0511

Do High Rheumatoid Factor Levels Impact Response to Certolizumab Pegol in Patients with Inadequately Controlled Rheumatoid Arthritis? A Post Hoc Analysis of a Phase 3b Trial

Josef S. Smolen,[1] Ted R. Mikuls,[2] James Galloway,[3] Ulf Müller-Ladner,[4] Jeffrey R. Curtis,[5] Motomu Hashimoto,[6] Tsutomu Takeuchi,[7,8] Ernest Choy,[9] Yoshiya Tanaka,[10] Carlos Cara,[11] Bernard Lauwerys,[12] Nicola Tilt,[13] Baran Ufuktepe,[14] Peter C. Taylor[15]

Objective

To assess the impact of rheumatoid factor (RF) levels and previous therapies on certolizumab pegol (CZP) efficacy in patients with rheumatoid arthritis (RA), in a post hoc analysis of the REALISTIC trial.

Background

- In patients with RA, high RF levels are associated with higher disease activity and decreased response to monoclonal antibodies targeting tumor necrosis factor (TNF).[1,2,3]
- Such patients may have better clinical responses to TNF inhibitors (TNFis) without a fragment crystallizable (Fc) portion, such as CZP, compared to TNFis with an Fc.[4,5,6]
- However, in patients with RA and high RF levels who have had previous inadequate responses or intolerance to TNFis (TNFi-IR), data on response to CZP are limited. These patients generally have poorer responses to subsequent biologic or targeted synthetic disease-modifying anti rheumatic drugs (DMARDs).[7]

Methods

- REALISTIC (NCT00717236) was double-blind and placebo-controlled to Week 12.[8] Patients with inadequately controlled RA were randomized 4:1 to receive either CZP (400 mg subcutaneous [SC] at Weeks 0, 2, and 4, followed by 200 mg SC every 2 weeks) or placebo (PBO) for a 12-week period, after which all patients received open-label CZP.
- We report the following outcomes to Week 36: Disease Activity Score 28 C-reactive protein (DAS28-CRP), DAS28-CRP <2.6, Clinical Disease Activity Index (CDAI), and CDAI remission (CDAI ≤2.8).
 - Results are stratified by baseline RF level (<4th quarter [≤Q3] vs 4th quarter [Q4]) and prior TNFi use; data are reported as observed case (OC).

Results

Patient Characteristics

- Overall, 751 CZP-randomized patients (RF ≤Q3 [<180 IU/mL]: n=560, RF Q4 [≥180 IU/mL]: n=191) and 179 PBO-randomized patients (RF ≤Q3: n=135; RF Q4: n=44) were included. Baseline demographics were generally similar between patients with RF ≤Q3 and Q4, including number of previous DMARDs (**Table 1**).

Clinical Outcomes

- At Week 12, TNFi-naïve patients treated with CZP had lower DAS28-CRP than PBO-randomized patients regardless of RF levels, indicating no effect of RF on response to CZP (**Figure 1**).
 - In TNFi-IR patients DAS28-CRP was similar in CZP-treated patients with RF ≤Q3 and RF Q4; in PBO-randomized patients the difference was larger. There was a greater difference in DAS28-CRP between CZP- and PBO-randomized patients in those with RF Q4 compared with RF ≤Q3.
 - Responses increased to Week 36 in all CZP-randomized groups.
- Similar results were observed for CDAI (**Figure 1**).
- At Week 36, the proportions of CZP-randomized patients who achieved DAS28-CRP <2.6 and CDAI ≤2.8 were similar across RF levels and prior TNFi use (**Figure 2**).

Conclusions

Patients with RA and high RF levels who were treated with certolizumab pegol had similar clinical responses to those with low RF levels regardless of previous TNFi treatment, indicating that RF level does not influence response to certolizumab pegol. These data support previous findings,[5] and expand them to a TNFi-IR population. Results may have treatment choice implications in patients with RA and high RF levels who have had inadequate responses to previous TNFi treatment.

Summary

The **impact of rheumatoid factor level and prior TNFi use** on efficacy outcomes over 36 weeks in patients with **rheumatoid arthritis** who received treatment with **certolizumab pegol** or placebo was assessed

High RF | Low RF

CZP-treated patients with high RF levels experienced **similar clinical responses** to patients with lower RF levels

Clinical response to CZP

TNFi-IR | TNFi-naïve

Similar clinical responses were observed with CZP treatment across RF level stratifications, **irrespective of previous TNFi use**

Clinical response to CZP

These findings indicate that **RF level does not influence response to CZP**, and thus support the use of CZP in patients with RA and high RF levels. Furthermore, these data **expand this notion to those with prior inadequate response to TNFis**

Table 1 Baseline demographics and disease characteristics, stratified by RF level and prior TNFi use

