Bimekizumab treatment resulted in improvements in MRI inflammatory and structural lesions in the sacroiliac joints of patients with axial spondyloarthritis: 52-week results and post hoc analyses from two phase 3 studies

Walter P. Maksymowych,[1] Sofia Ramiro,[2,3] Denis Poddubnyy,[4–6] Xenofon Baraliakos,[7] Robert G. Lambert,[8] Ute Massow,[9] Tom Vaux,[10] Chetan Prajapati,[10] Alexander Marten,[9] Natasha de Peyrecave,[11] Mikkel Østergaard[12,13]

Objective

To evaluate the impact of bimekizumab (BKZ) on MRI inflammatory and structural lesions in the sacroiliac joints (SIJ) of patients with axial spondyloarthritis (axSpA) to Week 52 in two phase 3 studies.

Background

- Inflammatory and structural lesions in the SIJ are key characteristics of axSpA, and can be visualized using MRI assessment.[1,2]
- BKZ, a monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A, has demonstrated consistent and sustained efficacy to 2 years in patients with non-radiographic (nr-) and radiographic (r-)axSpA, in the parallel phase 3 studies BE MOBILE 1 (NCT03928704) and BE MOBILE 2 (NCT03928743), respectively, and their combined open-label extension (OLE).[3–6]
- BKZ has also demonstrated improvements in MRI inflammation scores in the SIJ and spine of patients with axSpA to 1 year.[3]
- The impact of dual inhibition of IL-17A and IL-17F on structural lesions in patients with axSpA has not yet been demonstrated.

Methods

- BE MOBILE 1 and BE MOBILE 2 study designs have been reported previously.[7] From Weeks 16 to 52, all patients received subcutaneous BKZ 160 mg Q4W.
- Spondyloarthritis Research Consortium of Canada (SPARCC) SIJ inflammation scores and SPARCC SIJ Structural Score (SSS; erosions, backfill, fat lesions, ankylosis) were assessed at baseline, Week 16, and Week 52 in patients in the MRI sub-studies.
- MRIs were read centrally by two independent expert readers, with an adjudicator in cases of disagreement. Inflammatory and structural lesions were assessed independently by different readers, hence the number of MRIs successfully scored could differ. All readers were blinded to timepoint and any clinical data; structural lesions were analyzed post hoc.
- We report observed case data for patients across the full disease spectrum of axSpA with valid MRI assessments at all 3 timepoints (baseline, Week 16, and Week 52).

Results

Patients

- Overall, 60% (152/254) of patients with nr-axSpA and 42% (139/332) of patients with r-axSpA were enrolled in the MRI sub-studies.
- Of these, 76% (115/152) and 78% (109/139) of patients had valid SPARCC SIJ inflammation assessments at all 3 timepoints, respectively, and 84% (128/152) and 83% (116/139) of patients had valid SPARCC SSS (structural lesions) assessments at all 3 timepoints.
- Baseline characteristics in the MRI sub-studies were largely comparable between treatment arms and reflected those of the overall patient population (**Table 1**).

SPARCC SIJ Inflammation Scores

- Across the full disease spectrum of axSpA, BKZ demonstrated substantially larger reductions in SPARCC SIJ inflammation scores compared with placebo (PBO) at Week 16, with marked decreases from baseline (**Figure 1**).
 - Patients continuing or switching to BKZ showed further decreases at Week 52 (**Figure 1**).
- At Week 16, a larger proportion of BKZ-randomized patients with SPARCC SIJ inflammation score ≥2 at baseline achieved MRI SIJ remission (SPARCC SIJ inflammation score <2) compared with PBO-randomized patients (**Figure 2**).
 - At Week 52, at least 75% continuous BKZ patients achieved MRI SIJ remission (**Figure 2**).

SPARCC SSS (Structural Lesions)

- At Week 16, across the full disease spectrum of axSpA, BKZ demonstrated substantially larger reductions in erosion scores compared with placebo. At Week 52, patients continuing or switching to BKZ showed further decreases in erosion scores (**Figure 3**).
- At Week 16, BKZ showed larger increases in backfill and fat lesion scores versus PBO in patients with nr-axSpA and r-axSpA. At Week 52, these backfill and fat lesion scores continued to increase, or were largely sustained, in patients continuing or switching to BKZ (**Figure 3**).
- No or minimal changes in ankylosis score were observed following treatment with BKZ in patients with nr-axSpA and r-axSpA, respectively, at Week 16 and Week 52 (**Figure 3**).

