Comprehensive Analysis of Lennox-Gastaut Syndrome in Europe: Treatment Patterns, Healthcare Utilisation, and Quality of Life

Adam Strzelczyk[1] ;
Antonio Gil-Nagel[2];
Pasquale Striano[3,4];
Richard Chin[5];
Iryna Leunikava[6];
Beatriz Rodriguez[7];
Alexander Gillespie[8];
Amelie Lothe[9];
Alexis Arzimanoglou[10];

[1]Epilepsy Center Frankfurt Rhine-Main, Department of Neurology, Goethe-University Frankfurt, Frankfurt am Main, Germany; [2]Hospital Ruber Internacional, Madrid, Spain; [3]Istituto Di Ricovero E Cura A Carattere Scientifico (IRCCS) Istituto Giannina Gaslini, Member of the ERN Epicare, Genoa, Italy; [4]Department of Neurosciences, Rehabilitation, Ophthalmology, Genetics, Maternal and Child Health, University of Genova, Genova, Italy; [5]Royal Hospital for Children and Young People, Edinburgh, United Kingdom; [6]UCB Pharma, Monheim am Rhein, Germany; [7]UCB Pharma, Madrid, Spain; [8]Adelphi Real World, Bollington, United Kingdom; [9]UCB Pharma S.A., Colombes, France; [10]Director Epilepsy Program, Children's University Hospital San Juan de Dios, Coordinating member of the ERN EpiCARE, Barcelona, Spain

Poster #1.344

Background

- Lennox-Gastaut syndrome (LGS) is a rare developmental and epileptic encephalopathy that onsets in childhood, with an estimated incidence of 0.1-0.28 per 100,000 population.[1]
- The disease is characterised by repeated and varying seizures , treatment resistance, cognitive impairments, and as a result, an impaired quality of life (QoL). [2, 3]
- There are limited real-world data exploring the population of adults diagnosed with LGS, patients' current treatment usage (including use of rescue medication), healthcare resource utilisation (HCRU) or patient QoL.

Objective

- To describe the real-world evidence of patient demographics, treatment patterns, healthcare resource utilisation and quality of life among the Lennox-Gastaut Syndrome population in Europe, stratified by different age groups, including adults.

Methods

- Data were drawn from the Adelphi Real World LGS Disease Specific Programme™ a cross-sectional survey with retrospective data collection conducted in France, Germany, Italy, Spain and the United Kingdom from July 2022 - August 2023. [4, 5] Descriptive analyses were performed.
- Neurologists and paediatric neurologists completed record forms for the next ten consecutively consulting patients with LGS. The forms collected data on patient demographics, seizure frequency and type, antiseizure medications (ASMs), rescue medications (RMs), HCRU and QoL for patients with a confirmed LGS diagnosis.
- Patient data were analysed by age groups: <6 years, 6-18 years and >18 years and by seizure types (generalised tonic-clonic (GTC), "all drop" (tonic/atonic/GTC/secondary GTC), "non-drop" (atypical absences, myoclonic, myoclonic-atonic-myoclonic-tonic-clonic, clonic, focal onset aware, focal onset impaired awareness, hyperkinetic, non-motor onset, behaviour arrest, cognitive emotional, focal with observable motor signs) seizures, experienced in the past month).

Results

Patient Demographics & Characteristics

- In total, 93 neurologists and 21 paediatric neurologists completed records for 454 patients with a physician-confirmed LGS diagnosis across five countries: France (83), Germany (78), Italy (131), Spain (118), and the UK (44).
- Mean age (Standard Deviation; SD): 16.0 years (10.8); >18 years old: 34%; Male: 65%; Mean age at first seizure: 4.0 years (2.5) (Table 1).
- In the past month, 75% had experienced drop seizures and 65% non-drop seizures. <6 years (y) and 6-18y experienced tonic seizures most commonly, and atypical seizures were most prevalent for >18y.

Table 1: Patient Demographics & Characteristics

	Overall (n=454)	<6y (n=78)	6-18y (n=221)	>18y (n=155)	All drop seizures in past month (n=342)	Non-drop seizures in past month (n=294)
Age (mean years, (SD))	16.0 (10.8)	4.1 (1.0)	12.3 (4.5)	27.2 (9.7)	15.9 (10.6)	15.8 (11.1)
Male, n (%)	297 (65)	51 (65)	150 (68)	96 (62)	229 (67)	197 (67)
Age of first seizure (mean years, (SD))	4.0 (2.5)	2.7 (1.5)	4.6 (2.2)	4.0 (3.2)	4.1 (2.6)	4.1 (2.7)
Seizures within the last month (Top 5; %)						
Any drop seizure	75	76	75	76	100	62
Any non-drop seizure	65	71	65	61	53	100
Tonic	48	60	51	37	63	43
Atypical absence	40	40	39	41	35	62
Atonic	37	37	35	39	49	35

SD, standard deviation; y, years.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Treatment Patterns

- Patients had a mean (SD) of 2.3 (1.5) ASM treatment regimens since diagnosis. Most common prescribed ASMs: valproate, clobazam and lamotrigine **(Figure 1).**
- RMs were prescribed to 70% of patients, consistent between age groups (70-71%). The two highest prescribed RMs were diazepam and midazolam **(Figure 2)**. The third most prescribed RM product was buccolam for all age groups apart from >18y, where clonazepam was third most common.