Baseline population characteristic	RF ≤Q3 (<180 IU/mL) (N=695)				RF Q4 (≥180 IU/mL) (N=235)			
	TNFi-IR (N=259)		TNFi-naïve (N=436)		TNFi-IR (N=93)		TNFi-naïve (N=142)	
	PBO (n=52)	CZP (n=207)	PBO (n=83)	CZP (n=353)	PBO (n=16)	CZP (n=77)	PBO (n=28)	CZP (n=114)
Mean age, years (SD)	50.3 (13.6)	54.2 (12.3)	54.7 (13.1)	55.3 (13.4)	57.3 (6.6)	54.9 (11.5)	53.6 (12.7)	57.6 (11.4)
Female, n (%)	45 (86.5)	168 (81.2)	65 (78.3)	282 (79.9)	10 (62.5)	53 (68.8)	21 (75.0)	78 (68.4)
Mean disease duration, years (SD)	9.7 (9.3)	11.8 (9.8)	8.4 (9.8)	6.3 (7.7)	11.8 (7.6)	11.1 (9.0)	5.9 (6.2)	7.0 (7.1)
Disease duration <2 years, n (%)	7 (13.5)	17 (8.2)	29 (34.9)	133 (37.7)	0 (0)	5 (6.5)	9 (32.1)	35 (30.7)
Positive RF (≥14 IU/mL) at baseline, n (%)	43 (82.7)	141 (68.1)	50 (60.2)	223 (63.2)	16 (100)	77 (100)	28 (100)	114 (100)
Mean RF, IU/mL (SD)	58.4 (49.5)	47.3 (46.0)	47.8 (46.9)	45.6 (46.3)	714.4 (1265.3)	602.3 (558.7)	519.9 (399.1)	544.4 (435.8)
Concomitant methotrexate use, n (%)	35 (67.3)	142 (68.6)	55 (66.3)	255 (72.2)	9 (56.3)	46 (59.7)	23 (82.1)	77 (67.5)
Number of previous DMARDs, n (%)								
0	13 (25.0)	59 (28.5)	32 (38.6)	134 (38.0)	7 (43.8)	19 (24.7)	15 (53.6)	39 (34.2)
1	25 (48.1)	81 (39.1)	32 (38.6)	130 (36.8)	4 (25.0)	37 (48.1)	9 (32.1)	40 (35.1)
2	8 (15.4)	26 (12.6)	10 (12.0)	59 (16.7)	2 (12.5)	10 (13.0)	4 (14.3)	22 (19.3)
≥3	6 (11.5)	41 (19.8)	9 (10.8)	30 (8.5)	3 (18.8)	11 (14.3)	0 (0)	13 (11.4)
Mean DAS28-CRP (SD)	5.7 (0.9)	5.7 (0.8)	5.6 (0.8)	5.6 (0.8)	6.3 (1.0)	6.0 (1.1)	5.7 (0.8)	5.9 (1.0)
Mean CDAI (SD)	38.2 (12.5)	38.5 (11.6)	37.2 (11.1)	37.2 (12.2)	44.5 (14.0)	42.4 (14.7)	37.7 (10.2)	39.4 (13.0)
Mean anti-CCP (ACPA), IU/mL (SD)	139.0 (194.0)	111.5 (188.4)	99.5 (171.8)	115.2 (194.6)	260.8 (309.4)	302.7 (285.0)	340.3 (322.7)	268.5 (256.7)

Full analysis set.

Figure 1 Mean DAS28-CRP and CDAI over time in a) overall population, b) TNFi-IR patients, and c) TNFi-naïve patients, stratified by RF level (RF ≤Q3 [<180 IU/mL] or Q4 [≥180 IU/mL]) (OC)

