Poster **288**

Evidence of misdiagnosis in administrative claims data for individuals with myasthenia gravis

AANEM 2024; Savannah, GA, USA; October 15–18, 2024

Judith Thompson[1], Bo Zhang[1], Joshua N. Liberman[2], Jonathan Darer[2]

[1]UCB, Smyrna, GA, USA; [2]Health Analytics, LLC, Clarksville, MD, USA

Introduction

- Misdiagnosis occurs when the named diagnosis given to a patient does not represent the patient's true condition[1]
- The true incidence of misdiagnosis is not easily assessed, but several studies comparing the initial and final diagnosis during a hospitalization demonstrate discordance of 9% or greater[2,3]
- Establishing a diagnosis of MG, a rare neuromuscular disorder, can be complicated by variations in symptom presentation

Study objectives

- To estimate the prevalence of misdiagnosis in MG and establish whether social determinants of health are associated with misdiagnosis in MG[4]

Methods

- Total study period: September 1, 2017 to August 31, 2022 (**Figure 1**)
- Patient selection period: September 1, 2018 to August 31, 2022
- Inclusion criteria:
 - ≥2 outpatient claims ≥90 days apart or one inpatient claim with an ICD-10-CM G70.0x
 - ≥18 years of age at index
 - A ≥12-month pre-index baseline period with continuous claims coverage
 - A ≥12-month post-index follow-up period with continuous claims coverage
 - ≥1 pharmacy claim in the study period
 - Assignment to a ZIP5 code (based on PCP ZIP code)
- Exclusion criteria:
 - History of MG, thymectomy, AChEI, or biologic treatment during the Exclusion and Diagnostic Assessment Periods
- Covariate measures:
 - Demographics: age, sex, insurance type
 - Comorbidities: medical and psychiatric
 - Measures assessing social determinants of health included community risk measures of housing, food, economic, transportation, health literacy, social connectedness, and digital landscape. Individuals were assigned to a community (i.e., ZIP5) based on rendering PCP address
- Outcome measures:
 - Possible misdiagnosis: defined by select conditions with ≥1 medical claim(s) before index followed by absence of corresponding medical claims following index
 - Conditions included stroke, COPD, chronic fatigue syndrome, MS, thyroid eye disease, Hashimoto's thyroiditis, malignant thymus neoplasm, Guillain-Barré syndrome, Graves' disease, systemic lupus erythematosus, amyotrophic lateral ALS, and myositis
- Analysis:
 - Univariate analysis of possible misdiagnoses and time from initial misdiagnosis to index. Unadjusted comparison of measures assessing social determinants of health by those with and without possible misdiagnosis
 - Logistic regression of baseline characteristics associated with misdiagnosis (yes=1, no=0)
 - Chi-squared test for categorical variables and Student's t-test for continuous variables

Results

- A total of 3,873 individuals met the eligibility criteria and were included
- Possible misdiagnosis was present in 425 (11.0%) of the eligible individuals
- Most common possible misdiagnoses were stroke (5.5%), COPD (4.5%), chronic fatigue syndrome (2.2%), Graves' disease (1.5%), MS (1.4%), and Guillain-Barré syndrome (1.1%) (**Figure 2**)
- Of those with a possible misdiagnosis, most (51.5%) received only one possible misdiagnosis, while 30.1% received two, and 18.4% received three or more possible misdiagnoses
- Of those with a possible misdiagnosis, 56.0% received that diagnosis more than 6 months before the initial MG diagnosis (**Figure 3**)
- Individuals who were assigned to geographies with high-risk housing ($p<0.10$) or high-risk social connectedness ($p<0.05$) scale scores were more likely to receive a possible misdiagnosis (**Figure 4**)
- Diagnoses of several symptoms associated with MG, including muscle weakness, dysarthria, dysphagia, and diplopia, were all associated with elevated likelihood of receiving a misdiagnosis (**Figure 5**)
- Medical comorbidities (CCI, hypertension, sleep apnea), psychiatric comorbidity (anxiety), social connectedness risk, and receiving the initial MG diagnosis in an inpatient facility were all associated with elevated odds of receiving a possible misdiagnosis (**Figure 5**)