Overview

QUESTION

What are the characteristics, treatment usage, healthcare resource utilisation and quality of life for patients with LGS across different age groups in Europe?

INVESTIGATION

From the Adelphi LGS Disease Specific Programme™ [4, 5], a real-world, cross-sectional survey with retrospective data collection, conducted in France, Germany, Italy, Spain and the United Kingdom from July 2022 - August 2023

RESULTS

Sample

Records for 454 patients with a physician-confirmed diagnosis of LGS were collected.

Adult Population

34%

...of the sample were >18 years old

Most Common Seizures

3/4

...of patients across all age groups had experienced a **drop seizure** in the last month

70%

...of patients are prescribed **RMs**

Treatment for **seizure-related complications** were the leading cause of hospitalisations in the last year, with 52% admitted for this reason

Daytime seizures were reported by physicians to have the greatest impact on patient QoL (32-49%)

47%

...of patients with LGS have **poor QoL** as reported by physicians

CONCLUSIONS

Rescue medications are prescribed to the majority of patients with LGS, yet drop and non-drop seizures persist across the age-groups and remain the primary reason for patients being admitted to hospital within the last 12 months.

Figure 1: Currently Prescribed ASMs, Top 10 (%)

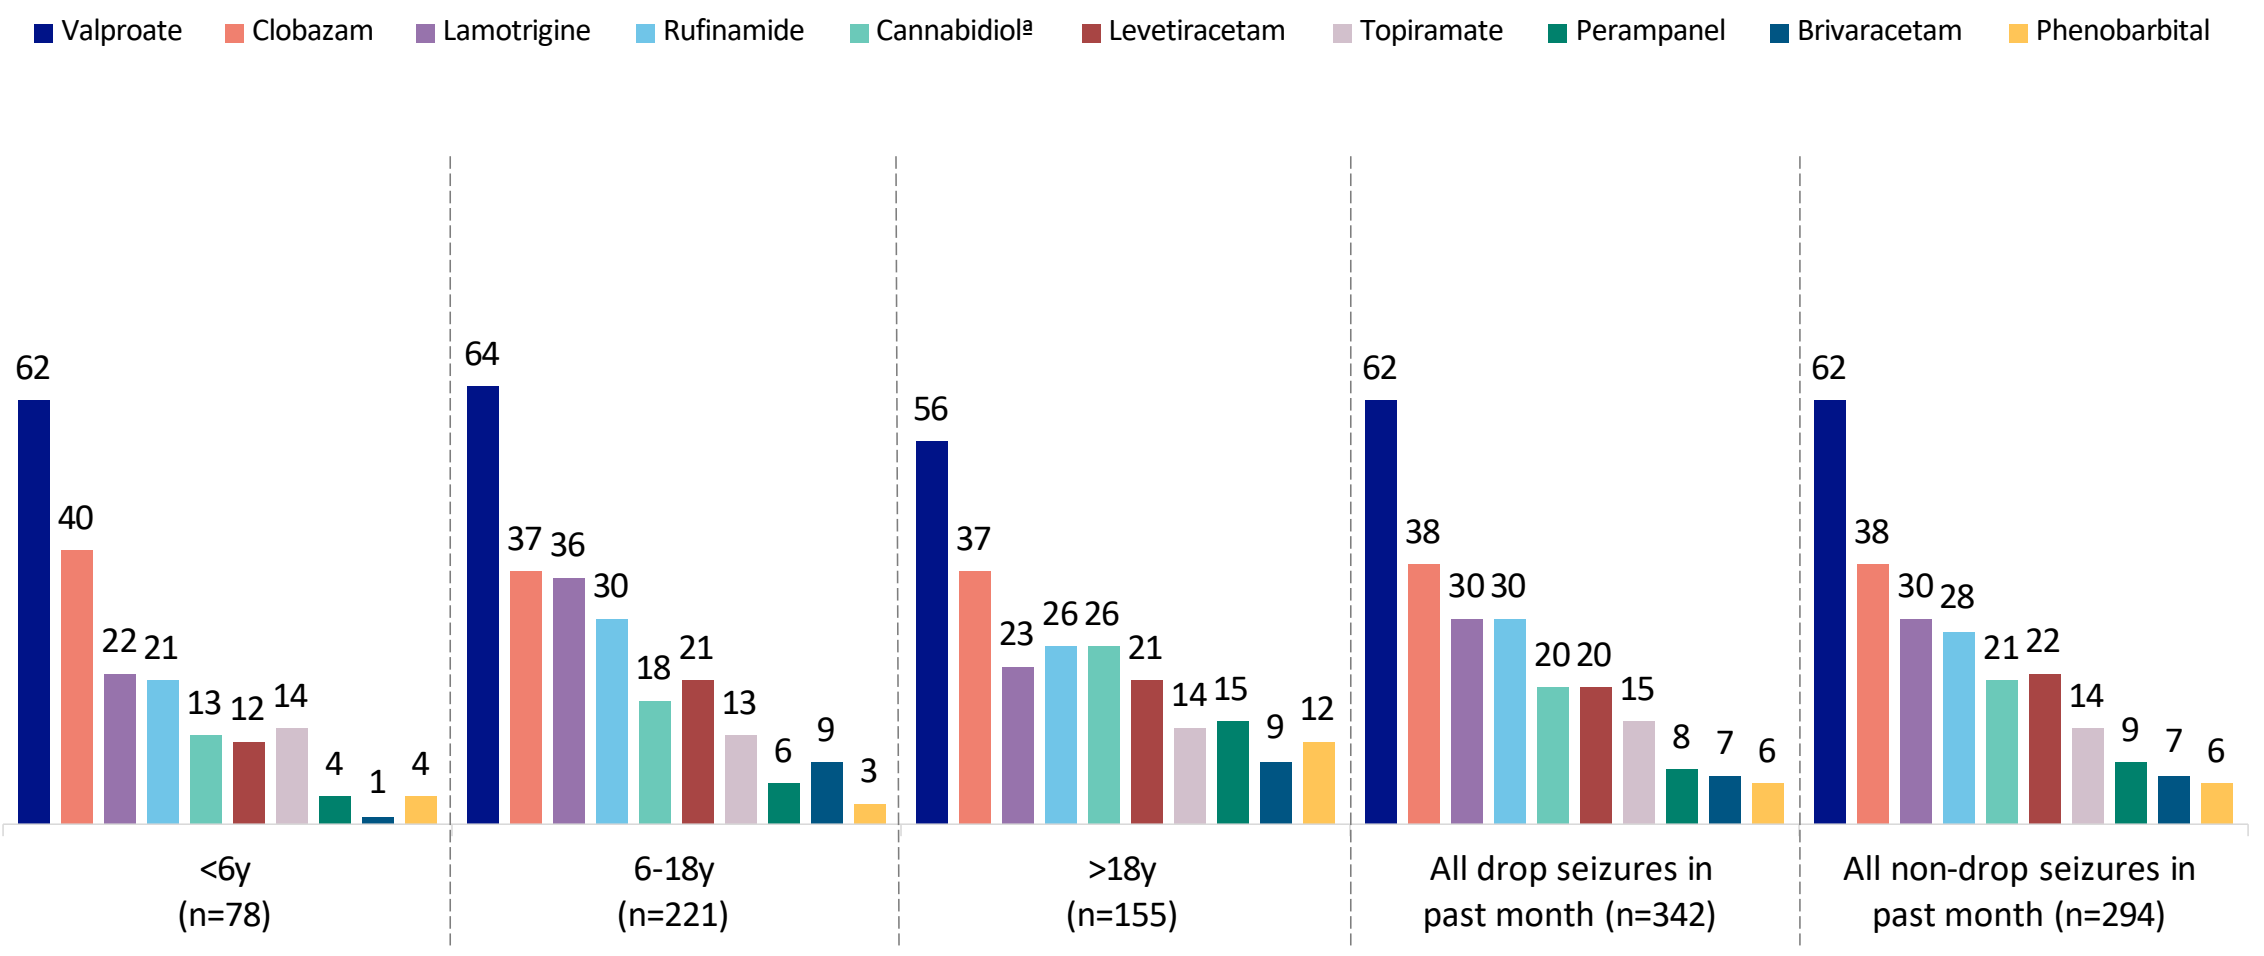

Valproate
Clobazam
Lamotrigine
Rufinamide
Cannabidiol[a]
Levetiracetam
Topiramate
Perampanel
Brivaracetam
Phenobarbital
62 40 22 21 13 12 14 4 1 4
64 37 36 30 18 21 13 6 9 3
56 37 23 26 26 21 14 15 9 12
62 38 30 30 20 20 15 8 7 6
62 38 30 28 21 22 14 9 7 6
<6y (n=78)
6-18y (n=221)
>18y (n=155)
All drop seizures in past month (n=342)
All non-drop seizures in past month (n=294)