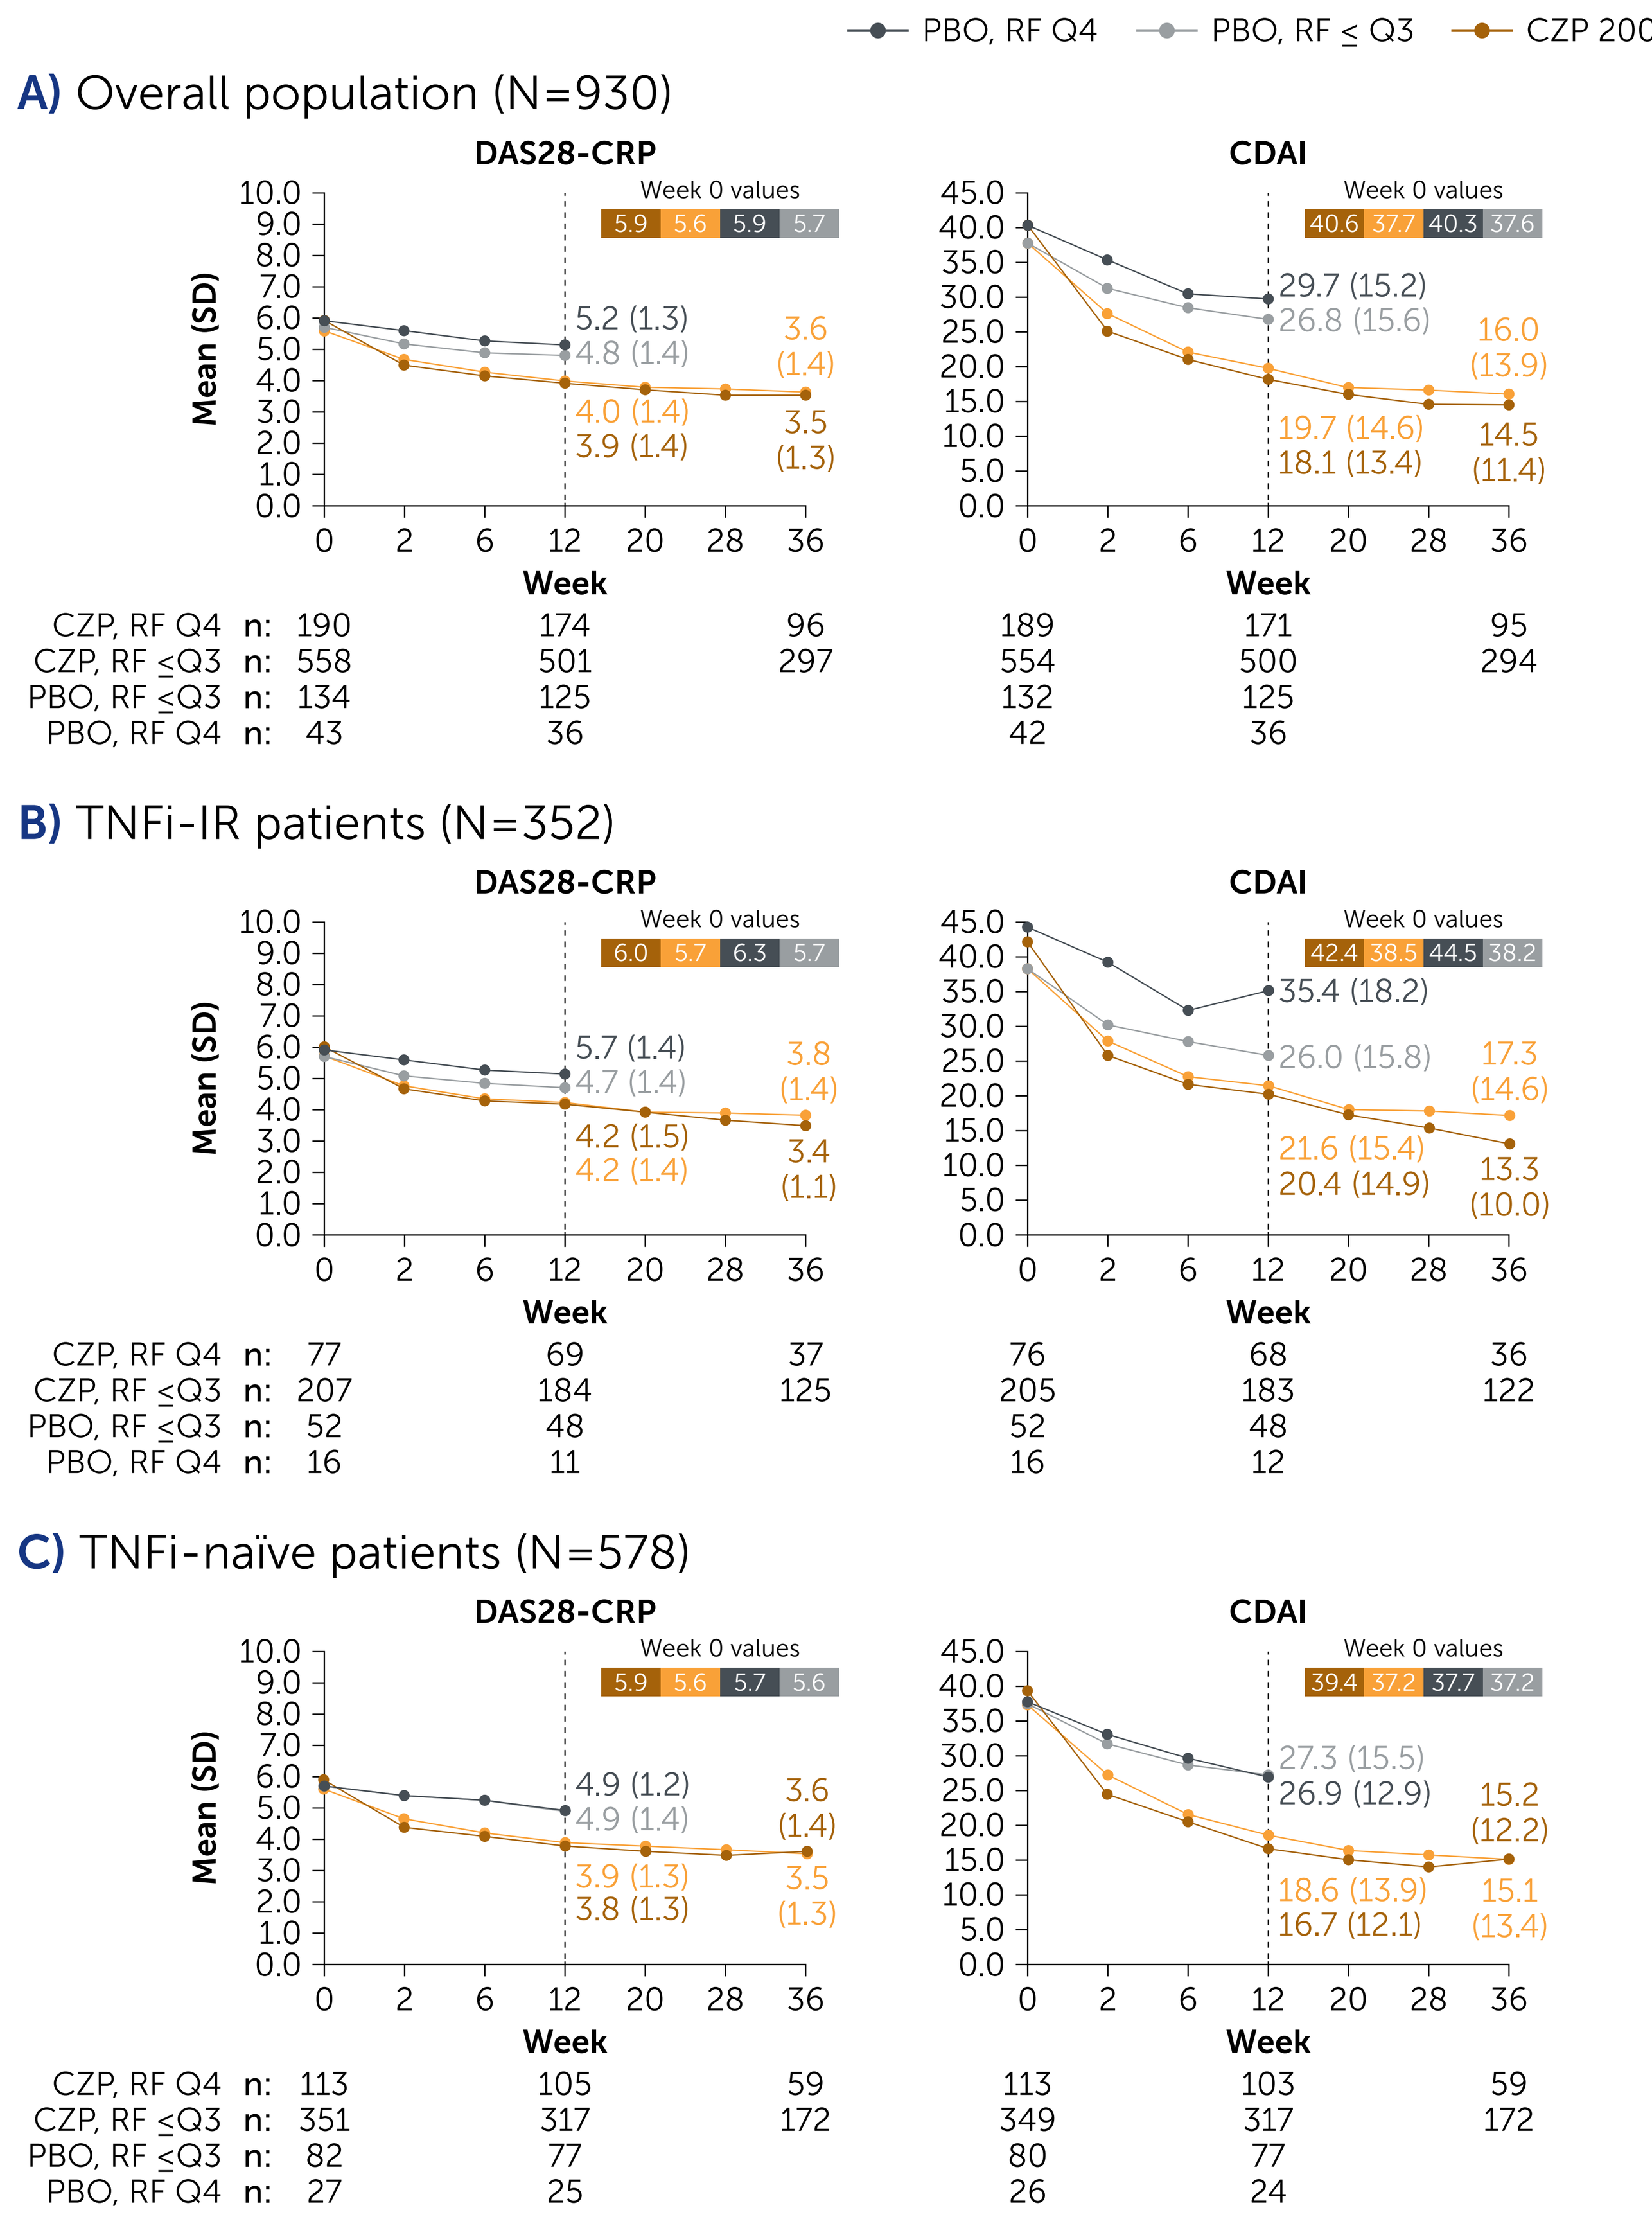

PBO, RF Q4
PBO, RF ≤ Q3
CZP 200 mg Q2W, RF Q4
CZP 200 mg Q2W, RF ≤ Q3
A) Overall population (N=930)
DAS28-CRP
Week 0 values 5.9 5.6 5.9 5.7
5.2 (1.3) 4.8 (1.4) 4.0 (1.4) 3.9 (1.4) 3.6 (1.4) 3.5 (1.3)
Mean (SD)
Week
CZP, RF Q4 n: 190 174 96
CZP, RF ≤Q3 n: 558 501 297
PBO, RF ≤Q3 n: 134 125
PBO, RF Q4 n: 43 36
CDAI
Week 0 values 40.6 37.7 40.3 37.6
29.7 (15.2) 26.8 (15.6) 19.7 (14.6) 18.1 (13.4) 16.0 (13.9) 14.5 (11.4)
189 171 95
554 500 294
132 125
42 36
B) TNFi-IR patients (N=352)
DAS28-CRP
Week 0 values 6.0 5.7 6.3 5.7
5.7 (1.4) 4.7 (1.4) 4.2 (1.5) 4.2 (1.4) 3.8 (1.4) 3.4 (1.1)
CZP, RF Q4 n: 77 69 37
CZP, RF ≤Q3 n: 207 184 125
PBO, RF ≤Q3 n: 52 48
PBO, RF Q4 n: 16 11
CDAI
Week 0 values 42.4 38.5 44.5 38.2
35.4 (18.2) 26.0 (15.8) 21.6 (15.4) 20.4 (14.9) 17.3 (14.6) 13.3 (10.0)
76 68 36
205 183 122
52 48
16 12
C) TNFi-naïve patients (N=578)
DAS28-CRP
Week 0 values 5.9 5.6 5.7 5.6
4.9 (1.2) 4.9 (1.4) 3.9 (1.3) 3.8 (1.3) 3.6 (1.4) 3.5 (1.3)
CZP, RF Q4 n: 113 105 59
CZP, RF ≤Q3 n: 351 317 172
PBO, RF ≤Q3 n: 82 77
PBO, RF Q4 n: 27 25
CDAI
Week 0 values 39.4 37.2 37.7 37.2
27.3 (15.5) 26.9 (12.9) 18.6 (13.9) 16.7 (12.1) 15.2 (12.2) 15.1 (13.4)
113 103 59
349 317 172
80 77
26 24

Full analysis set.