Figure 1 Study design

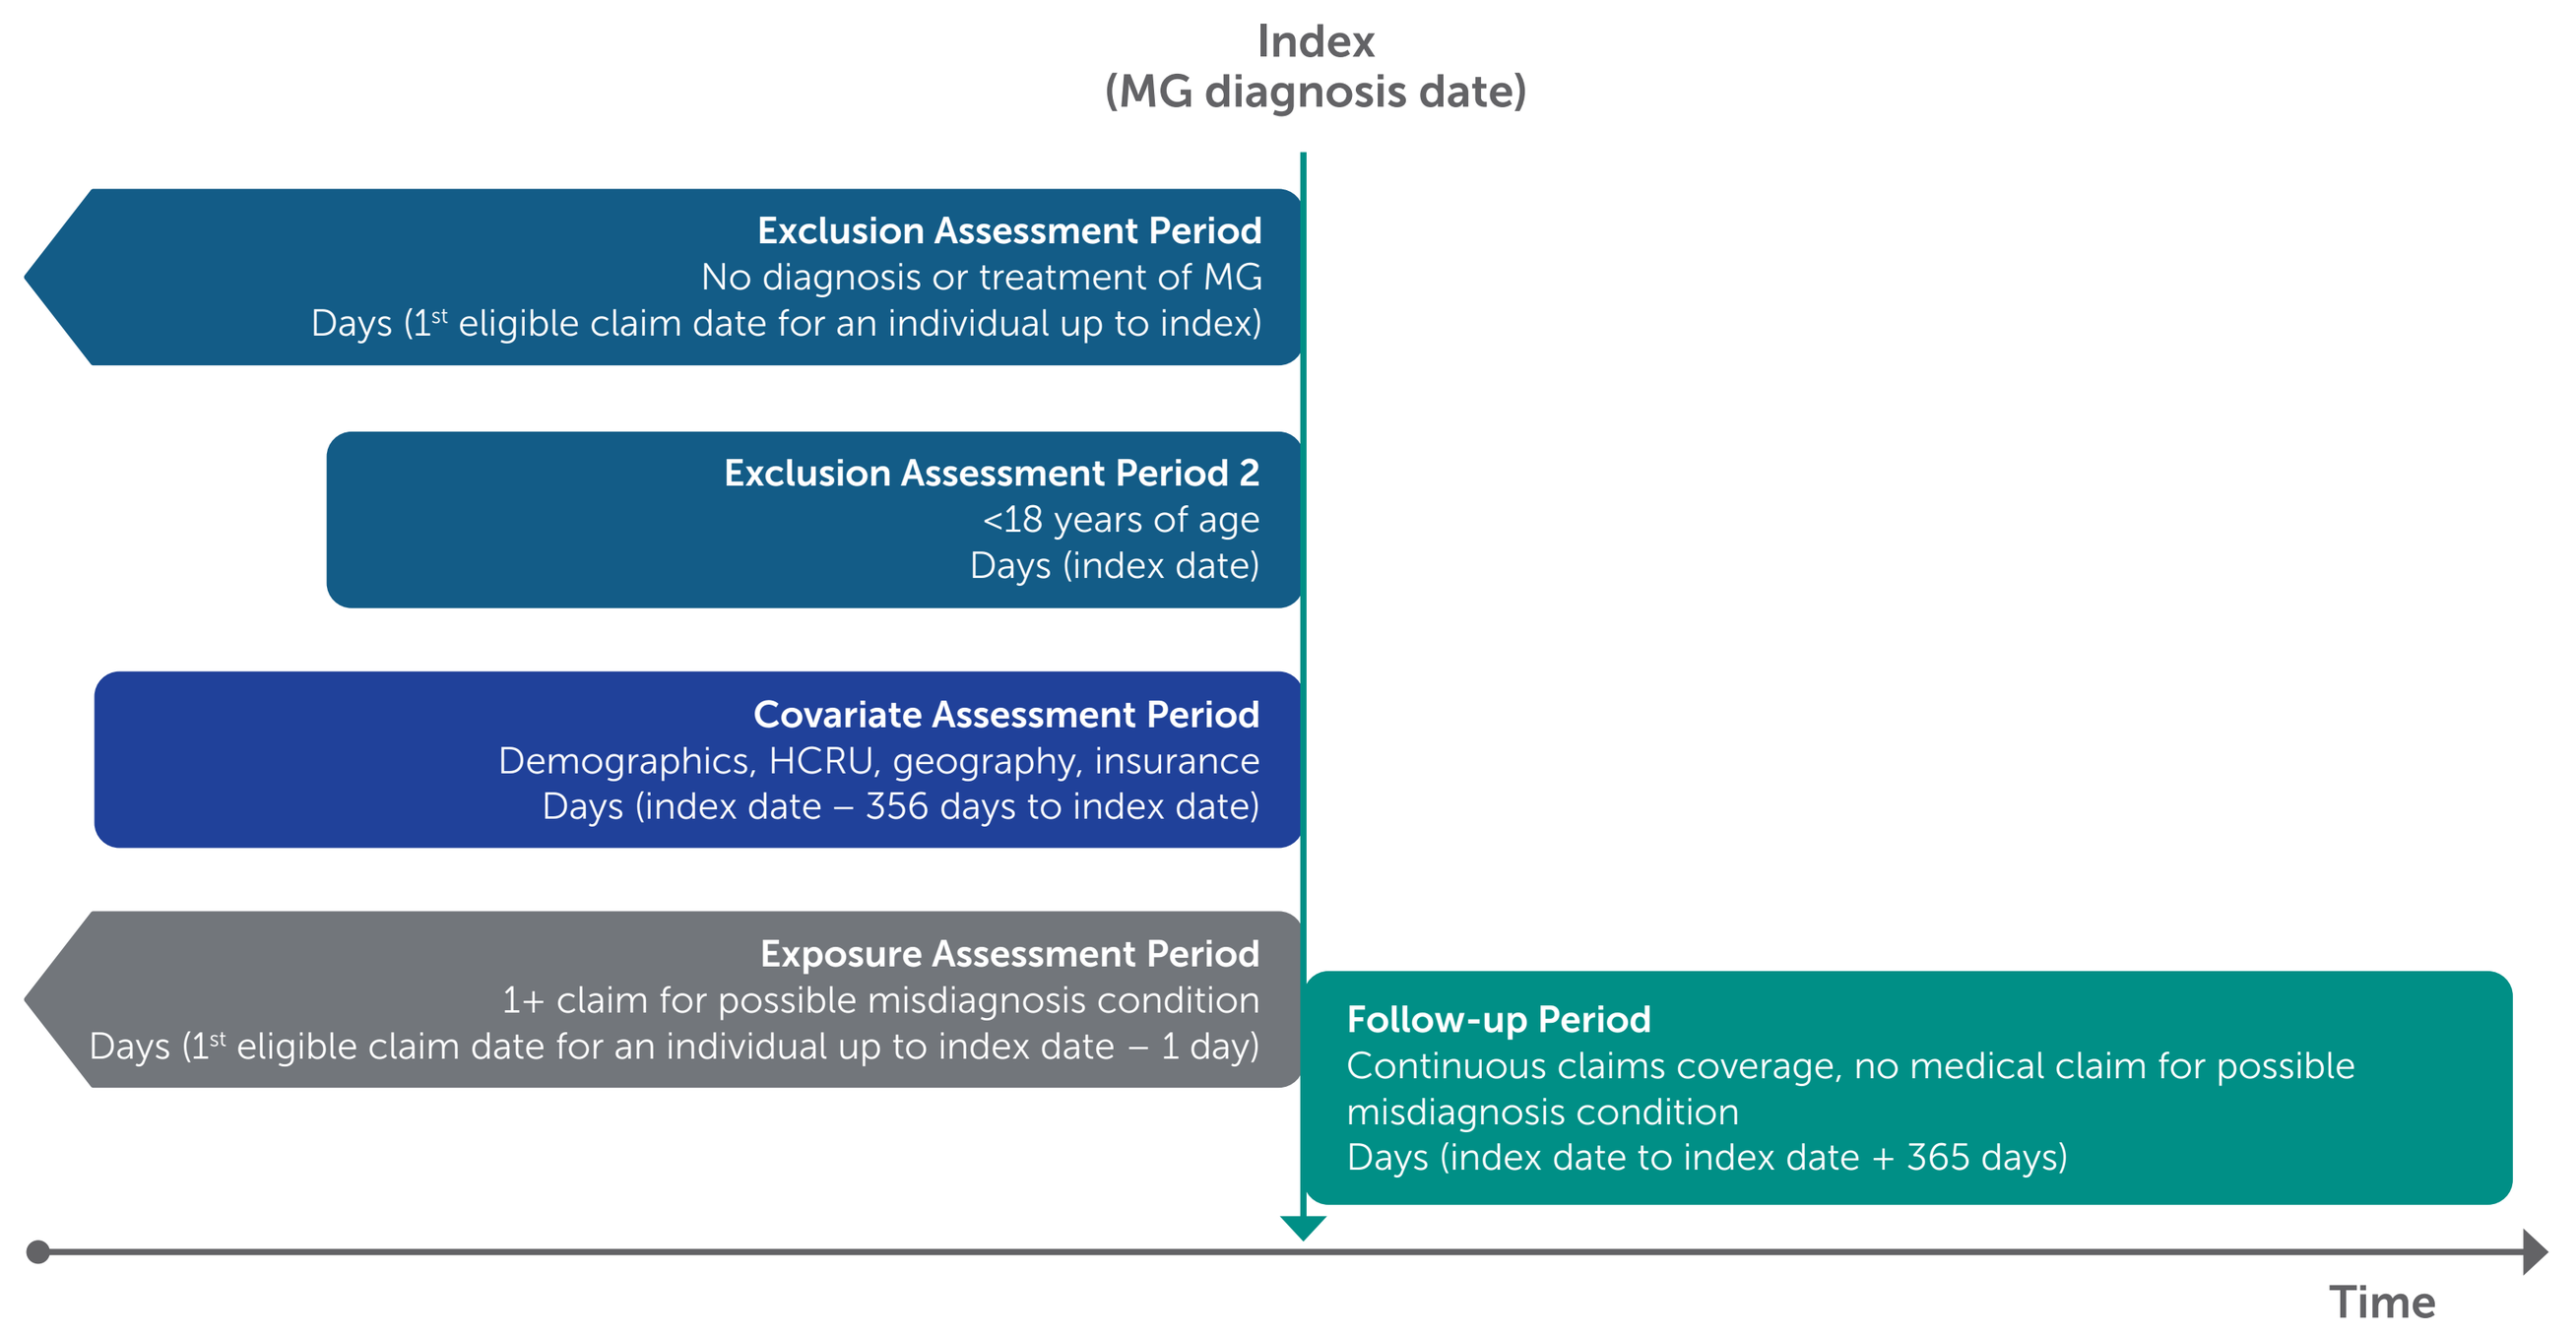
Index
(MG diagnosis date)
Exclusion Assessment Period
No diagnosis or treatment of MG
Days (1st eligible claim date for an individual up to index)
Exclusion Assessment Period 2
<18 years of age
Days (index date)
Covariate Assessment Period
Demographics, HCRU, geography, insurance
Days (index date – 356 days to index date)
Exposure Assessment Period
1+ claim for possible misdiagnosis condition
Days (1st eligible claim date for an individual up to index date – 1 day)
Follow-up Period
Continuous claims coverage, no medical claim for possible misdiagnosis condition
Days (index date to index date + 365 days)
Time