ASM, anti-seizure medications; y, years; [a]Combined cannabidiol Epidiolex and cannabidiol other than Epidiolex.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Figure 2: Currently Prescribed Rescue Medications (%)

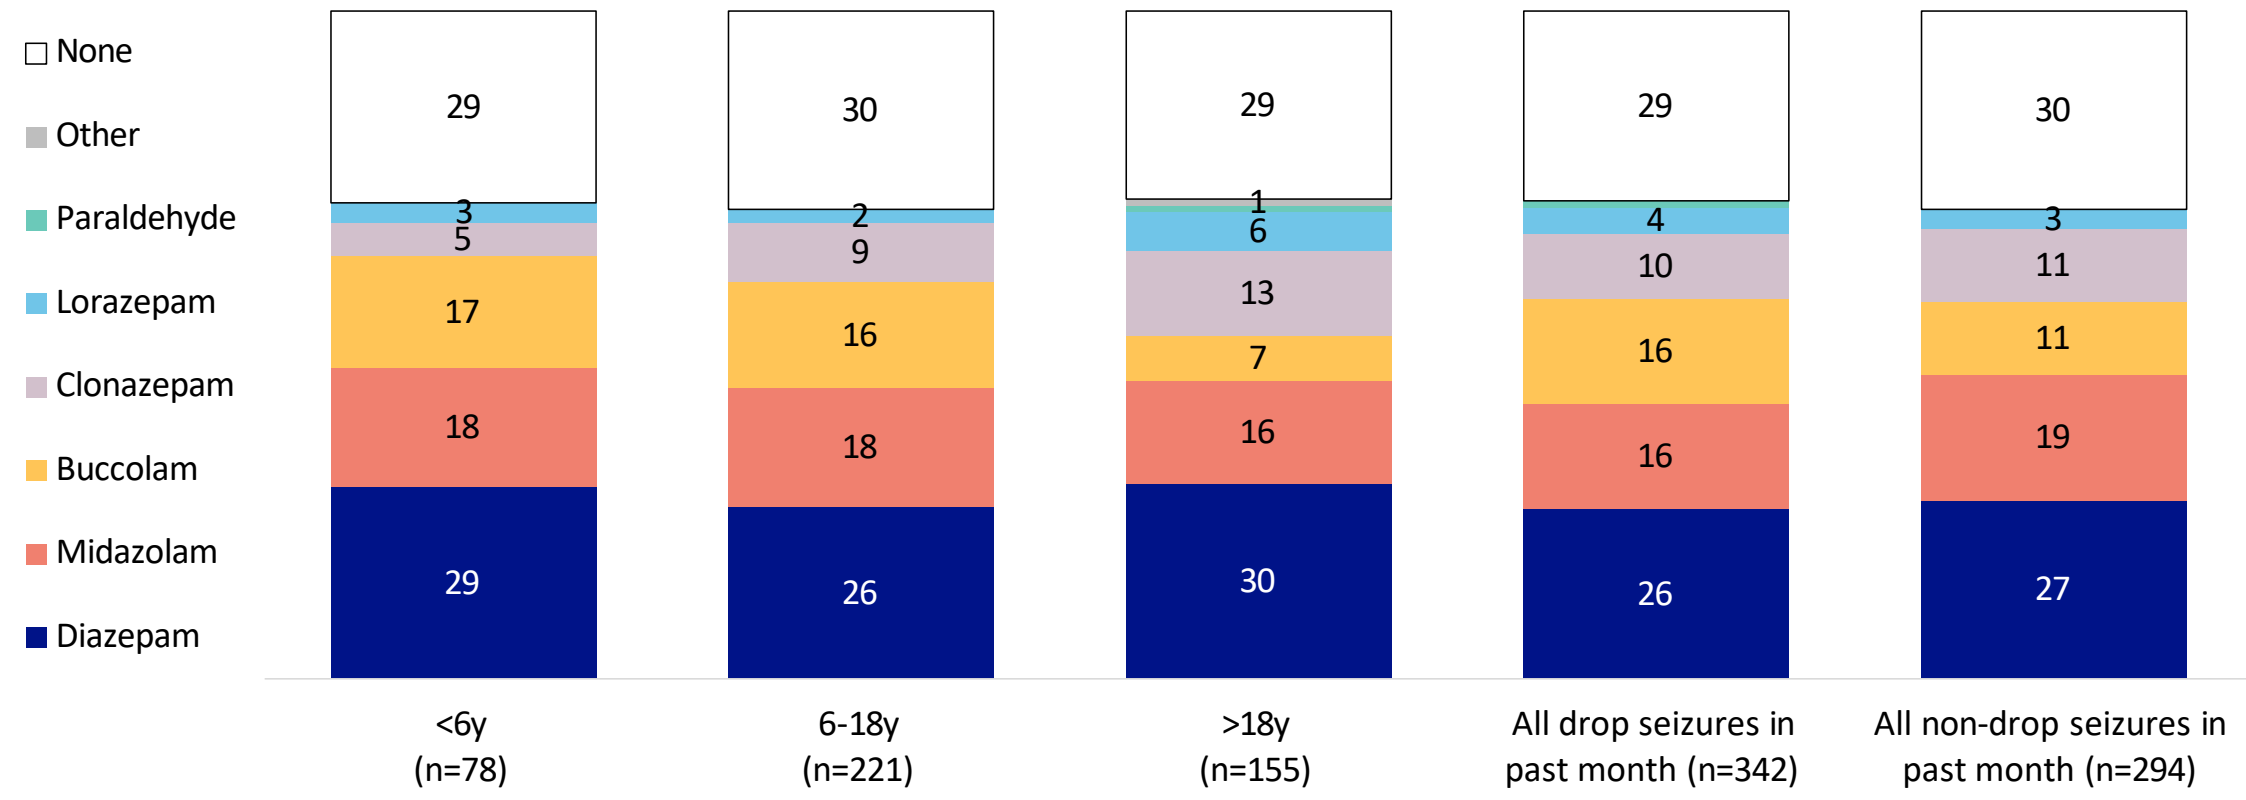

None
Other
Paraldehyde
Lorazepam
Clonazepam
Buccolam
Midazolam
Diazepam
29 3 5 17 18 29
30 2 9 16 18 26
29 1 6 13 7 16 30
29 4 10 16 16 26
30 3 11 11 19 27
<6y (n=78)
6-18y (n=221)
>18y (n=155)
All drop seizures in past month (n=342)
All non-drop seizures in past month (n=294)

RM, rescue medications; y, years.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Healthcare Resource Utilisation

HCRU records from the previous 12 months showed that 16% of patients had been hospitalised at least once, of whom:

- 52% had been admitted to treat a seizure-related complication;
- 92% had been admitted via the emergency room (ER);
- 19% had been admitted to the intensive care unit (ICU) **(Table 2).**

Patient QoL

Physicians were most likely to report patients with LGS QoL as poor **(Figure 3).** Day-time seizures had the greatest impact on patients' QoL, especially in those aged <6y and >18y **(Figure 4).**