Figure 2 Proportions of patients achieving DAS28-CRP <2.6 and CDAI ≤2.8 in a) overall population, b) TNFi-IR patients, and c) TNFi-naïve patients, stratified by RF level (RF ≤Q3 [<180 IU/mL] or Q4 [≥180 IU/mL]) (OC)

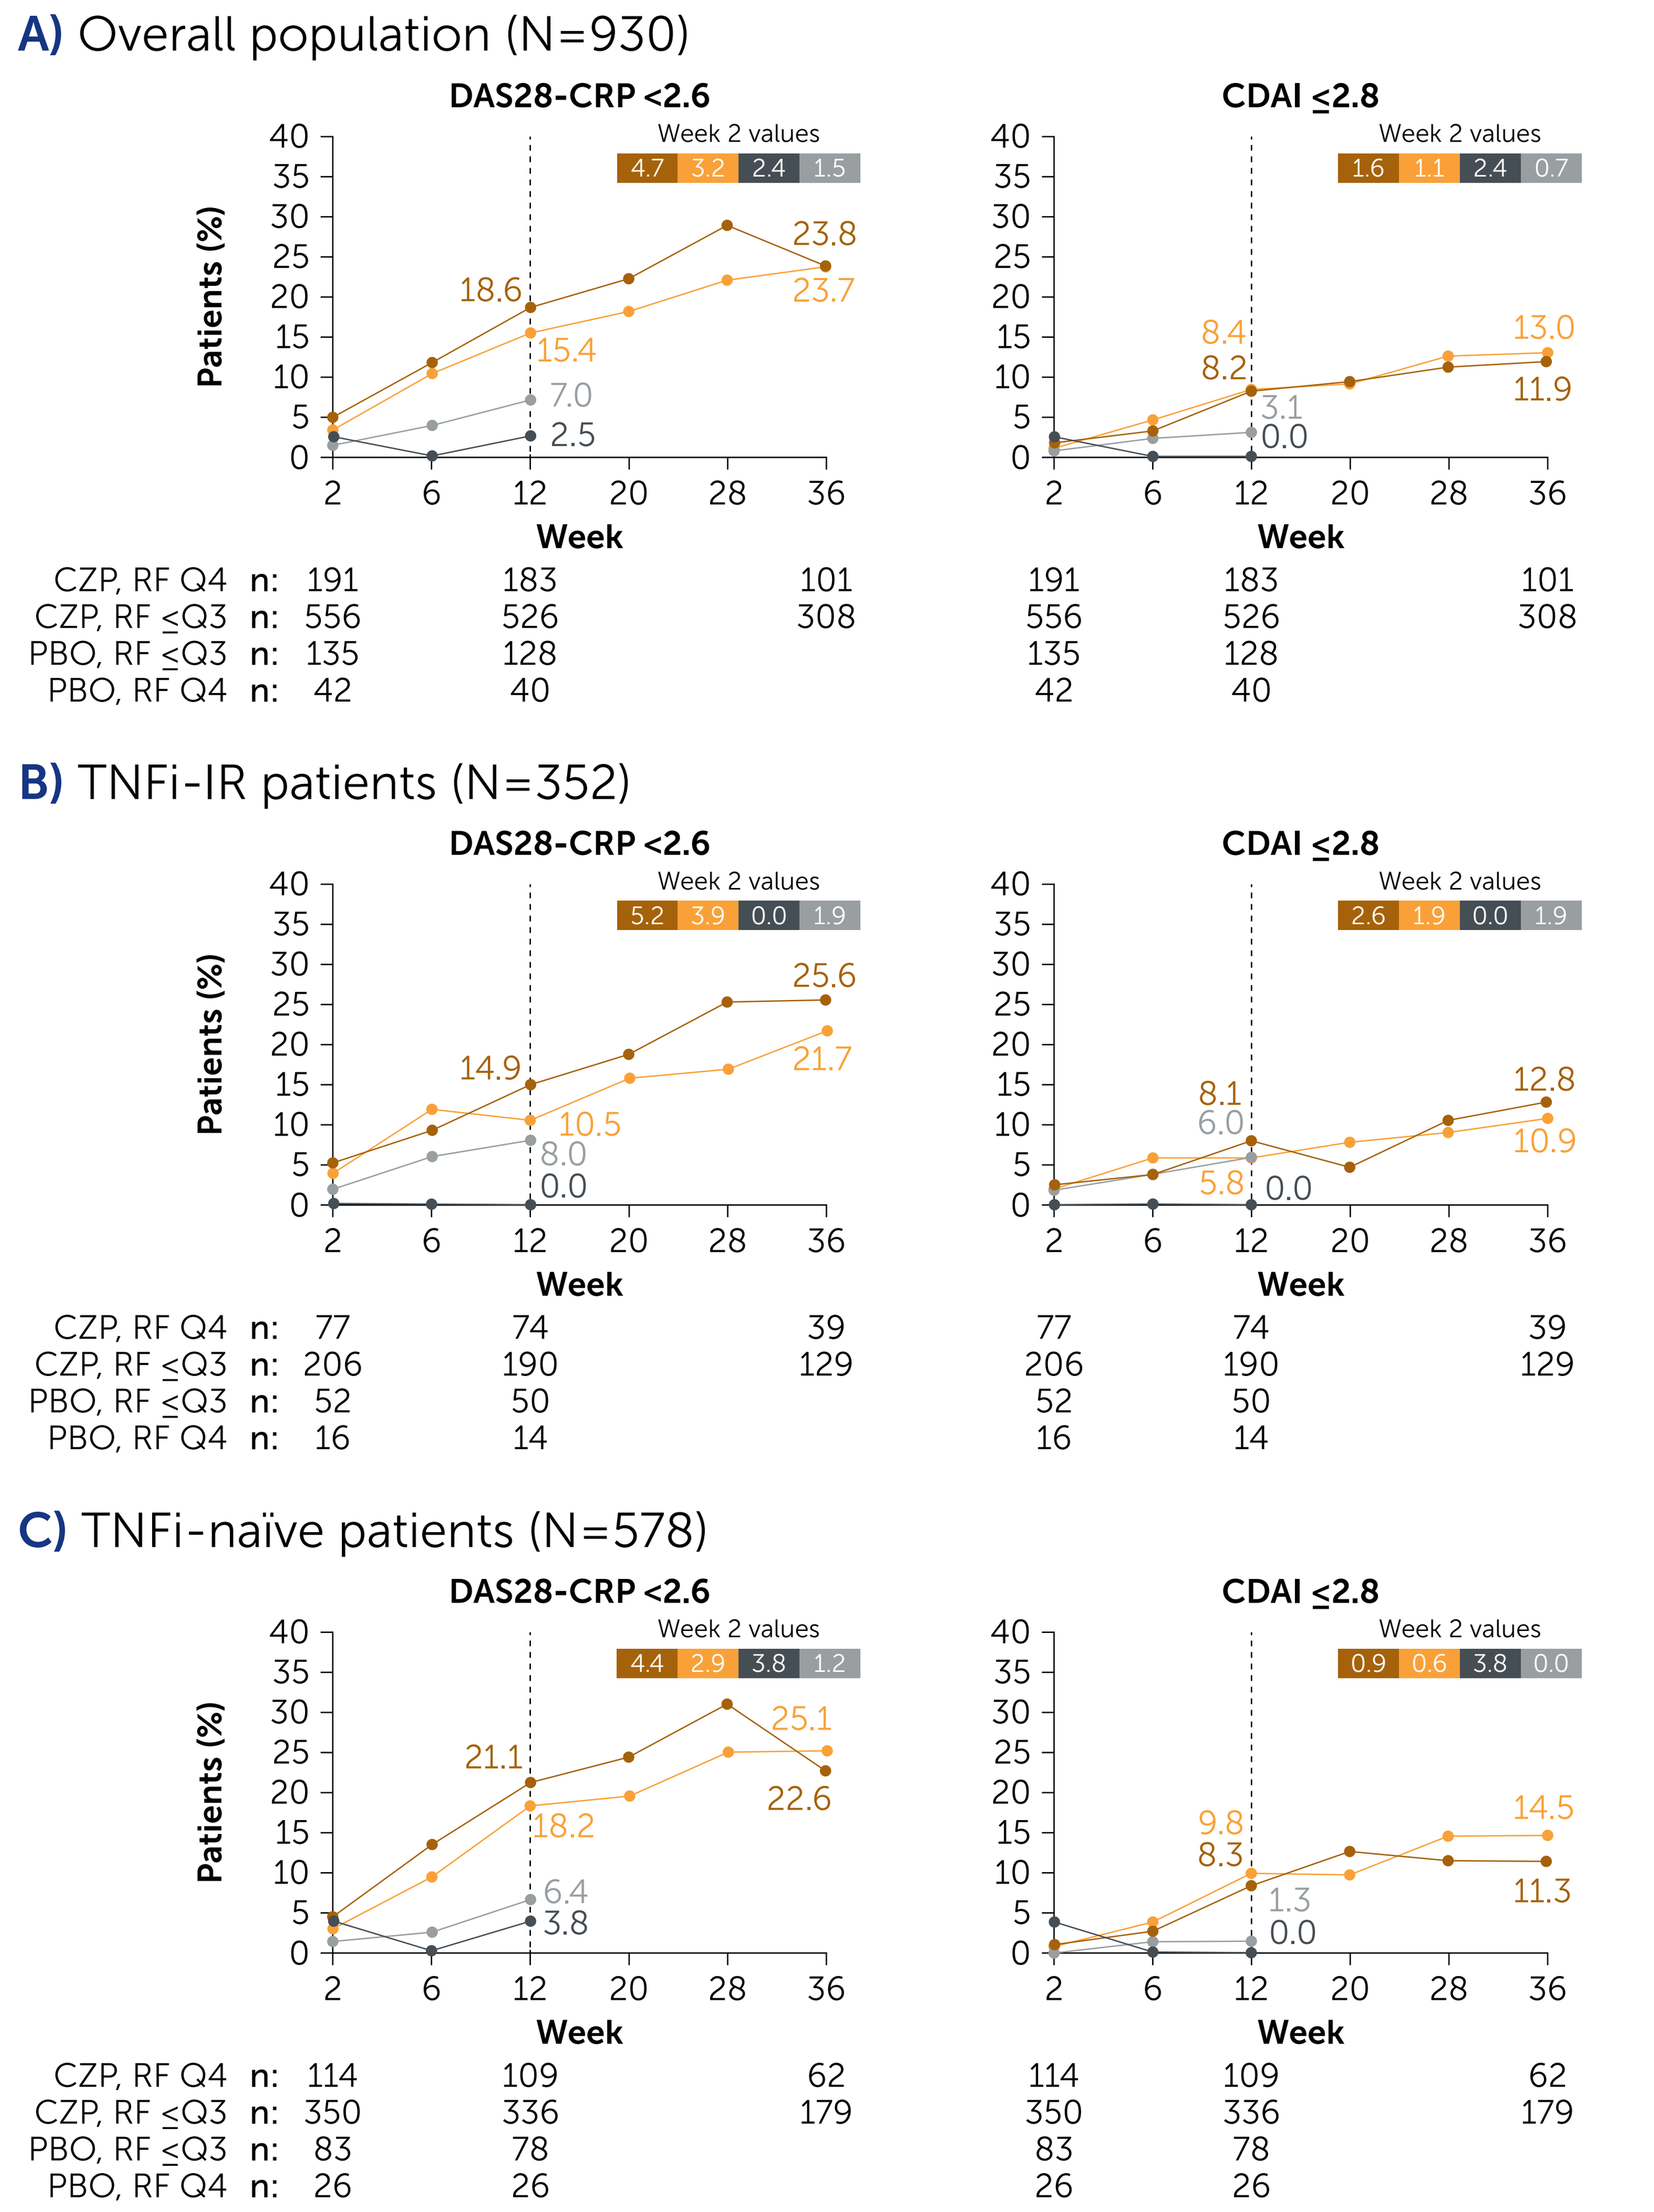

A) Overall population (N=930)
DAS28-CRP <2.6
Week 2 values 4.7 3.2 2.4 1.5
18.6 15.4 7.0 2.5 23.8 23.7
Patients (%)
Week
CZP, RF Q4 n: 191 183 101
CZP, RF ≤Q3 n: 556 526 308
PBO, RF ≤Q3 n: 135 128
PBO, RF Q4 n: 42 40
CDAI ≤2.8
Week 2 values 1.6 1.1 2.4 0.7
8.4 8.2 3.1 0.0 13.0 11.9
191 183 101
556 526 308
135 128
42 40
B) TNFi-IR patients (N=352)
DAS28-CRP <2.6
Week 2 values 5.2 3.9 0.0 1.9
14.9 10.5 8.0 0.0 25.6 21.7
CZP, RF Q4 n: 77 74 39
CZP, RF ≤Q3 n: 206 190 129
PBO, RF ≤Q3 n: 52 50
PBO, RF Q4 n: 16 14
CDAI ≤2.8
Week 2 values 2.6 1.9 0.0 1.9
8.1 6.0 5.8 0.0 12.8 10.9
77 74 39
206 190 129
52 50
16 14
C) TNFi-naïve patients (N=578)
DAS28-CRP <2.6
Week 2 values 4.4 2.9 3.8 1.2
21.1 18.2 6.4 3.8 25.1 22.6
CZP, RF Q4 n: 114 109 62
CZP, RF ≤Q3 n: 350 336 179
PBO, RF ≤Q3 n: 83 78
PBO, RF Q4 n: 26 26