Figure 2 Percent of eligible study participants with a possible misdiagnosis

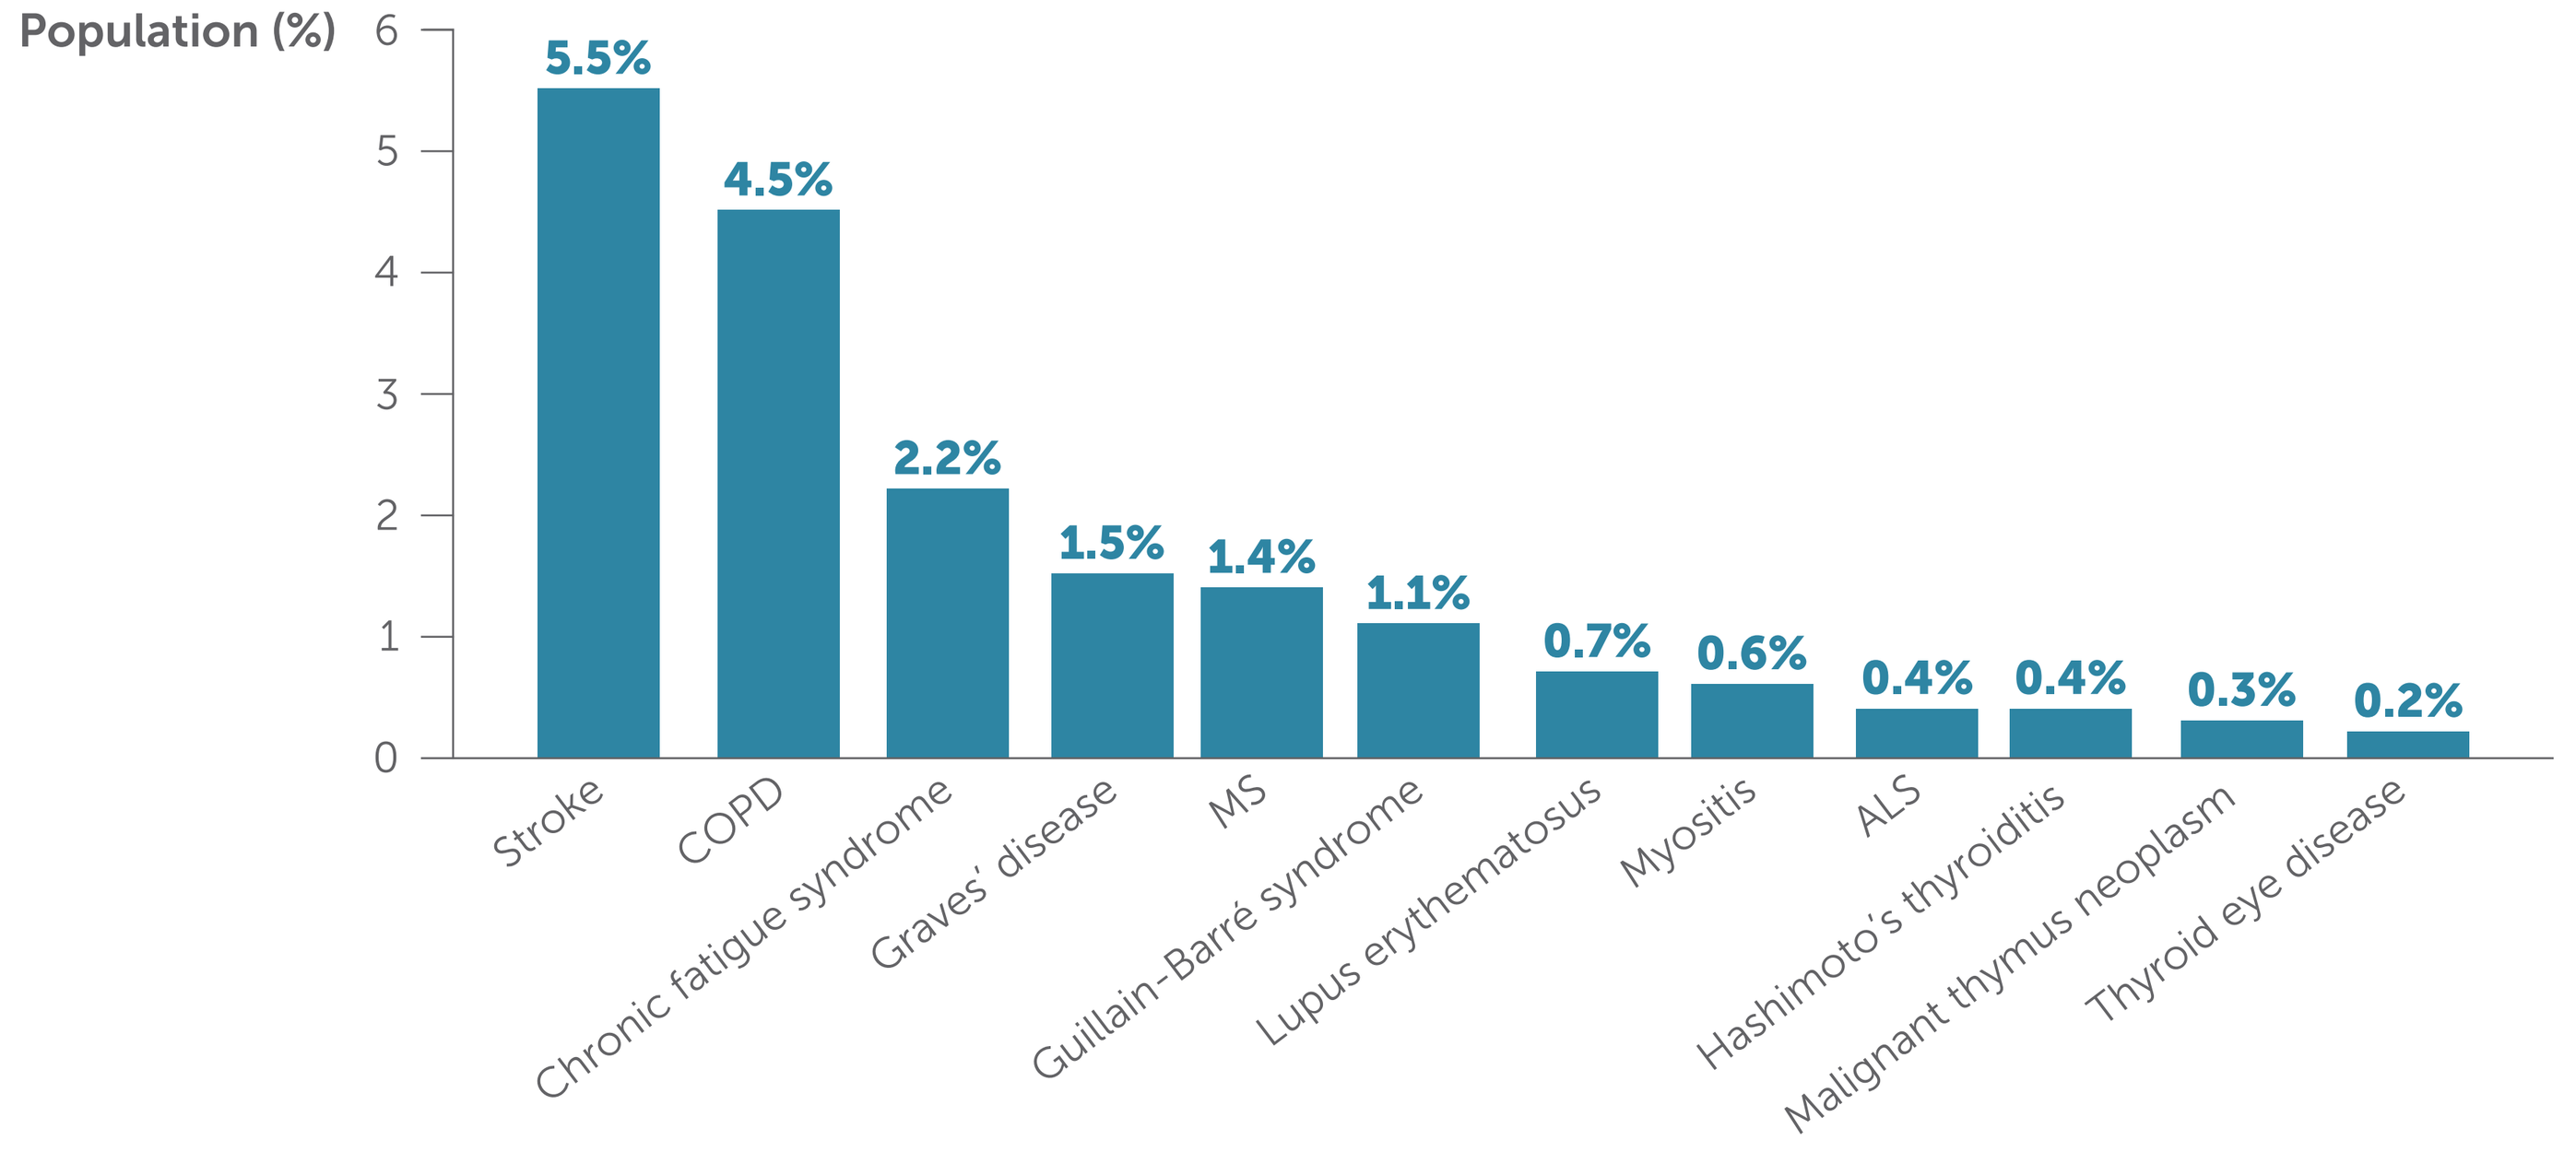
Population (%)
6
5
4
3
2
1
0
5.5%
4.5%
2.2%
1.5%
1.4%
1.1%
0.7%
0.6%
0.4%
0.4%
0.3%
0.2%
Stroke
COPD
Chronic fatigue syndrome
Graves' disease
MS
Guillain-Barré syndrome
Lupus erythematosus
Myositis
ALS
Hashimoto's thyroiditis
Malignant thymus neoplasm
Thyroid eye disease

