Poster **MG76**

Effect of rozanolixizumab on myasthenia gravis-specific outcome subdomain scores: *Post hoc* analyses from the Phase 3 MycarinG study

MGFA Scientific Session 2024; Savannah, GA, USA; October 15, 2024

Robert M. Pascuzzi[1], Ali A. Habib[2], John Vissing[3], Tuan Vu[4], Asha Hareendran[5], Francesca Pannullo[5], Thaïs Tarancón[6], Vera Bril[7] on behalf of the MycarinG study team

[1]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [2]MDA ALS & Neuromuscular Center, Department of Neurology, University of California, Irvine, Orange, CA, USA; [3]Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [4]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [5]UCB, Slough, UK; [6]UCB, Madrid, Spain; [7]University Health Network, Toronto, ON, Canada

Introduction

- Rozanolixizumab is a humanized IgG4 mAb FcRn inhibitor approved for the treatment of adults with AChR Ab+ or MuSK Ab+ gMG[1,2]
- In the randomized, double-blind, placebo-controlled, Phase 3, MycarinG study (NCT03971422), one 6-week cycle of rozanolixizumab significantly improved MG-specific outcomes versus placebo in patients with gMG (**Figure 1**)[1]
- Treatment for gMG may have differential effects across the different muscle groups affected by the disease[3–5]
 - These differential effects have been evaluated using the subdomain scores of MG-ADL and QMG[5–7]
 - This *post hoc* analysis aimed to evaluate the effect of rozanolixizumab treatment on MG-ADL and QMG subdomain scores assessing ocular, bulbar, respiratory and limb weakness/gross motor muscle groups

Methods

- Patients enrolled in MycarinG were aged ≥18 years with AChR Ab+ or MuSK Ab+ gMG, MGFA Disease Class II–IVa, with an MG-ADL score ≥3 (for non-ocular symptoms) and a QMG score ≥11[1]
- Patients were randomized 1:1:1 to receive weekly subcutaneous rozanolixizumab 7 mg/kg, rozanolixizumab 10 mg/kg, or placebo for 6 weeks, followed by an 8-week observation period[1]
- The primary endpoint was CFB to Day 43 in MG-ADL total score; secondary endpoints included CFB to Day 43 in QMG total score[1]
- MG-ADL and QMG subdomain scores were derived by grouping individual items into muscle groups (**Table 1**)[8,9]
- The CFB to Day 43 in MG-ADL and QMG subdomains was analyzed *post hoc*
 - All *post hoc* analyses were descriptive

Results

- Overall, 200 patients received rozanolixizumab 7 mg/kg (n=66), 10 mg/kg (n=67), or placebo (n=67)
- Baseline demographics and disease characteristics were generally balanced between the treatment groups (**Table 2**)
- Mean CFB at Day 43 across all MG-ADL subdomain scores was higher for the rozanolixizumab treatment groups compared with the placebo group (**Figure 2**)
 - In rozanolixizumab-treated patients, mean CFB was highest in the bulbar subdomain score
- Mean CFB at Day 43 across all QMG subdomain scores was also higher for the rozanolixizumab treatment groups compared with the placebo group (**Figure 3**)
 - In rozanolixizumab-treated patients, mean CFB was highest in the gross motor subdomain score
- In all patients, mean CFB was lowest in the respiratory subdomain score for both MG-ADL and QMG. However, mean respiratory subdomain scores at baseline were low (**Figure 2** and **3**); patients with severe respiratory or oropharyngeal muscle weakness (MGFA Disease Class IVb/V) were excluded from the study
- Rozanolixizumab treatment was generally well tolerated, and most adverse events were mild or moderate (**Table 3**)
 - The most frequent TEAEs in rozanolixizumab-treated patients were headache (41.4%), diarrhea (20.3%), pyrexia (16.5%), and nausea (9.8%)