CDAI ≤2.8
Week 2 values 0.9 0.6 3.8 0.0
9.8 8.3 1.3 0.0 14.5 11.3
114 109 62
350 336 179
83 78
26 26

Full analysis set.

ACPA: anti-citrullinated protein antibody; **anti-CCP:** anti-cyclic citrullinated peptide; **CDAI:** clinical disease activity index; **CZP:** certolizumab pegol; **DAS28-CRP:** Disease Activity Score 28 - C-reactive protein; **DMARD:** disease-modifying antirheumatic drug; **Fc:** fragment crystallizable; **IU/mL:** international units per milliliter; **PBO:** placebo; **OC:** observed case; **≤Q3:** <4th quarter; **Q4:** 4th quarter; **RA:** rheumatoid arthritis; **RF:** rheumatoid factor; **SC:** subcutaneous; **SD:** standard deviation; **TNF:** tumor necrosis factor; **TNFi:** tumor necrosis factor inhibitor; **TNFi-IR:** prior inadequate response or intolerance to tumor necrosis factor inhibitors.

Institutions: [1]Medical University of Vienna, Division of Rheumatology, Department of Medicine, Vienna, Austria; [2]University of Nebraska Medical Center, Division of Rheumatology and Immunology, Omaha, NE, USA; [3]King's College London, Centre for Rheumatic Diseases, London, UK; [4]Justus-Liebig University Giessen, Department of Rheumatology and Clinical Immunology, Campus Kerckhoff, Bad Nauheim, Germany; [5]University of Alabama at Birmingham, Division of Clinical Immunology and Rheumatology, Birmingham, AL, USA; [6]Osaka Metropolitan University, Department of Clinical Immunology, Osaka, Japan; [7]Keio University School of Medicine, Division of Rheumatology, Department of Internal Medicine, Tokyo, Japan; [8]Saitama Medical University, Department of Rheumatology and Allied Immunology, Saitama, Japan; [9]Cardiff University School of Medicine, Division of Infection and Immunity, Cardiff, United Kingdom; [10]University of Occupational and Environmental Health, The First Department of Internal Medicine, Kitakyushu, Fukuoka, Japan; [11]UCB, Madrid, Spain; [12]UCB, Brussels, Belgium; [13]UCB, Slough, UK; [14]UCB, Istanbul, Turkey; [15]Botnar Research Centre, Nuffield Department of Orthopaedics, Rheumatology and Musculoskeletal Sciences, University of Oxford, Oxford, UK.