Table 2: Healthcare Resource Utilisation

	Overall LGS (n=454)	<6y (n=78)	6-18y (n=221)	>18y (=155)	All drop seizures in past month (n=342)	Non-drop seizures in past month (n=294)
Hospitalised at least once in last 12 months (%)	16	24	16	12	17	17
Admitted via ER in last 12 months (%)	92	100	90	88	92	88
Admitted to the ICU in last 12 months (%)	19	28	18	12	18	19
Reasons for hospitalisations in last 12 months (%)						
To treat a seizure-related complication	52	61	52	41	52	53
Fall/fracture following seizure	31	28	38	24	34	28
Status epilepticus	27	44	28	6	24	28
Other	17	6	21	24	20	16
Related adverse event from ASM	6	6	3	12	6	7
Hospitalisations in last 12 months (Mean (SD))	0.3 (1.0)	0.5 (0.9)	0.3 (1.2)	0.2 (0.4)	0.3 (1.1)	0.3 (0.8)
ER admissions in last 12 months (Mean (SD))	1.0 (1.6)	2.0 (2.5)	0.9 (1.3)	0.5 (1.0)	1.1 (1.7)	1.1 (1.7)

SD, standard deviation; y, years; ER, emergency room; ICU, intensive care unit; LGS, Lennox-Gastaut Syndrome.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Figure 3: Physician-Rated Patient QoL (%)

