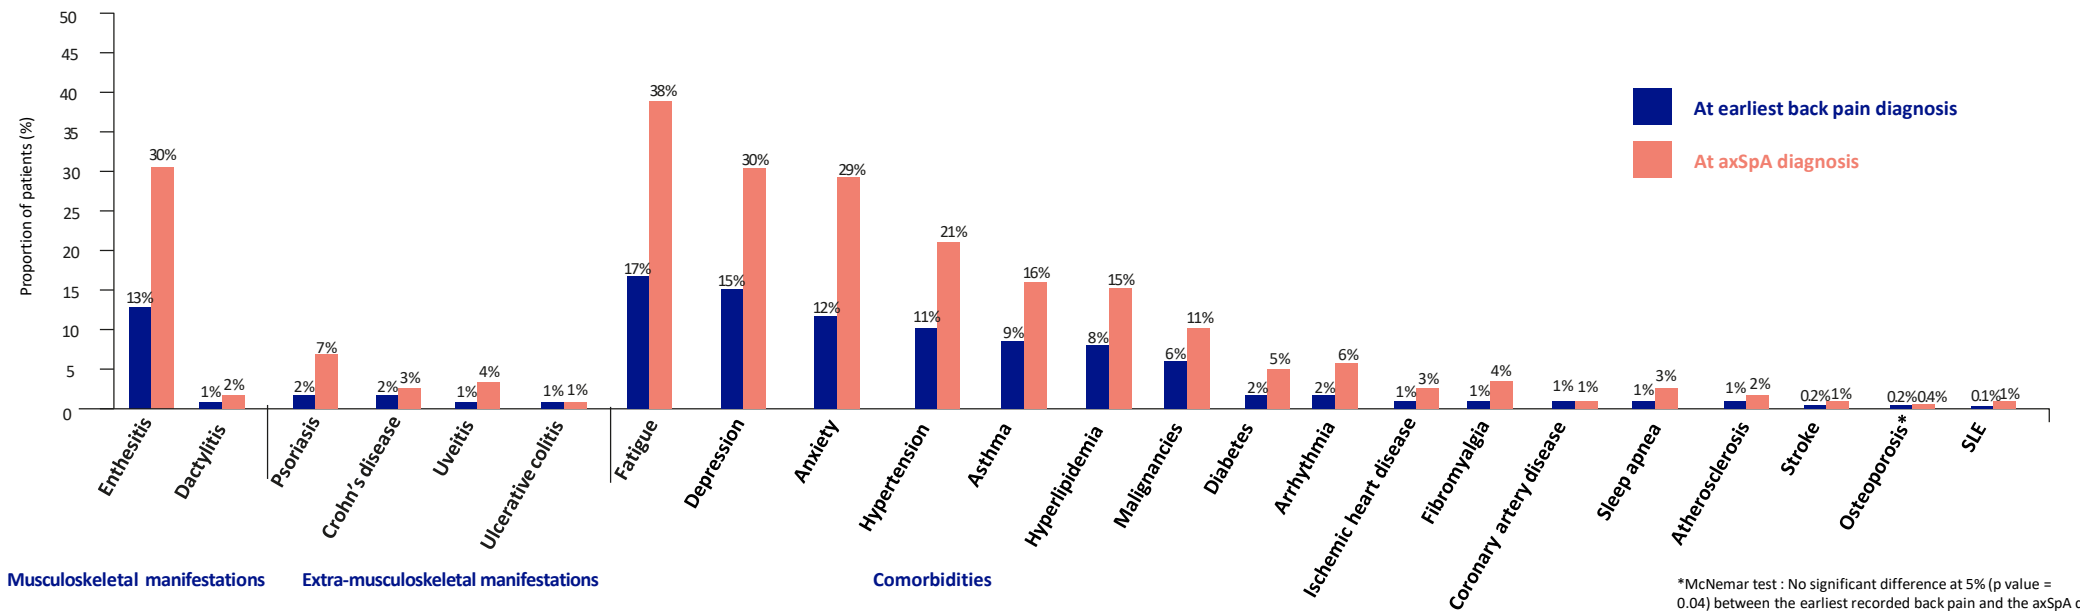
Proportion of patients (%)
At earliest back pain diagnosis
At axSpA diagnosis
Enthesitis 13% 30%
Dactylitis 1% 2%
Psoriasis 2% 7%
Crohn's disease 2% 3%
Uveitis 1% 4%
Ulcerative colitis 1% 1%
Fatigue 17% 38%
Depression 15% 30%
Anxiety 12% 29%
Hypertension 11% 21%
Asthma 9% 16%
Hyperlipidemia 8% 15%
Malignancies 6% 11%
Diabetes 2% 5%
Arrhythmia 2% 6%
Ischemic heart disease 1% 3%
Fibromyalgia 1% 4%
Coronary artery disease 1% 1%
Sleep apnea 1% 3%
Atherosclerosis 1% 2%
Stroke 0.2% 1%
Osteoporosis* 0.2% 0.4%
SLE 0.1% 1%
Musculoskeletal manifestations
Extra-musculoskeletal manifestations
Comorbidities