References: [1]Vastesaeger N. Rheum 2009;48:1114–21; [2]Cuchacovich M. Clin Rheumatol 2014;33:1707–14; [3]Takeuchi T. Arthritis Res 2017;19:194; [4]Nakayama Y. Rheumatol Int 2022;42:1227–34; [5]Smolen J.S. Rheum 2024;00:1–10; [6]Bidgood S. Ann Rheum Dis 2024;83 (suppl 1):727; [7]Pappas D.A. Rheumatol Int 2021;41:585–93; [8]Weinblatt M.E. Rheum 2012;51:2204–14.

Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **JSS**, **TRM**, **JG**, **UM-L**, **JRC**, **MH**, **TT**, **EC**, **YT**, **CC**, **BL**, **NT**, **BU**, **PCT**; Drafting of the publication, or reviewing it critically for important intellectual content: **JSS**, **TRM**, **JG**, **UM-L**, **JRC**, **MH**, **TT**, **EC**, **YT**, **CC**, **BL**, **NT**, **BU**, **PCT**; Final approval of the publication: **JSS**, **TRM**, **JG**, **UM-L**, **JRC**, **MH**, **TT**, **EC**, **YT**, **CC**, **BL**, **NT**, **BU**, **PCT**. **Author Disclosures: JSS**: Research grants from: Abbvie, AstraZeneca, Eli Lilly, Novartis, and Roche; honoraria from: AbbVie, Amgen, AstraZeneca, Astro, BMS, Celgene, Celltrion, Chugai, Eli Lilly, Gilead, ILTOO, Janssen, Merck Sharp & Dohme, Novartis, Sandoz, Pfizer, R-Pharma, Roche, Samsung, Sanofi, and UCB; editor of: Annals of the Rheumatic Diseases; co-editor of: Rheumatology 7E/8E; convenor of: EULAR Task Forces and T2T Task Forces. **TRM**: Consultant for: Horizon Therapeutics; Olatech Therapeutics, Pfizer, Sanofi, and UCB; research support from: Horizon Therapeutics; royalties from: Wolters Kluwer Health (UpToDate), and Elsevier. **JG:** Grant/research support/speaker fees from: AbbVie, Galapagos, Janssen, Eli Lilly, Pfizer, and UCB. **UM-L:** Speaker/advisor for UCB. **JRC:** Grant/research support and consultancy fees from AbbVie, Amgen, BMS, Janssen, Lilly, Novartis, Pfizer, and UCB. **MH:** Research grants from: Abbvie, Asahi Kasei, Astellas, BMS, Eisai, Daiichi Sankyo, Eli Lilly, Novartis, and Taisho Toyama; speaker fees from: BMS, Chugai, Eisai, Eli Lilly, Tanabe, and Mitsubishi. **TT:** Grant/honoraria and consultancy fees from: AbbVie, Asahi Kasei, Astellas, AYUMI, BMS, Chugai, Daiichi Sankyo, Eisai, Janssen, Mitsubishi Tanabe, and Pfizer Japan; served on speakers bureaus for: Abbott, BMS, Chugai, Eisai, Janssen, Mitsubishi Tanabe, Pfizer, Takeda, and UCB; consultant for: Asahi Kasei, AstraZeneca, Eli Lilly and Company, Mitsubishi Tanabe, and Novartis; received research support from Abbott, Astellas, BMS, Chugai, Daiichi Sankyo, Eisai, Janssen, Mitsubishi Tanabe, Nippon Shinyaku, Otsuka, Pfizer, Sanofi, Santen, Takeda, and Teijin. **EC:** Research grants from: Bio-Cancer, Biogen, Novartis, Pfizer, and Sanofi; consultancy fees from: Abbvie, Amgen, Biogen, Biocon, Chugai, Eli Lilly, Fresenius Kabi, Gilead, Janssen, Novartis, Pfizer, Regeneron, Roche, RPharm, and Sanofi; speaker fees from AbbVie, Amgen, BMS, Chugai Pharma, Eli Lilly, Fresenius Kai, Galapagos, Gilead, Janssen, Novartis, Pfizer, Regeneron, RPharm, Roche, Sanofi, and UCB. **YT:** Speaker fees and/or honoraria from: AbbVie, AstraZeneca, Boehringer Ingelheim, BMS, Chugai, Eisai, Eli Lilly, Gilead, GlaxoSmithKline Pfizer, Taiho, and UCB; received grants from: Boehringer Ingelheim, Chugai, and Taisho. **CC**, **BL**, **NT:** Employee and stockholder of UCB. **BU:** Employee of UCB. **PCT:** Research grants from: Galapagos; Consultancy fees from: AbbVie, Eli Lilly, UCB, Pfizer, Biogen, Janssen, Fresenius, Galapagos, Gilead, GlaxoSmithKline, Nordic Pharma, and Acelyrin Inc.; participation on a Data Safety Monitoring Board/Advisory Board for Immunovant, Sanofi, and Kymab. **Acknowledgements:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Erin Clarkson, BSc, Costello Medical, Manchester, UK for medical writing and editorial assistance, and the Costello Medical Creative team for graphic design assistance. This study was funded by UCB. All costs associated with development of this presentation were funded by UCB.

To receive a copy of this poster, scan the QR code or visit:
UCBposters.com/ACR2024
Poster ID: 0511
Link expiration:
February 17, 2025

Presented at ACR Convergence 2024 | November 14–19 | Washington, DC