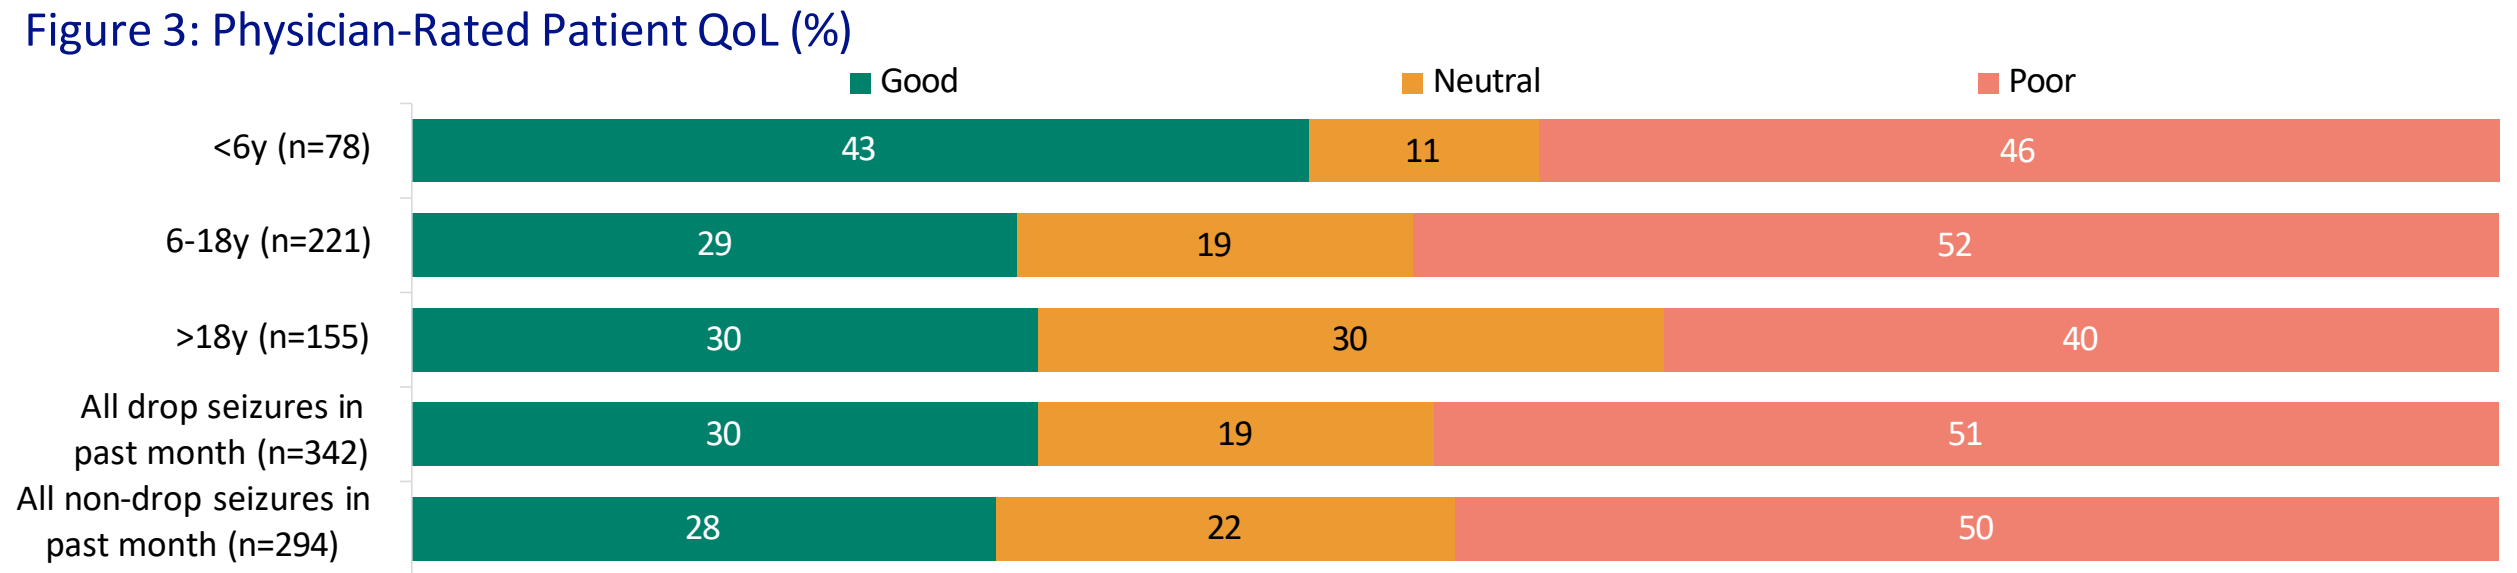

Good
Neutral
Poor
<6y (n=78) 43 11 46
6-18y (n=221) 29 19 52
>18y (n=155) 30 30 40
All drop seizures in past month (n=342) 30 19 51
All non-drop seizures in past month (n=294) 28 22 50

QoL, quality of life; y, years.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Figure 4: Greatest Impact on Patient QoL (%)