Figure 3 Percent distribution of time from first possible misdiagnosis date to initial MG diagnosis date (n=425)

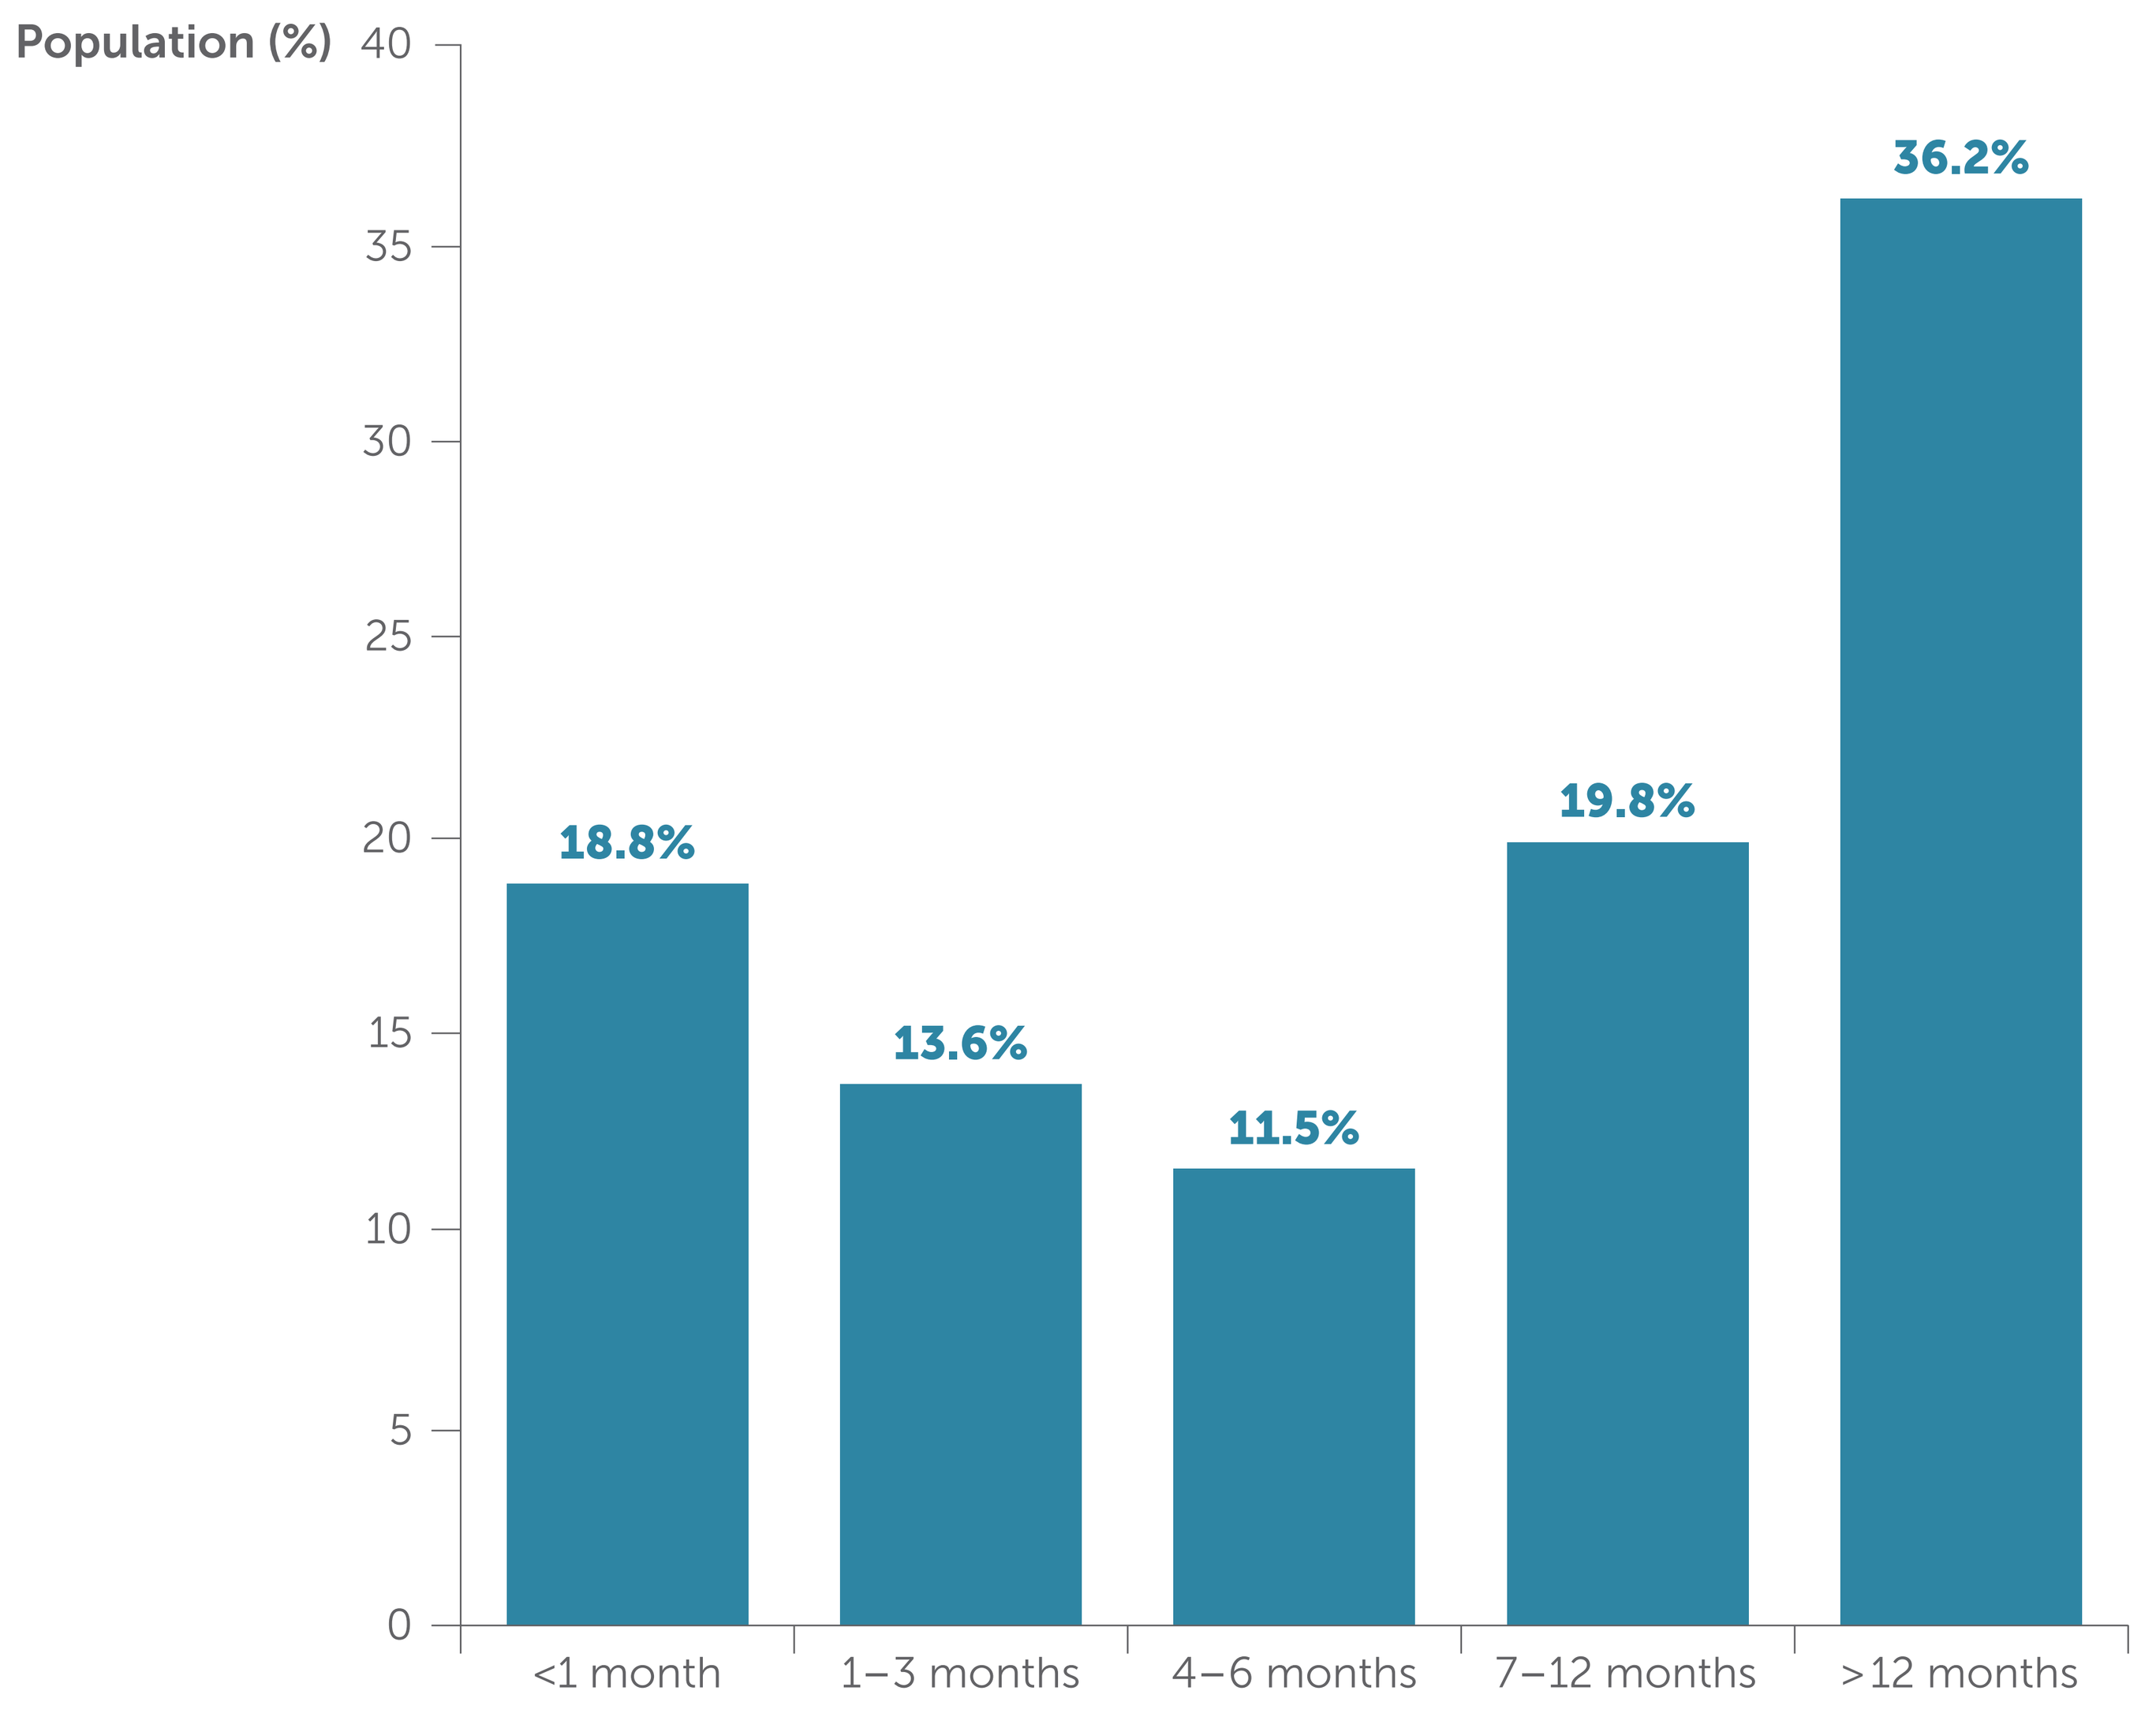
Population (%)
40
35
30
25
20
15
10
5
0
18.8%
13.6%
11.5%
19.8%
36.2%
<1 month
1–3 months
4–6 months
7–12 months
>12 months