Figure 1 CFB in MG-ADL and QMG total scores at Day 43

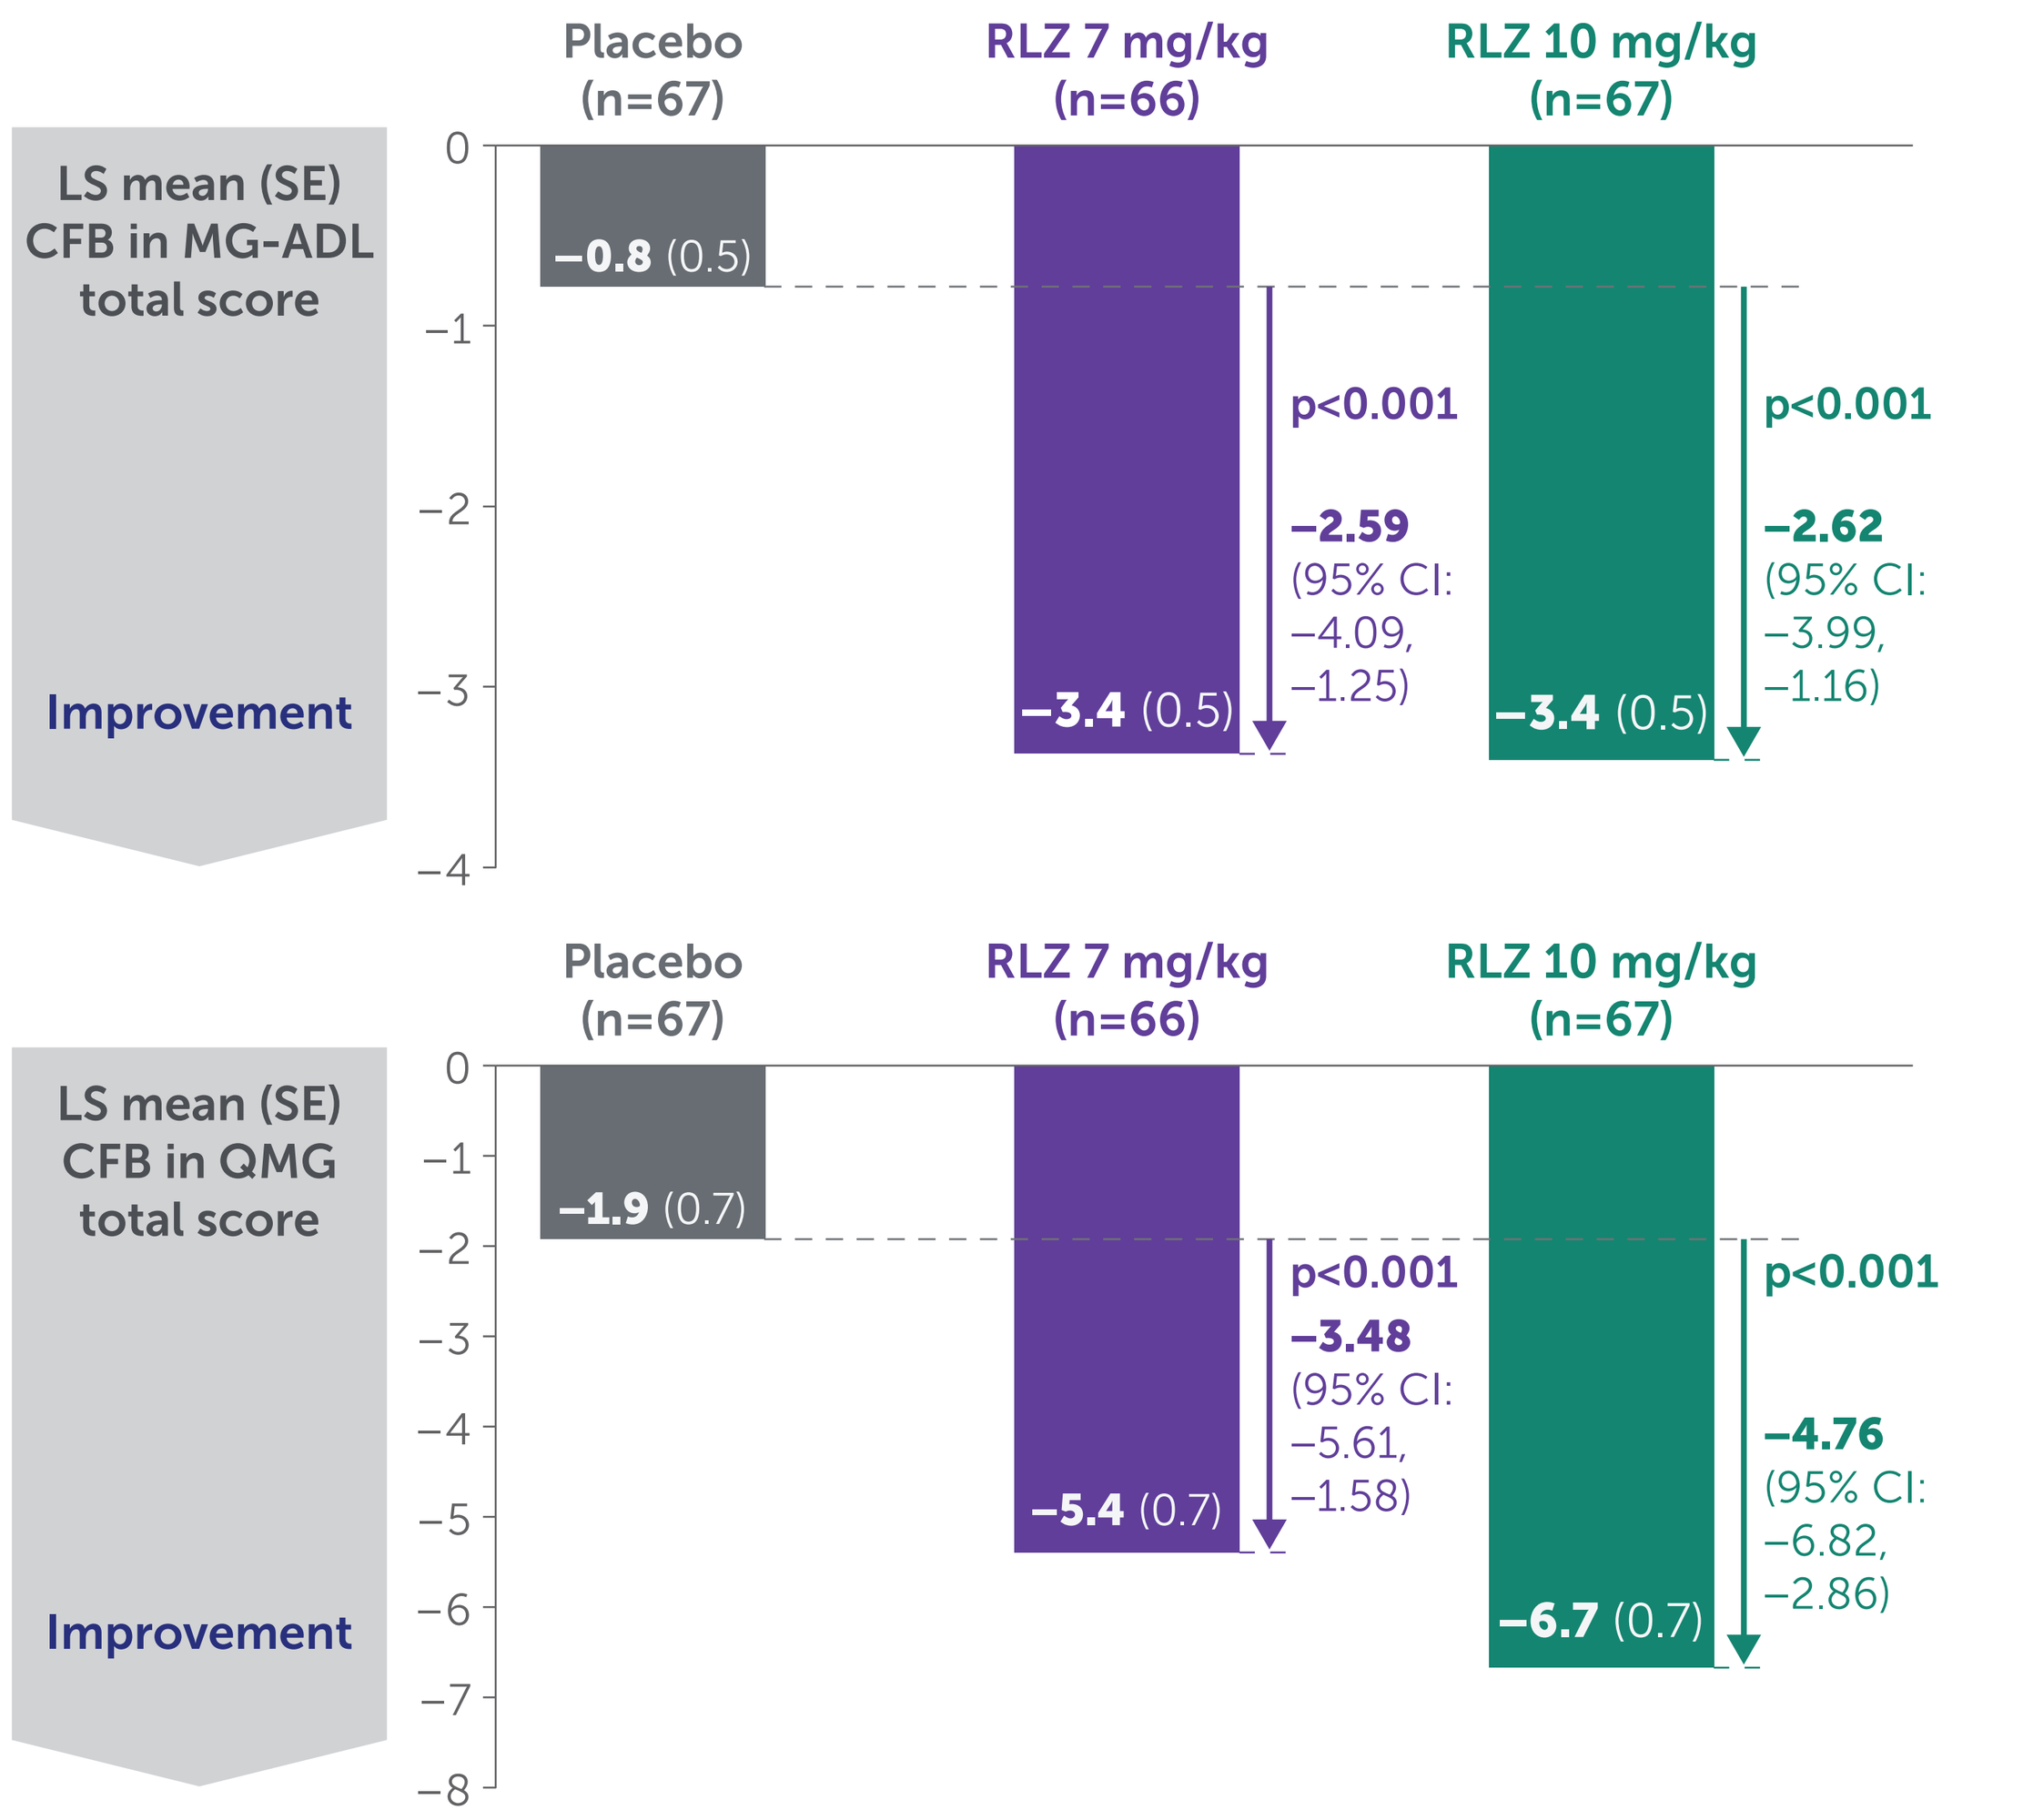
Placebo (n=67)
RLZ 7 mg/kg (n=66)
RLZ 10 mg/kg (n=67)
LS mean (SE) CFB in MG-ADL total score
Improvement
0
−1
−2
−3
−4
−0.8 (0.5)
−3.4 (0.5)
−3.4 (0.5)
p<0.001
−2.59 (95% CI: −4.09, −1.25)
p<0.001
−2.62 (95% CI: −3.99, −1.16)
LS mean (SE) CFB in QMG total score
−5
−6
−7
−8
−1.9 (0.7)
−5.4 (0.7)
−6.7 (0.7)
p<0.001
−3.48 (95% CI: −5.61, −1.58)
p<0.001
−4.76 (95% CI: −6.82, −2.86)