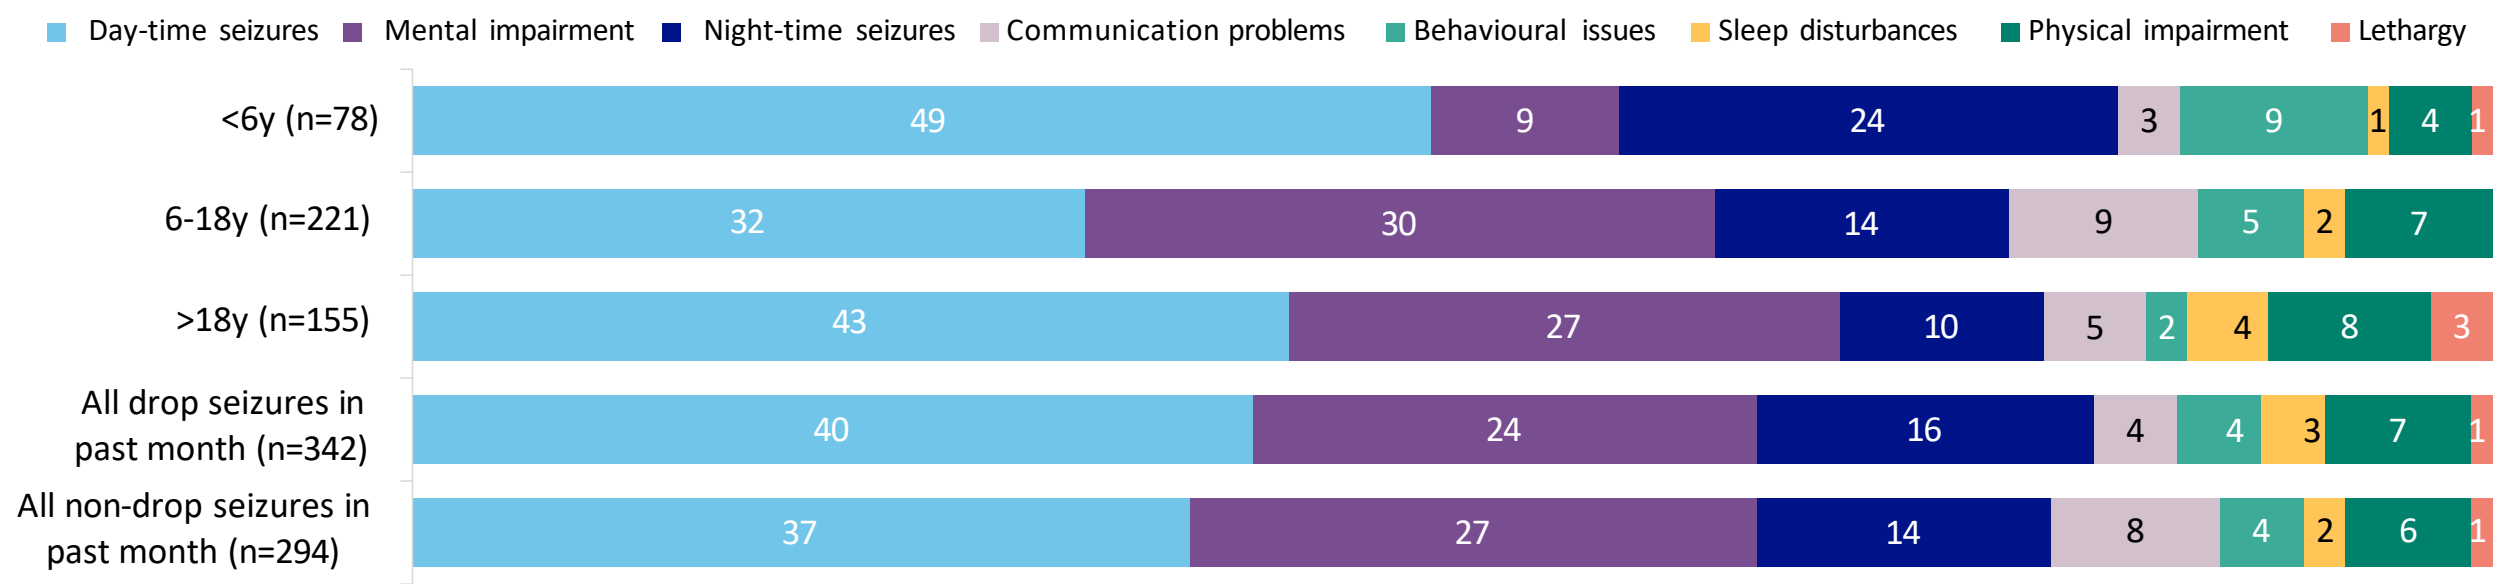

Day-time seizures
Mental impairment
Night-time seizures
Communication problems
Behavioural issues
Sleep disturbances
Physical impairment
Lethargy
<6y (n=78) 49 9 24 3 9 1 4 1
6-18y (n=221) 32 30 14 9 5 2 7
>18y (n=155) 43 27 10 5 2 4 8 3
All drop seizures in past month (n=342) 40 24 16 4 4 3 7 1
All non-drop seizures in past month (n=294) 37 27 14 8 4 2 6 1

QoL, quality of life; y, years.
For 'Drop seizures' & 'Non-drop seizures' group definitions, please see Supplementary Information.