Figure 4 Percent of population assigned to high-risk communities, by presence or absence of a possible misdiagnosis (n=3,873)

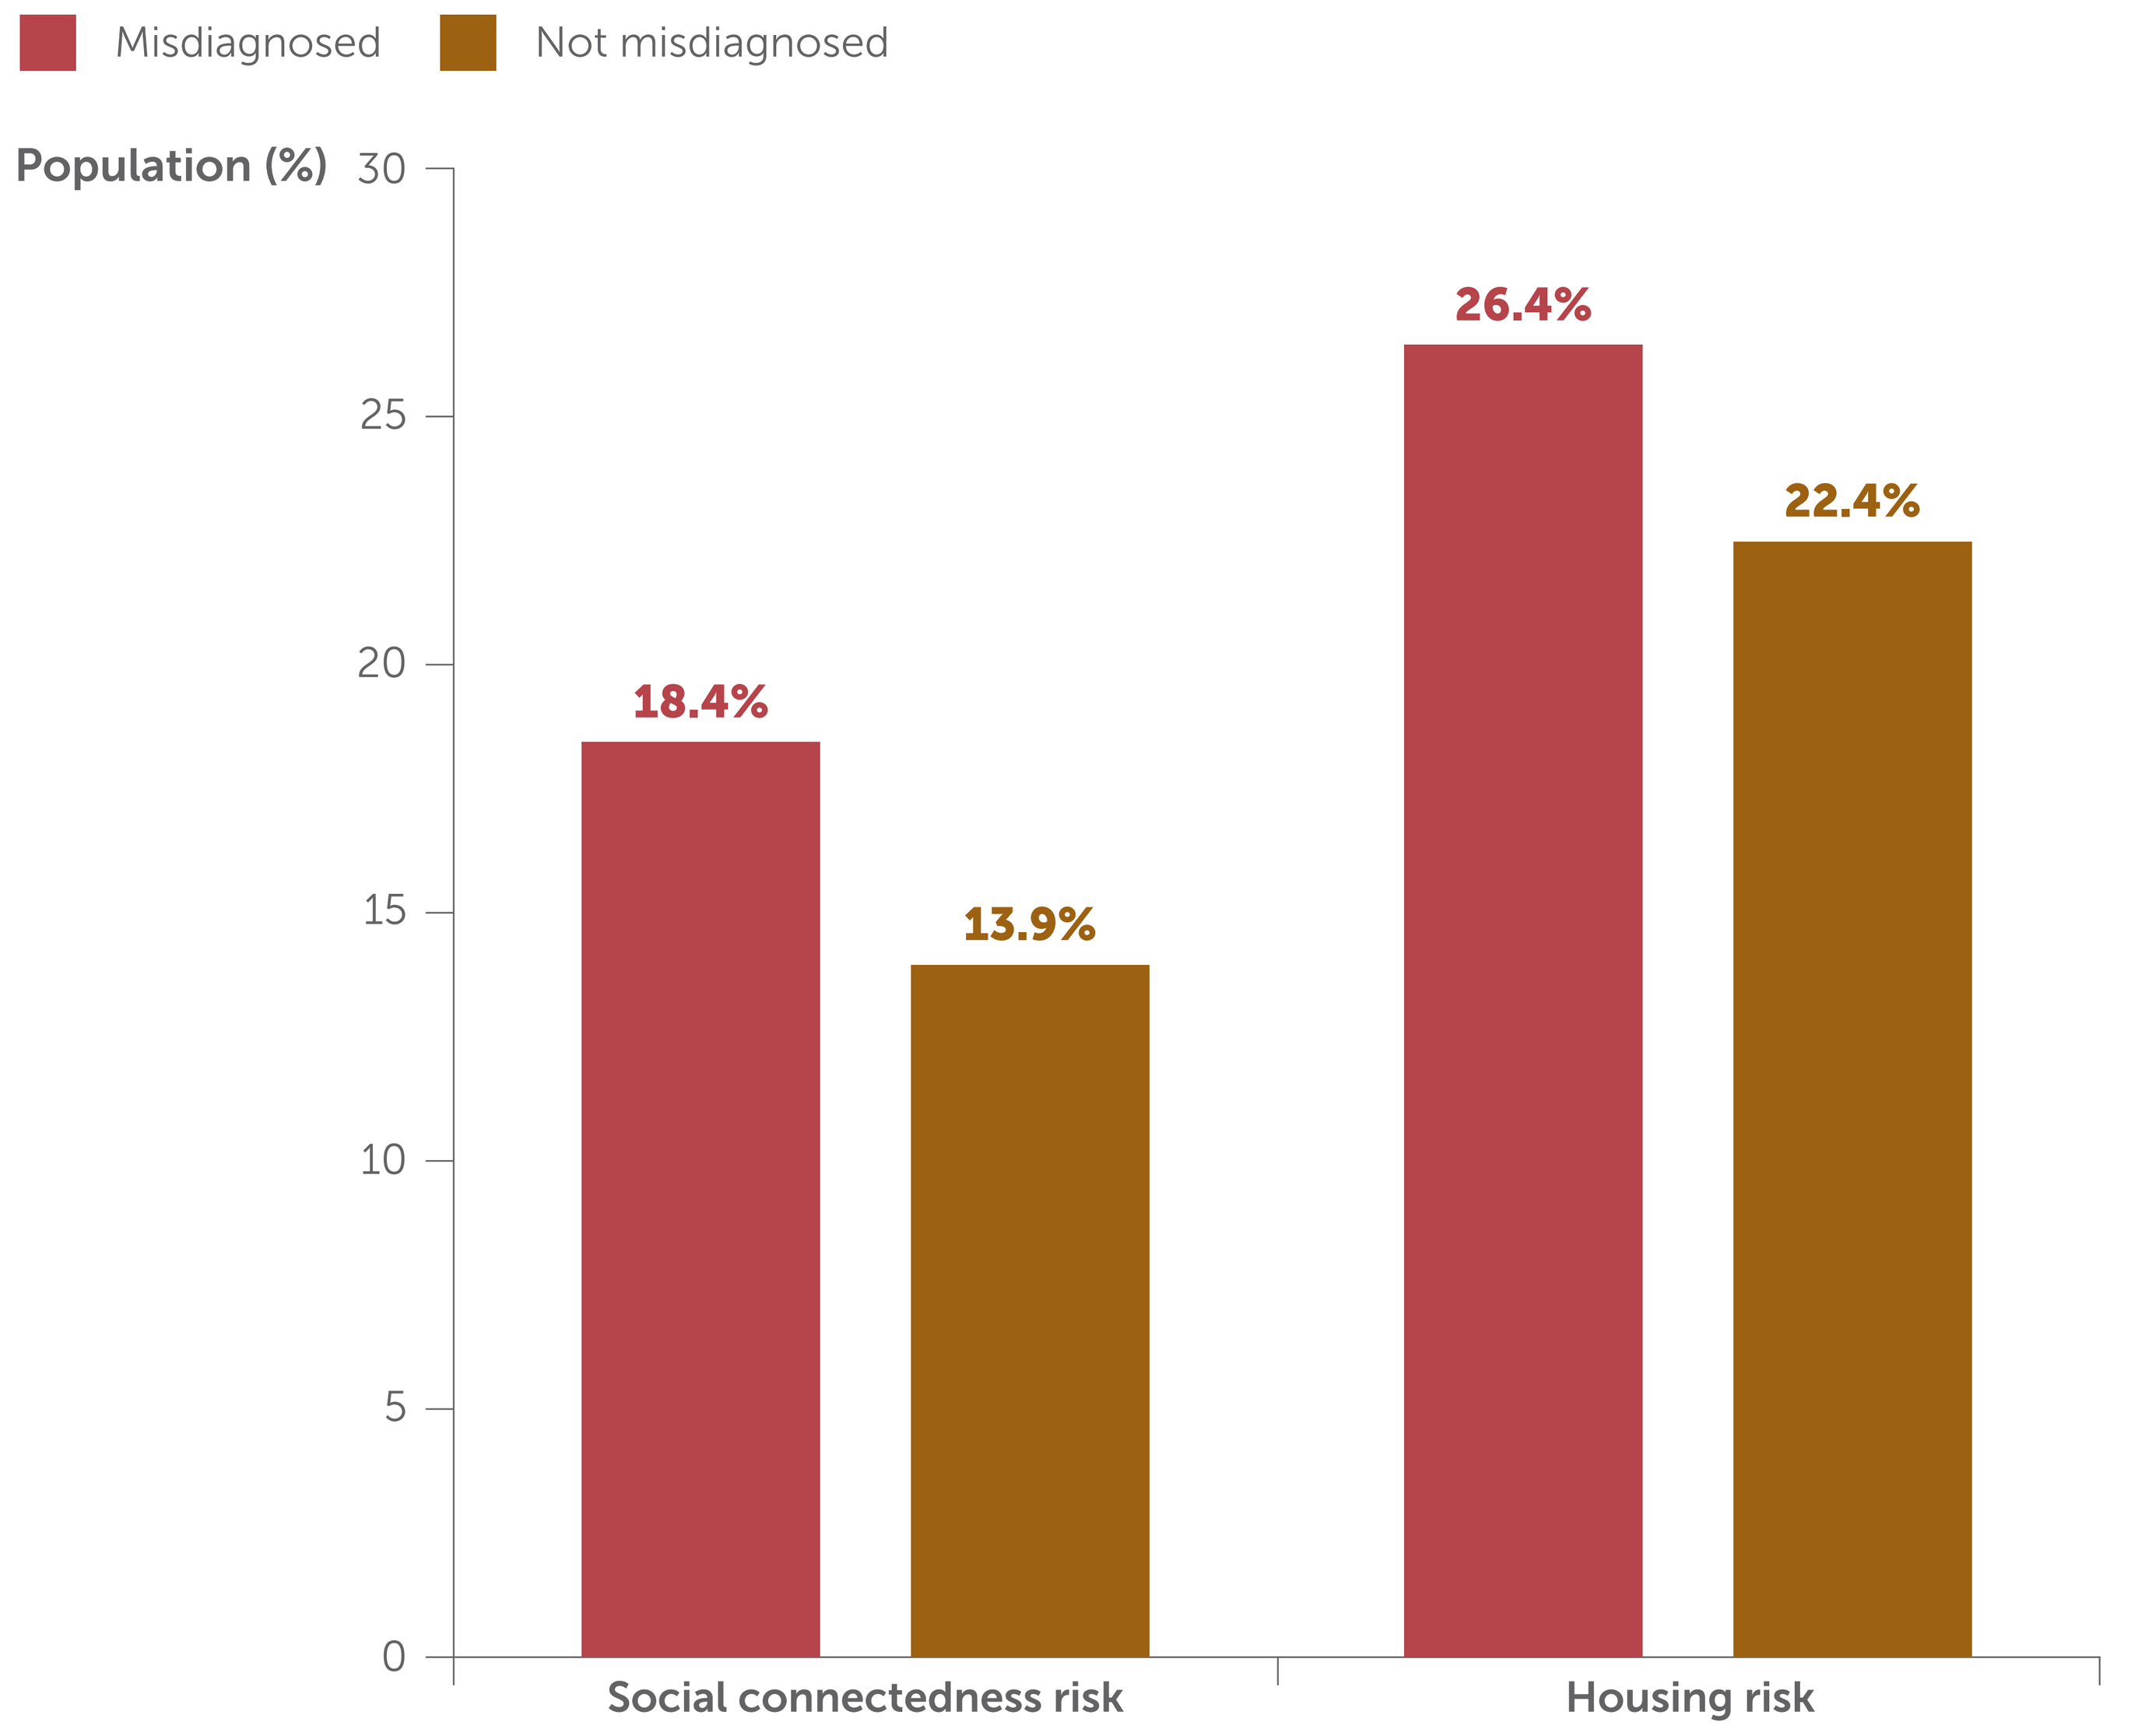
Misdiagnosed
Not misdiagnosed
Population (%)
30
25
20
15
10
5
0
18.4%
13.9%
26.4%
22.4%
Social connectedness risk
Housing risk