*McNemar test : No significant difference at 5% (p value = 0.04) between the earliest recorded back pain and the axSpA diagnosis

Conclusions

- **Diagnostic delay remains a key challenge for patients with axSpA in France, with several consultations for back pain in primary care prior to diagnosis and an average time to diagnosis of over 6 years after the earliest back pain consultation.**
- **An increased number of musculoskeletal, non-musculoskeletal manifestations and comorbidities were observed at the time of axSpA diagnosis compared to when back pain was first consulted for, highlighting a worsening disease burden over time.**
- **Interventions are needed to support GPs in recognising chronic back pain patients who may benefit from a timely referral to rheumatology to improve time to diagnosis.**

axSpA: axial spondyloarthritis; SD: standard deviation; GP: general practitioner; PsA: Psoriatic arthritis; RA: Rheumatoid arthritis; SLE: Systemic lupus erythematosus; Q1: upper quartile; Q3: lower quartile

References: [1]Yi E et al. Rheumatol Ther. 2020 Mar;7(1):65-87.

Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **CP, AC, ED, SLM, MR, JG, AV, EA, MD, CT, JGH**; Drafting of the publication, or revising it critically for important intellectual content: **CP, AC, ED, SLM, MR, JG, AV, EA, MD, CT, JGH**; Final approval of the publication: **CP, AC, ED, SLM, MR, JG, AV, EA, MD, CT, JGH**. **Author Disclosures: CP** Speakers bureau for AbbVie, Janssen, Novartis, Sandoz and UCB Pharma, Consultant for Celltrion, Janssen, Lilly, Novartis and Sandoz, Grant/research support from Fresenius and Pfizer, **AC** Speakers bureau for AbbVie, Amgen, Biogen, BMS, Boehringer Ingelheim, Celltrion, Fresenius Kabi, Galapagos, Janssen, Lilly, Medac, MSD, Novartis, Pfizer, Roche, Sanofi, Sandoz, UCB Pharma and Viatris, Consultant for AbbVie, Amgen, Boehringer Ingelheim, Celltrion, Fresenius Kabi, Galapagos, Janssen, Lilly, Novartis, Pfizer, Sanofi and UCB Pharma, Grant/research support from AbbVie, Biogen, BMS, Galapagos, Lilly, Hoffman-La Roche, MSD, Janssen, Novartis, Pfizer, Sanofi and UCB Pharma, **ED** Speakers bureau for Abbvie, BMS, Janssen, Lilly, Medac, MSD, Novartis, Pfizer, Roche and UCB Pharma, Consultant for Janssen, Lilly, Novartis, Pfizer and UCB Pharma, Grant/research support from Lilly and Novartis, **SLM** Paid instructor for Amgen and Sanofi, Consultant for Boiron, Kappa Santé and LEO Pharma, **MR**: None declared, **JG** Speakers bureau for Abbvie, BMS, Galapagos, Gilead, Lilly, MSD, Novartis, Pfizer, Sanofi and UCB Pharma, **CT** As a consultant in a research firm, we are called upon to collaborate with the entire pharmaceutical industry, **EA** Previous employee of Biocodex and Novartis, As a consultant in a research firm, we are called upon to collaborate with the entire pharmaceutical industry, **AV** Employee of UCB Pharma, **MD** As a consultant in a research firm, we are called upon to collaborate with the entire pharmaceutical industry, **JGH** Shareholder of UCB Pharma, Employee of UCB Pharma **Acknowledgements:** This study was sponsored by UCB Pharma. The authors acknowledge Celia Menckeberg, PhD, UCB Pharma, Brussels, Belgium for publication coordination and Chloe Foulds, Costello Medical, London, UK for review management support. All costs associated with development of this poster were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit: https://www.ucbposters.com/?PosterID=EULAR2024_POS0509

Poster ID: POS0509

Link expiration: 29/06/2024

Presented at EULAR 2024 | 12–15 June | Vienna, Austria