Randomized set.

Figure 2 Mean (SD) CFB in MD-ADL subdomain scores at Day 43

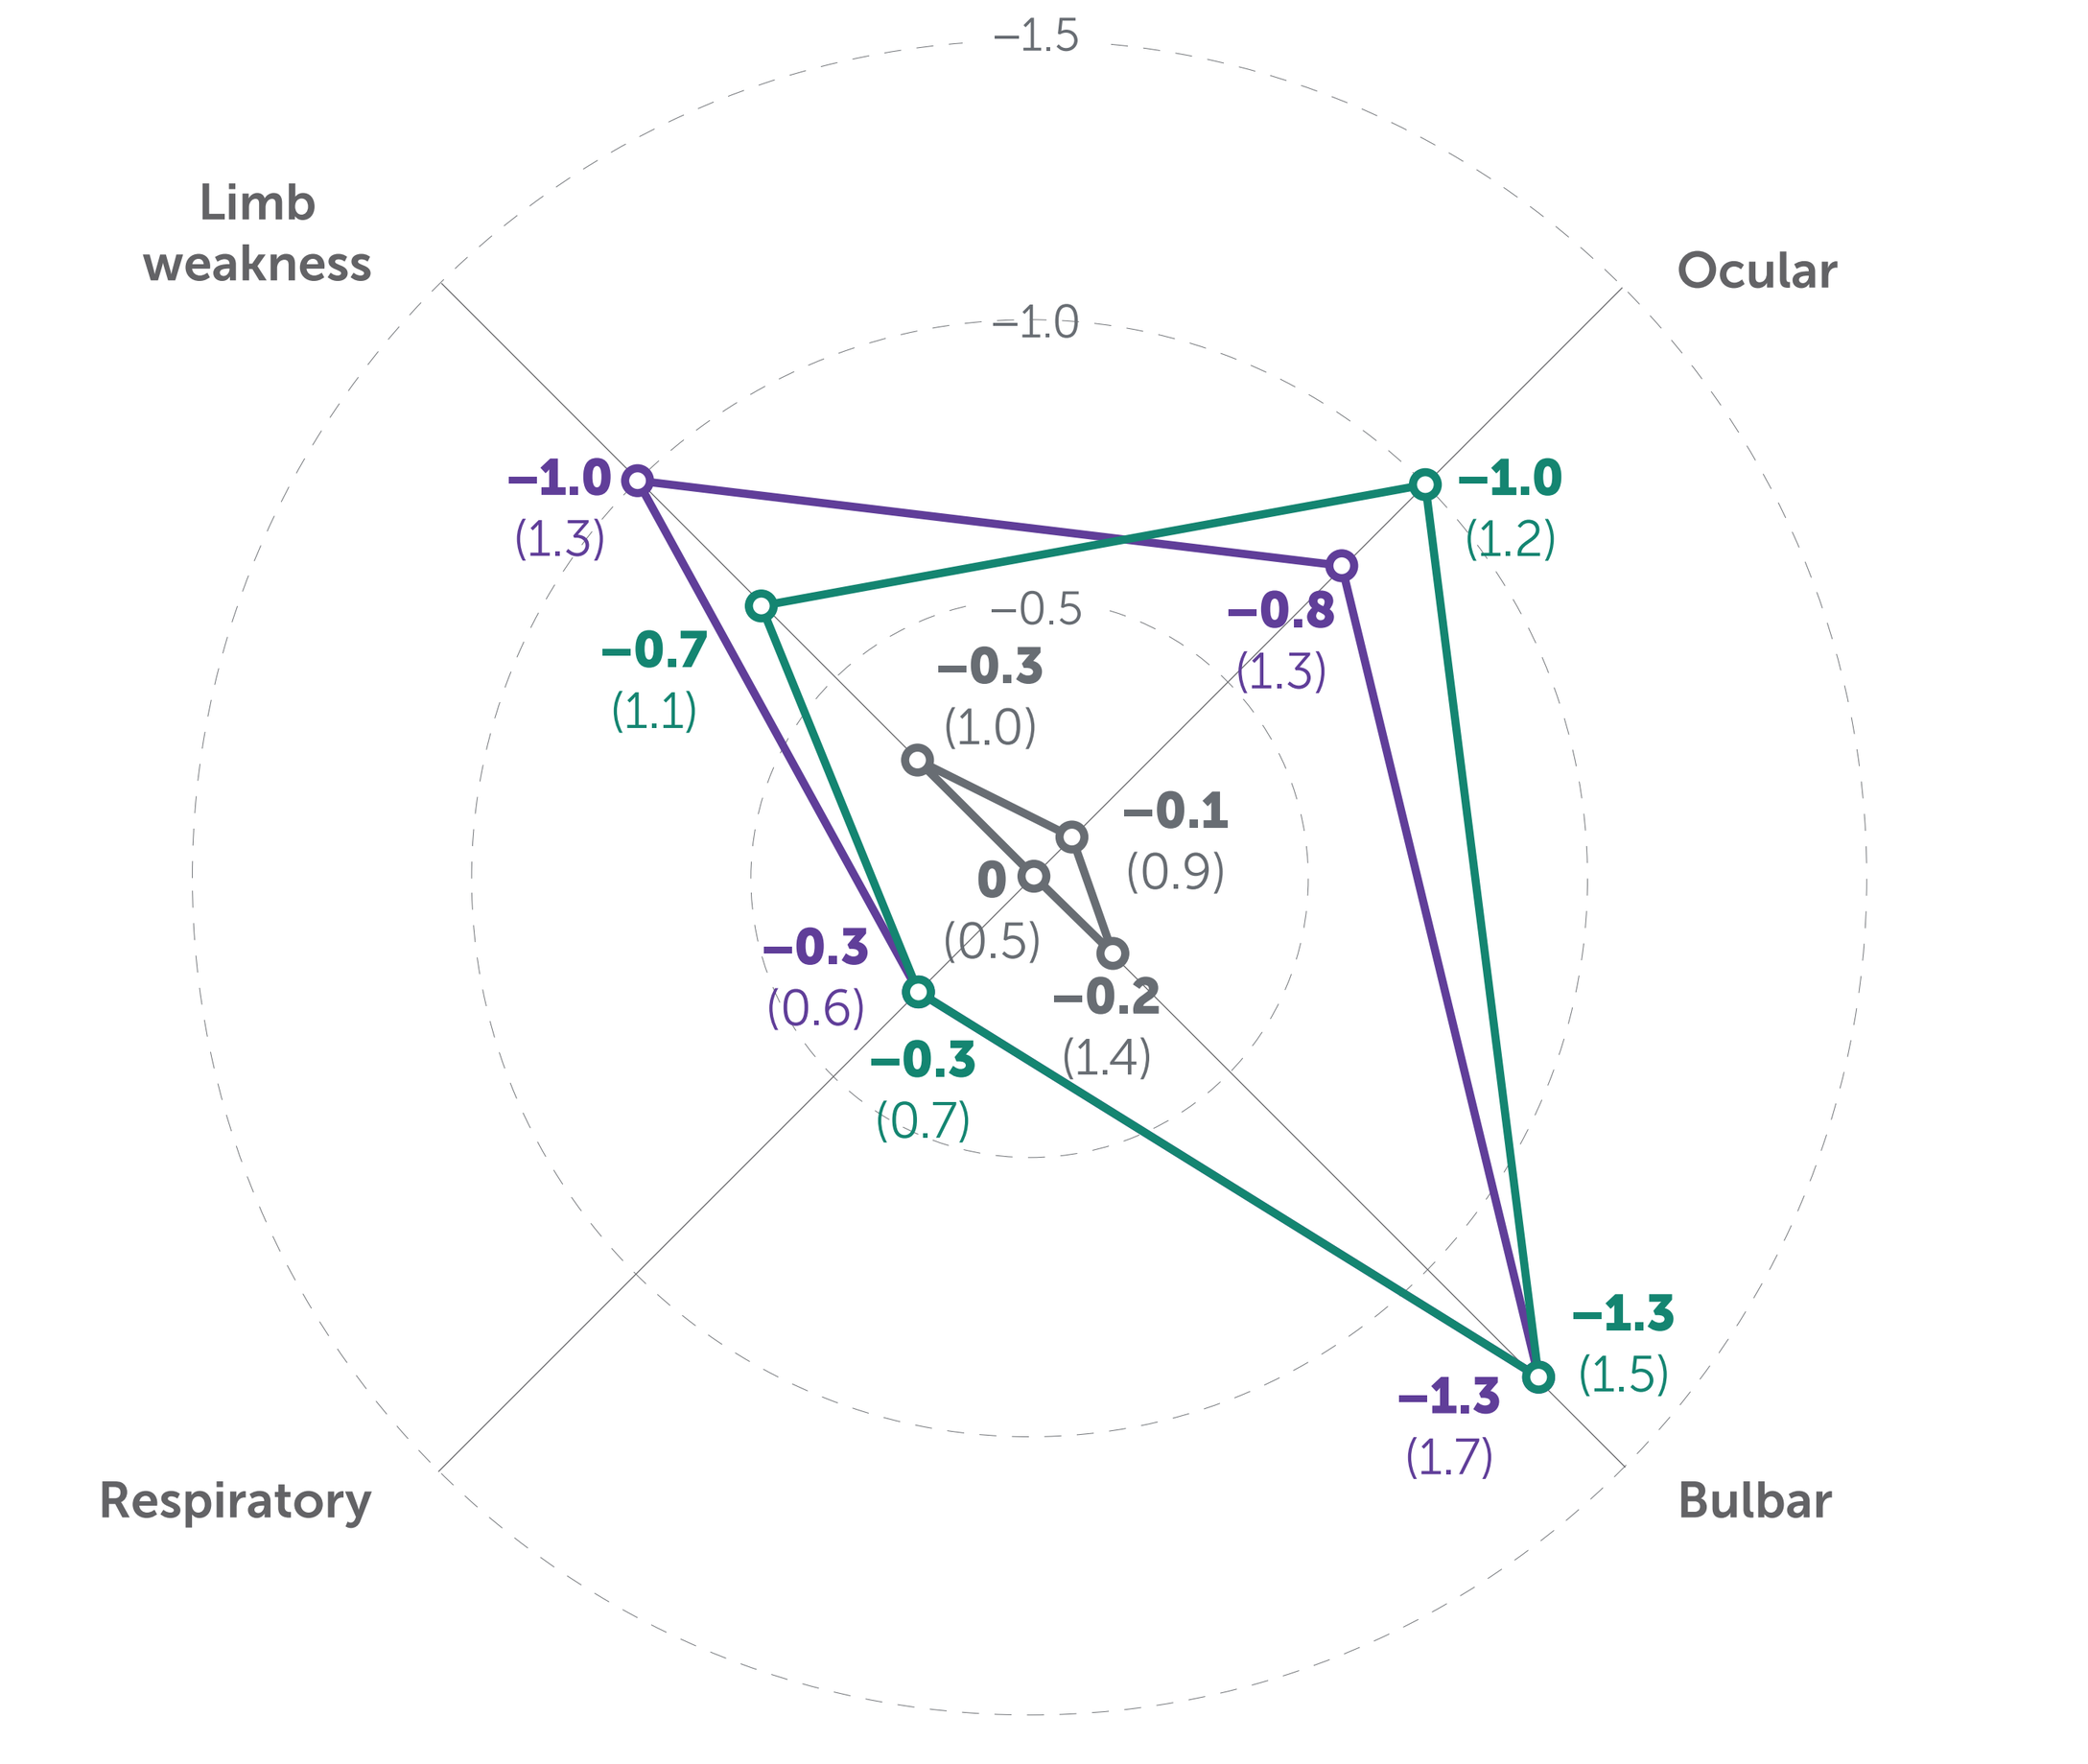
−1.5
−1.0
−0.5
Limb weakness
Ocular
Respiratory
Bulbar
−1.0 (1.3)
−0.7 (1.1)
−0.3 (1.0)
−0.8 (1.3)
−1.0 (1.2)
−0.1 (0.9)
0 (0.5)
−0.2 (1.4)
−0.3 (0.6)
−0.3 (0.7)
−1.3 (1.5)
−1.3 (1.7)