Summary and conclusions

- Receiving a possible misdiagnosis is associated with an extended period of time before receiving the MG diagnosis suggesting a delay in initiating appropriate treatment
- The association of misdiagnosis with being diagnosed as an inpatient also suggests the possibility of harm to the patient resulting from the possible misdiagnosis
- Almost half of individuals with a possible misdiagnosis received two or more diagnoses
- Residing in a high-risk community was associated with elevated odds of possible misdiagnosis
- Uncommon presentations of MG can also result in misdiagnosis. Symptoms such as slurred speech, dysphagia,[1] and hemibody weakness[2] may increase risk of incorrect diagnosis of MG as stroke

Figure 5 Adjusted OR and 95% CI for factors associated with receiving a possible misdiagnosis

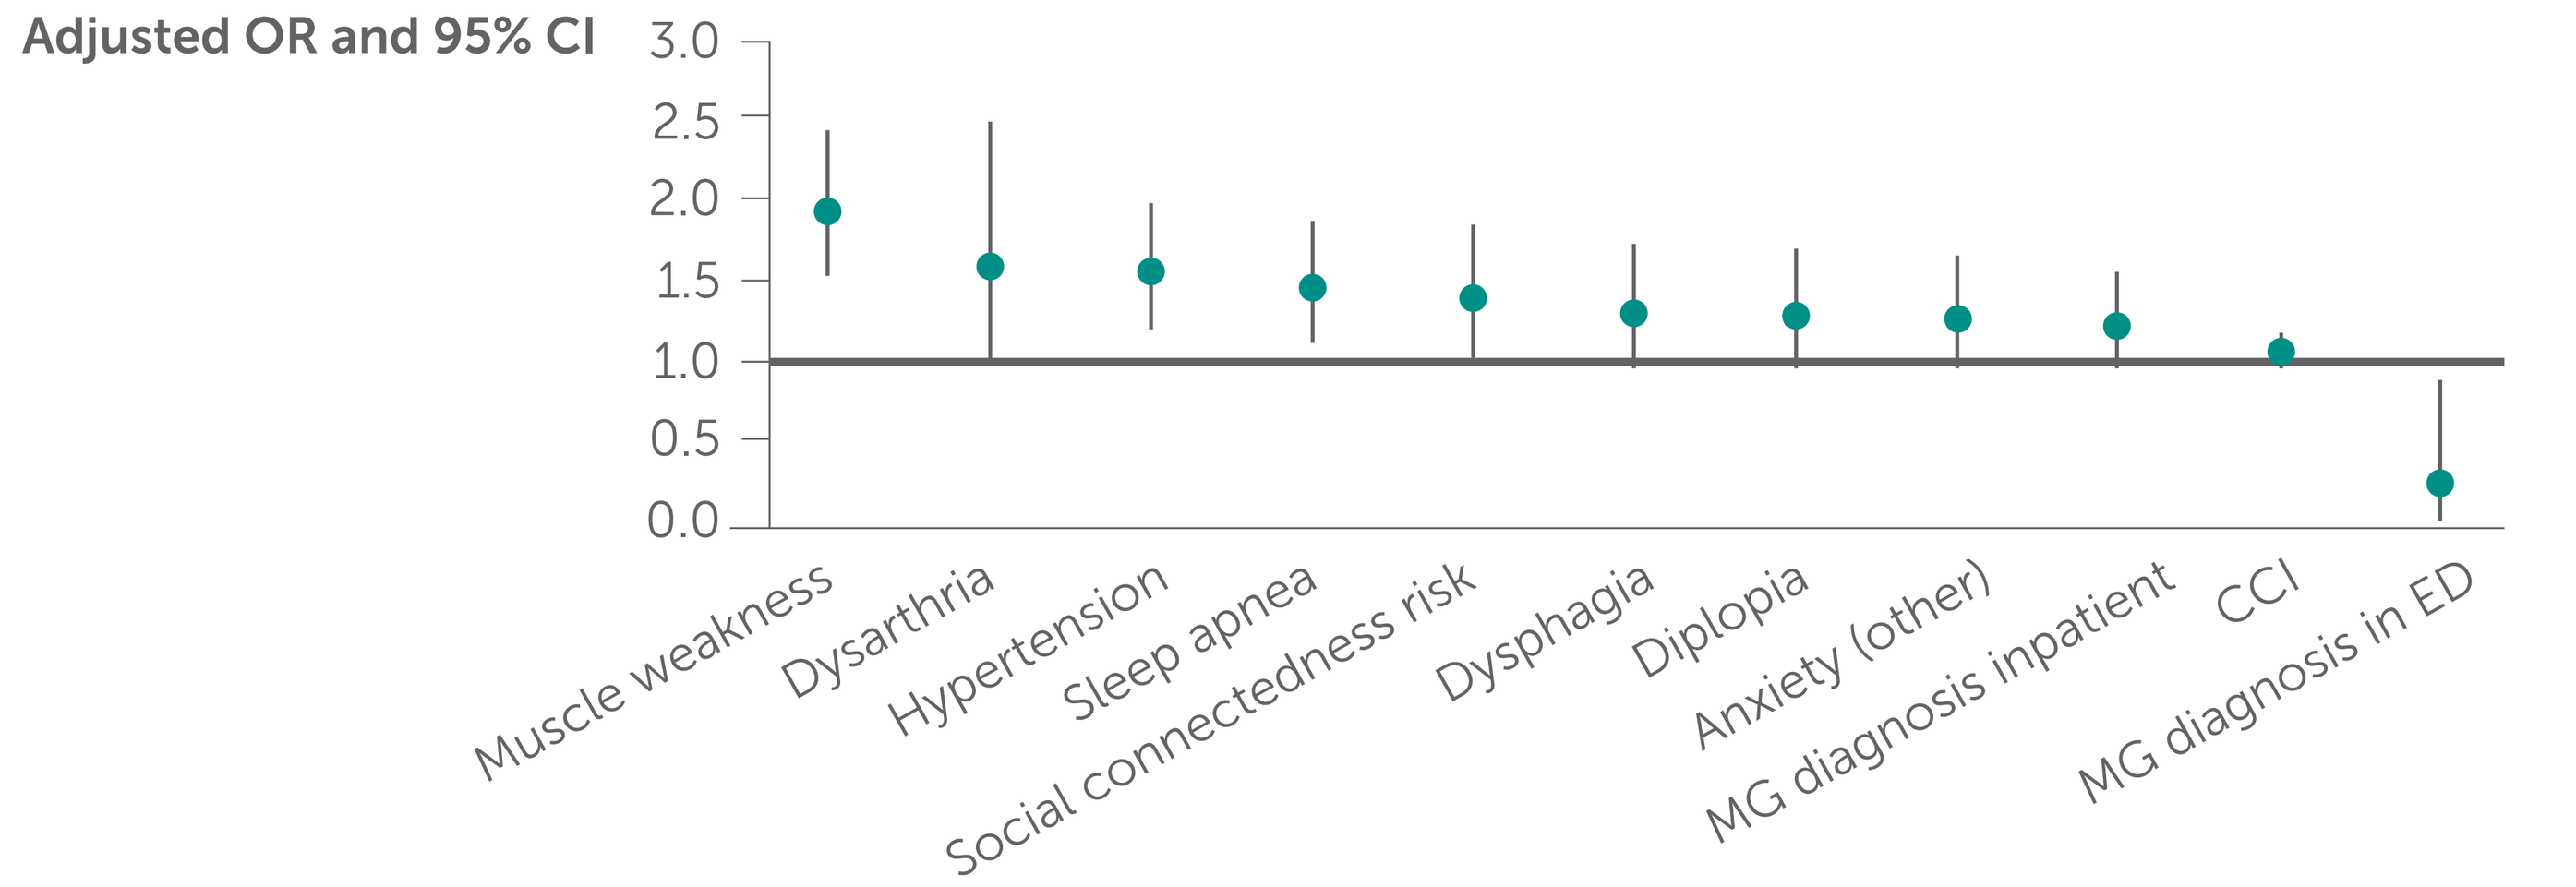
Adjusted OR and 95% CI
3.0
2.5
2.0
1.5
1.0
0.5
0.0
Muscle weakness
Dysarthria
Hypertension
Sleep apnea
Social connectedness risk
Dysphagia
Diplopia
Anxiety (other)
MG diagnosis inpatient
CCI
MG diagnosis in ED

Abbreviations: AChEI, acetylcholinesterase inhibitor; ALS, amyotrophic lateral sclerosis; CCI, Charlson Comorbidity Index; CI, confidence interval; COPD, chronic obstructive pulmonary disease; ED, emergency department; HCRU, health care resource utilization; ICD-10-CM, International Classification of Diseases, 10th Revision, Clinical Modification; MG, myasthenia gravis; MS, multiple sclerosis; OR, odds ratio; PCP, primary care provider; ZIP, Zone Improvement Plan.

Author disclosures: This study was funded by UCB. Editorial assistance was provided by Ogilvy Health, London, UK, which was funded by UCB. Judith Thompson and Bo Zhang are employees of UCB. Joshua N. Liberman and Jonathan Darer are employees of Health Analytics and were contracted by UCB to carry out the analysis.

References: 1. Committee on Diagnostic Error in Health Care, et al. Improving diagnosis in health care. Washington, DC, US: National Academies Press, 2015. 2. Chatterjee S, et al. Gap analysis between provisional diagnosis and final diagnosis in government and private teaching hospitals: a record-linked comparative study. J Family Med Prim Care. 2016;5:637–640. 3. Katongole SP, et al. Prevalence and classification of misdiagnosis among hospitalised patients in five general hospitals of Central Uganda. Clin Audit. 2022;14:65–77. 4. Socially Determined. Homepage. www.sociallydetermined.com. Accessed September 2024.

Please use this QR code to download a PDF of the poster.