Conclusions

- Despite receiving multiple treatment regimens, and 70% being prescribed RMs, patients with LGS continue to experience both drop and non-drop seizures into adulthood.
- Hospitalisations were reported in 16%, almost all of whom were admitted via ER and one-fifth to the ICU.
- Around half of patients' QoL is rated as "poor" by their physicians.
- Seizures remain both the most common reason for hospitalisations as well as having the greatest impact on patient QoL.
- Findings suggest a need for new ASMs & RMs to: better control seizures, reduce HCRU and improve patient QoL.

Limitations

- The Disease Specific Programme™ (DSP) is based on a pseudo-random sample. While minimal inclusion criteria governed the selection of participating physicians, participation was influenced by their willingness to complete the survey.
- Recall bias, a common limitation of surveys, might also have affected responses. Physicians had the ability to refer to patients' records, thus minimising the possibility of recall bias.

References:

1. *Trevathan, E, et al. 1997. Epilepsia, 38(12). 1283-8*
2. *Strzelczyk, A, at al. 2023. Orphanet J Rare. 18, 42*
3. *Gallop, K, et al. 2009. Seizure. 18(8). 554-8*
4. *Anderson, P, et al. 2023. Current Medical Research & Opinion. 39(12), 1707-15*
5. *Higgins, V., et al. 2016. Diabetes Metab Syndr Obes. 9. 371-380.*

Supplementary information:

Drop seizures: tonic, atonic (drop attacks), generalised tonic-clonic, secondary generalised tonic-clonic (focal to bilateral tonic-clonic) seizures.

Non-drop seizures: atypical absence, myoclonic, myoclonic-atonic, myoclonic-tonic-clonic, clonic, focal clonic/hemiclonic, convulsive status epilepticus, non-convulsive status epilepticus, epileptic spasm, focal onset aware (simple partial), focal onset impaired awareness (complex partial), hyperkinetic, non-motor onset, focal with observable motor signs, unidentified at the time.

Disclosures:

Data derived from Adelphi Real World (ARW) D+LGS DSP. UCB Pharma has subscribed to the DSP data. The authors thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors acknowledge Vincent Laporte, PhD (UCB Pharma, Brussels, Belgium) for managing the development of the poster and Nikisha Grant, MSc (Adelphi Real World, UK) and Becky Storm, BSc (Adelphi Real World, UK), for writing and editorial assistance, which was funded by UCB Pharma. Author contributions: J de Courcy designed the study; A Gillespie analysed and interpreted the data. All authors critically reviewed the poster and approved the final version for presentation. Author disclosures: A Strzelczyk reports personal fees and grants from Angelini Pharma, Desitin Arzneimittel, Eisai, Jazz/GW Pharmaceuticals companies, Marinus Pharma, Precisis, Takeda, UCB, UNEEG medical, and Zogenix. A Gil-Nagel received grants or honoraria as speaker or advisory board from Bial, Biocodex, Eisai, Stoke Therapeutics, GW Pharma, Esteve, UCB Pharma, Zogenix, and Arvelle Therapeutics. P Striano reports personal fees and grants from Angelini Pharma, Eisai, Jazz Pharmaceuticals Biomarin, UCB, Proveca, and Zogenix. R Chin has received consultancy fees from Eisai, GW Pharmaceuticals companies, and Zogenix, and has been a principal investigator for GW Research Ltd. I Leunikava and A Lothe are employees of UCB Pharma. A Gillespie is an employee of ARW. A Arzimanoglou received consultancy fees, funding for travel, and honoraria as speaker or advisory board from Eisai, GW Pharma, Takeda, UCB Pharma, and Zogenix, and has received research support from UCB Pharma and Caixa Foundation.

For a copy of this poster, use your smartphone to scan the QR code, download from the website below, visit the UCB medical affairs booth at the exhibit, or contact UCBCares®

Website: UCBposters.com/AES2024; Poster ID: 1.344

Phone: +1-844-599-CARE (2273)

Email: UCBCares@ucb.com

American Epilepsy Society Congress | Los Angeles, USA
December 6-10, 2024

Confidential - Not for Public Consumption or Distribution