Mean (SD) baseline subdomain scores	Ocular	Bulbar	Limb weakness	Respiratory
Placebo (n=67)	**2.9** (1.6)	**2.4** (1.6)	**2.2** (1.2)	**1.0** (0.6)
RLZ 7 mg/kg (n=66)	**2.6** (1.8)	**2.5** (1.7)	**2.2** (1.5)	**1.0** (0.5)
RLZ 10 mg/kg (n=67)	**2.6** (1.7)	**2.6** (1.6)	**2.0** (1.2)	**0.9** (0.5)

Randomized set. Patients with MGFA Disease Class IVb/V disease were excluded from MycarinG.

Figure 3 Mean (SD) CFB in QMG subdomain scores at Day 43

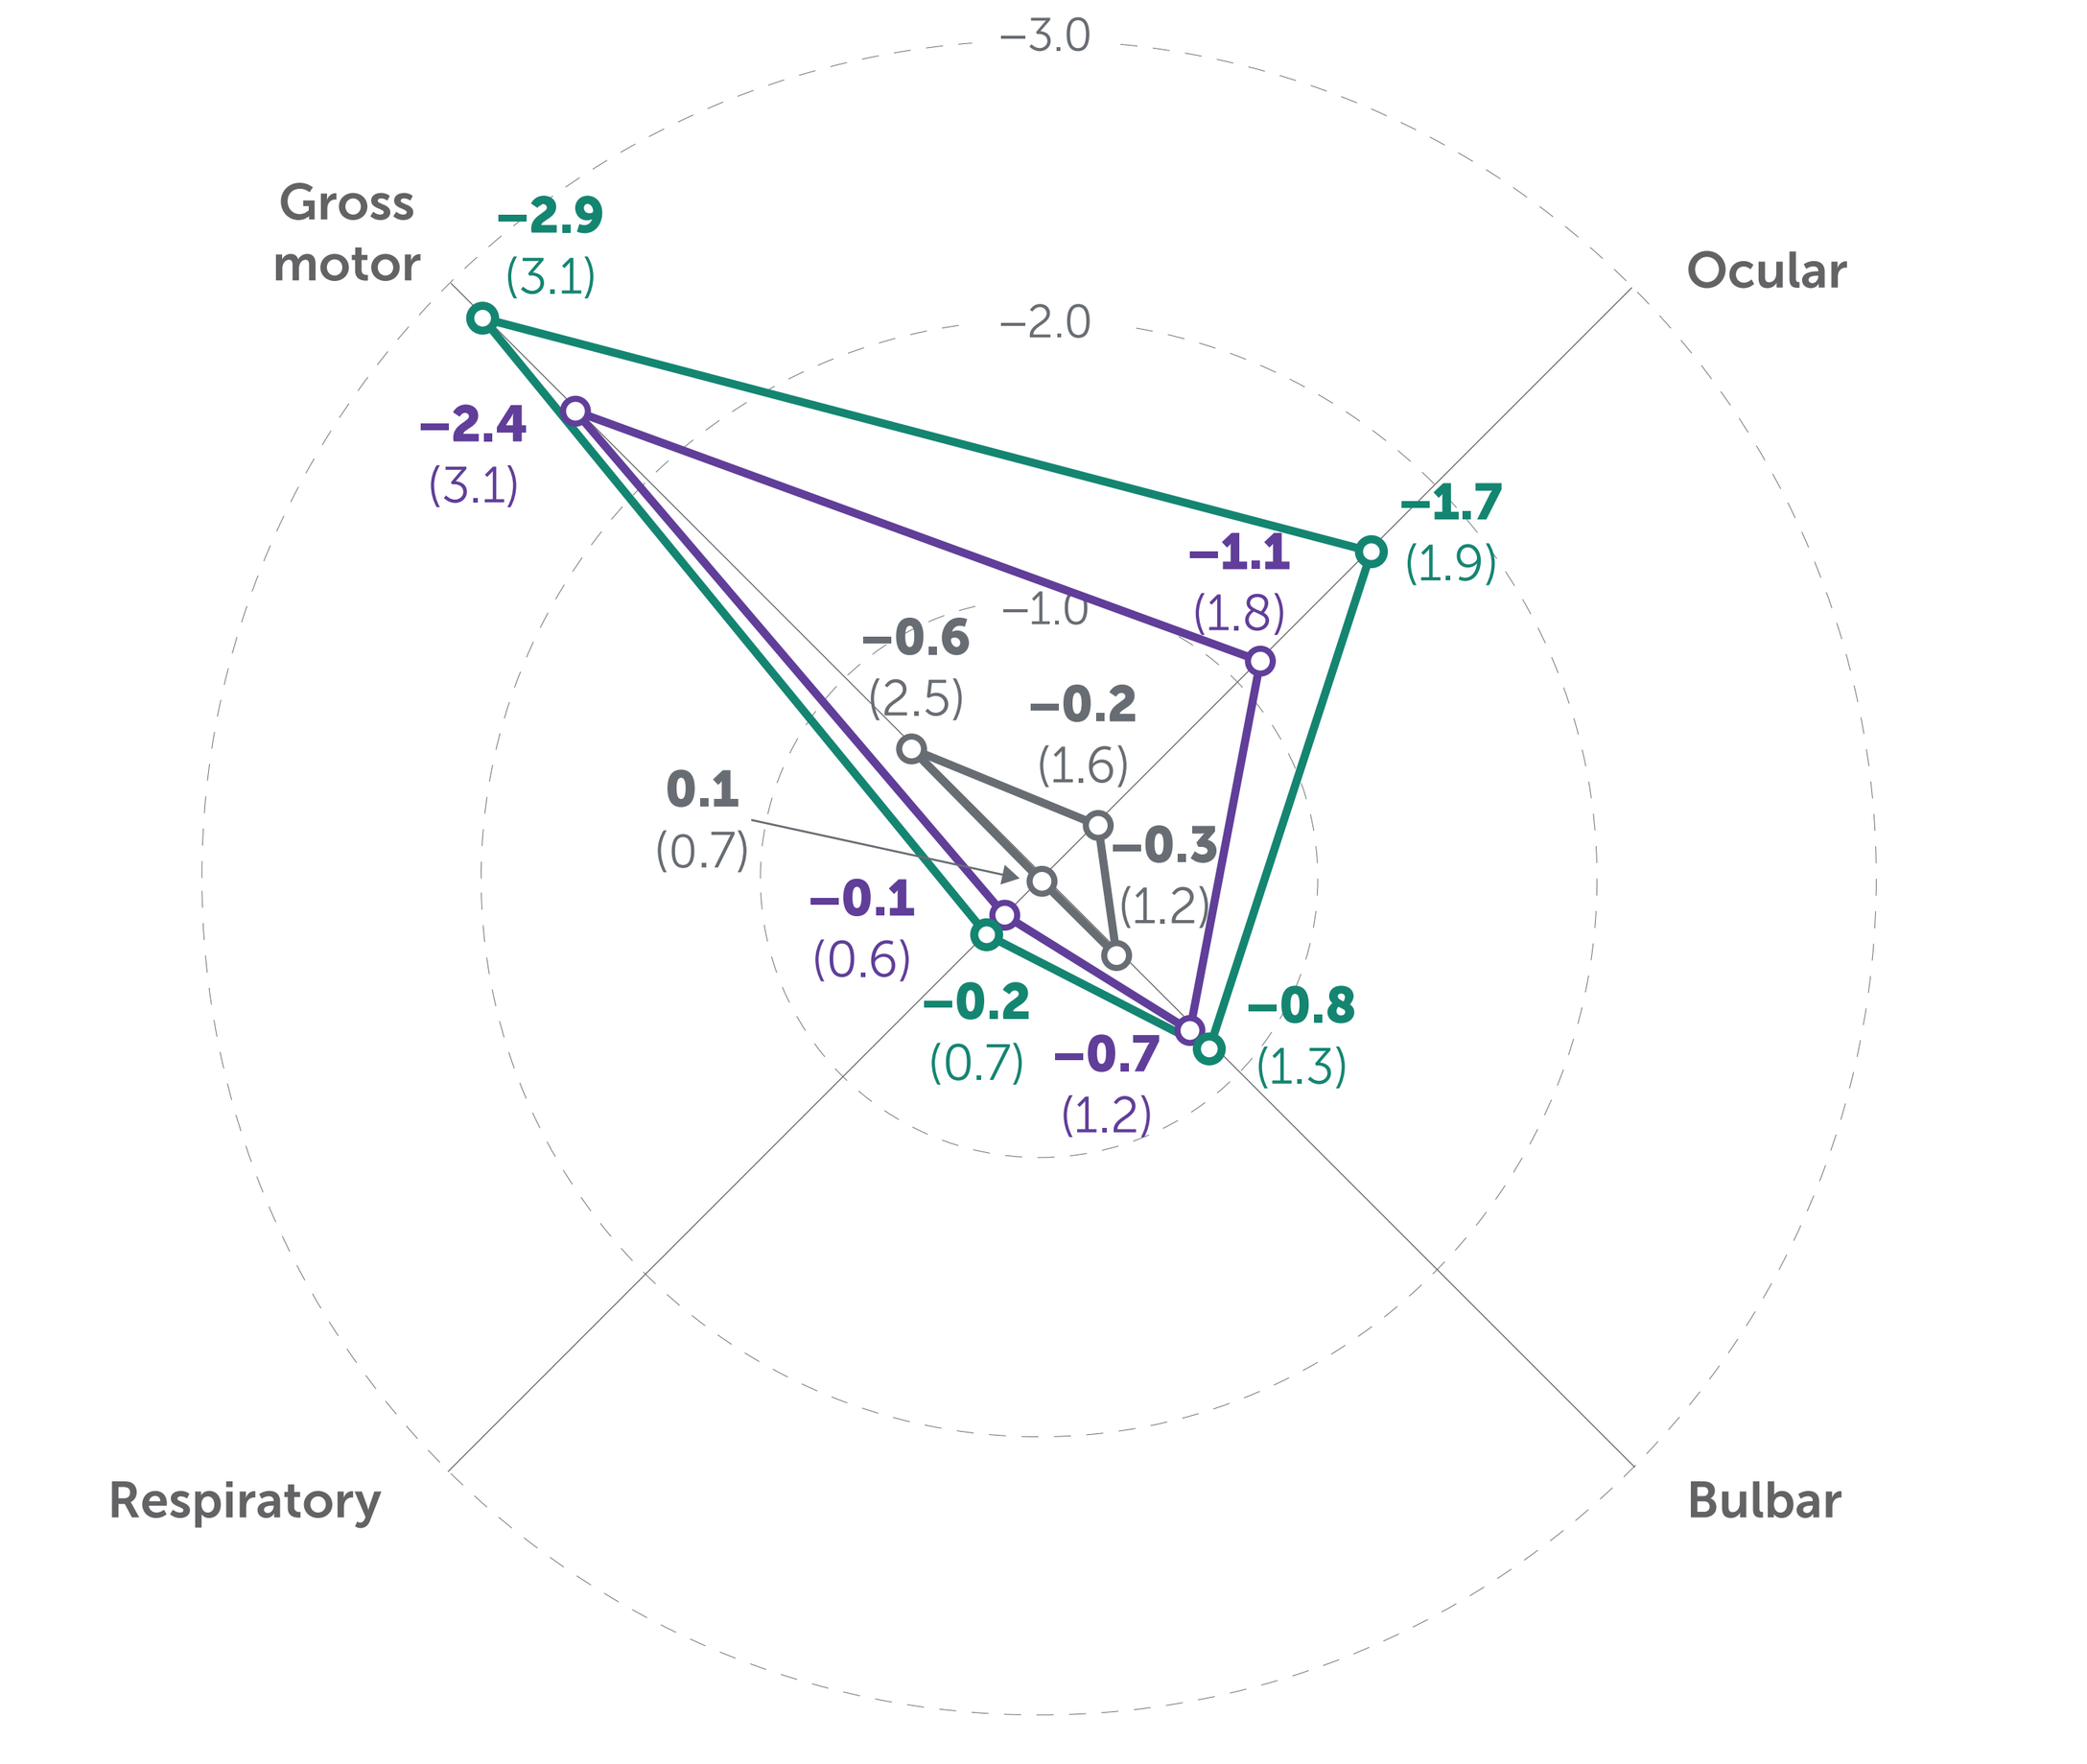
−3.0
−2.0
−1.0
Gross motor
Ocular
Respiratory
Bulbar
−2.9 (3.1)
−2.4 (3.1)
−0.6 (2.5)
−0.2 (1.6)
−1.1 (1.8)
−1.7 (1.9)
0.1 (0.7)
−0.1 (0.6)
−0.3 (1.2)
−0.2 (0.7)
−0.7 (1.2)
−0.8 (1.3)

Mean (SD) baseline subdomain scores	Ocular	Bulbar	Gross motor	Respiratory
Placebo (n=67)	**3.7** (2.1)	**1.5** (1.5)	**10.0** (2.5)	**0.6** (0.9)
RLZ 7 mg/kg (n=66)	**3.7** (2.0)	**1.2** (1.2)	**10.0** (2.7)	**0.5** (0.9)
RLZ 10 mg/kg (n=67)	**3.9** (1.9)	**1.5** (1.3)	**9.4** (2.9)	**0.7** (1.0)

Randomized set. Patients with MGFA Disease Class IVb/V disease were excluded from MycarinG.

Summary and conclusions

In the Phase 3 MycarinG study, one 6-week cycle of rozanolixizumab treatment significantly improved MG-ADL and QMG total scores versus placebo in patients with gMG

Compared with placebo, rozanolixizumab treatment led to improvements at Day 43 across ocular, bulbar, respiratory and limb weakness/gross motor muscle groups evaluated by MG-ADL and QMG subdomain scores

The findings from *post hoc* analyses support the efficacy of rozanolixizumab across a broad range of signs and symptoms experienced by patients with gMG, including those with ocular and bulbar symptoms

Table 1 MG-ADL and QMG items comprising the muscle group for each subdomain score

Muscle group	MG-ADL*	QMG†
Ocular	• Double vision • Eyelid droop	• Double vision • Ptosis (upward gaze) • Facial muscles
Bulbar	• Talking • Chewing • Swallowing	• Speech • Swallowing
Respiratory	• Breathing	• Forced vital capacity
Limb weakness/gross motor	• Impairment of ability to brush teeth or comb hair • Impairment of ability to arise from a chair	• Right arm outstretched • Left arm outstretched • Right hand grip • Left hand grip • Head lift • Right leg outstretched • Left leg outstretched

*The total MG-ADL score ranges from 0 to 24, with a higher score indicating more severe disability.
†The total QMG score ranges from 0 to 39, with a higher score indicating more severe disability.

Table 2 Baseline demographic and disease characteristics

		Placebo (n=67)	RLZ 7 mg/kg (n=66)	RLZ 10 mg/kg (n=67)
Age, years, mean (SD)		**50.4** (17.7)	**53.2** (14.7)	**51.9** (16.5)
Sex, female, n (%)		**47** (70.1)	**39** (59.1)	**35** (52.2)
AChR Ab+, n (%)		**59** (88.1)	**60** (90.9)	**60** (89.6)
MuSK Ab+, n (%)		**8** (11.9)	**5** (7.6)	**8** (11.9)
Duration of disease, years, mean (SD)		**9.4** (9.3)	**6.9** (6.8)	**9.6** (9.9)
MG-ADL score, mean (SD)		**8.4** (3.4)	**8.4** (3.8)	**8.1** (2.9)
QMG score, mean (SD)		**15.8** (3.5)	**15.4** (3.7)	**15.6** (3.7)
MGFA Disease Class, n (%)	IIa/b	**23** (34.3)	**29** (43.9)	**26** (38.8)
	IIIa/b	**41** (61.2)	**34** (51.5)	**39** (58.2)
	IVa/b*	**3** (4.5)	**3** (4.5)	**2** (3.0)

Randomized set.
*Only one patient, who was randomized to the placebo group, had Class IVb disease.

Table 3 Overview of TEAEs

Preferred term	Placebo (n=67), % (n)	RLZ 7 mg/kg (n=64), % (n)*	RLZ 10 mg/kg (n=69), % (n)*
Any TEAE	**67.2** (45)	**81.3** (52)	**82.6** (57)
Serious TEAEs	**9.0** (6)	**7.8** (5)	**10.1** (7)
Patient withdrawal from study due to TEAEs	**3.0** (2)	**3.1** (2)	**7.2** (5)
Treatment-related TEAEs†	**32.8** (22)	**50.0** (32)	**56.5** (39)
Severe TEAEs	**4.5** (3)	**4.7** (3)	**18.8** (13)
All deaths (AEs leading to death)	**0**	**0**	**0**

Safety set.
*Two patients in the rozanolixizumab 7 mg/kg group who incorrectly received 10 mg/kg were analyzed in the 10 mg/kg group for safety analyses.
†Treatment-related TEAEs as assessed by investigators.

Abbreviations: AChR Ab+, acetylcholine receptor autoantibody positive; AE, adverse event; CFB, change from baseline; FcRn, neonatal Fc receptor; (g)MG, (generalized) myasthenia gravis; IgG4, immunoglobulin G4; LS, least squares; mAb, monoclonal antibody; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MuSK Ab+, muscle-specific tyrosine kinase autoantibody positive; QMG, Quantitative Myasthenia Gravis; RLZ, rozanolixizumab; SD, standard deviation; SE, standard error; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Grace O'Malley, MSci, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Robert M. Pascuzzi is Professor Emeritus of Neurology at Indiana University and receives compensation for his professional work from Indiana University Health. He has no financial relationship with any pharmaceutical company and receives no compensation from any pharmaceutical company (present or past). Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organization Medical Education Resources (an educational organization with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation or publication. Ali A. Habib has received research support from Alexion Pharmaceuticals, argenx, Cabaletta Bio, Genentech/Roche, Immunovant, Regeneron Pharmaceuticals, UCB and Viela Bio (now Amgen). He has received honoraria from Alexion Pharmaceuticals, Alpine Immune Sciences, argenx, Genentech/Roche, Immunovant, Inhibrx, Regeneron Pharmaceuticals, NMD Pharma and UCB. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Arvinas, Biogen, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research, travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, Immunovant, Johnson & Johnson, NMD Pharmaceuticals, Regeneron Pharmaceuticals and UCB, and has served as a speaker for Alexion/AstraZeneca Rare Disease, argenx, and CSL Behring. He performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, ImmunAbs and UCB. Asha Hareendran and Thaïs Tarancón are employees and shareholders of UCB. Francesca Pannullo is a contractor for UCB. Vera Bril is a Consultant for Akcea, Alexion Pharmaceuticals, Alnylam, argenx, CSL, Grifols, Ionis, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson), Novo Nordisk, Octapharma, Pfizer, Powell Mansfield, Roche, Sanofi, Takeda Pharmaceuticals and UCB. She has received research support from Akcea, Alexion Pharmaceuticals, argenx, CSL, Grifols, Immunovant, Ionis, Momenta (now Johnson & Johnson), Octapharma, Takeda Pharmaceuticals, UCB and Viela Bio (now Amgen).

References: 1. Bril V, et al. Lancet Neurol. 2023;22(5):383–394. 2. Rozanolixizumab US PI. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/761286s000lbl.pdf. Accessed July 2024. 3. de Meel RHP, et al. Lancet Neurol. 2018;17(3):204–205. 4. Barnett C, et al. Neurology. 2017;89(23):2357–2364. 5. Zinman L, et al. Neurology. 2007;68(11):837–841. 6. Mantegazza R, et al. Ann Clin Transl Neurol. 2020;7(8):1327–1339. 7. Bril V, et al. Eur J Neurol. 2024;31(1):e16098. 8. Wolfe GI, et al. Neurology. 1999;52(7):1487–1489. 9. Barohn RJ, et al. Ann N Y Acad Sci. 1998;841(1):769–772.

Please use this QR code to download a PDF of the poster.