Reliability

- Smallest detectable change (SDC) and intra-class correlation (ICC) reliability data for inflammation and structural lesion scores are provided in **Figures 1 and 3**.

Conclusions

Dual inhibition of IL-17A and IL-17F with bimekizumab had a substantial impact on inflammatory and structural lesions at Week 16; improvements largely continued or were sustained at Week 52 across patients with nr-axSpA and r-axSpA.

Summary

Inflammation and **structural lesions** in the sacroiliac joints are key characteristics of axial spondyloarthritis

Across the **full disease spectrum of axial spondyloarthritis,** dual inhibition of **IL-17A and IL-17F** with **bimekizumab** demonstrated:

Rapid and substantial reductions in SIJ inflammation, as measured by SPARCC SIJ score

Over half of patients achieved **MRI SIJ remission** at Week 16 and Week 52, defined as SPARCC SIJ inflammation score <2

Substantial decreases in **erosions**, and increases in **backfill** and **fat lesions** compared with baseline, potentially indicating **tissue response** after only 16 weeks of treatment

Table 1 Baseline characteristics: MRI sub-studies

Mean (SD), unless otherwise stated	nr-axSpA (BE MOBILE 1)			r-axSpA (BE MOBILE 2)		
	PBO n=70[a]	BKZ 160 mg Q4W n=82[a]	Overall study population N=254[b]	PBO n=48[a]	BKZ 160 mg Q4W n=91[a]	Overall study population N=332[b]
Age, years	40.0 (12.5)	38.9 (11.6)	39.4 (11.5)	39.7 (12.9)	40.1 (12.2)	40.4 (12.3)
Sex, male, n (%)	31 (44.3)	50 (61.0)	138 (54.3)	31 (64.6)	68 (74.7)	240 (72.3)
HLA-B27 positive, n (%)	47 (67.1)	65 (79.3)	197 (77.6)	39 (81.3)	80 (87.9)	284 (85.5)
Symptom duration, years	8.7 (9.2)	9.0 (8.5)	9.0 (8.8)	12.7 (9.0)	13.7 (11.0)	13.5 (10.3)
ASDAS	3.6 (0.7)	3.6 (0.7)	3.7 (0.7)	3.6 (0.8)	3.9 (0.8)	3.7 (0.8)[c]
BASDAI	6.4 (1.3)	6.9 (1.2)	6.8 (1.3)	6.4 (1.4)	6.6 (1.4)	6.5 (1.3)
hs-CRP, mg/L, geometric mean (geometric CV, %)	4.9 (207.5)	3.8 (276.1)	4.8 (261.8)	6.1 (195.1)	7.6 (244.7)	6.6 (246.3)
Prior TNFi exposure, n (%)	8 (11.4)	5 (6.1)	27 (10.6)	7 (14.6)	13 (14.3)	54 (16.3)
Patients in MRI sub-studies with assessments at all 3 timepoints						
SPARCC SIJ[d]	8.7 (11.6)	8.6 (10.3)	-	3.9 (6.2)	6.0 (9.0)	-
SPARCC SIJ ≥2,[d] n (%)	32 (62.7)	39 (60.9)	-	18 (46.2)	36 (51.4)	-
SPARCC SSS (erosions)[e]	2.7 (4.0)	3.8 (5.3)	-	2.9 (3.4)	3.6 (4.9)	-
SPARCC SSS (backfill)[e]	0.6 (1.3)	0.4 (1.2)	-	0.8 (1.7)	1.8 (3.4)	-
SPARCC SSS (fat lesions)[e]	1.1 (2.9)	1.0 (2.6)	-	3.4 (6.6)	5.6 (7.9)	-
SPARCC SSS (ankylosis)[e]	0.0 (0.1)	0.1 (0.2)	-	5.0 (7.7)	5.9 (8.2)	-

Randomized set. **[a]** Includes only patients in the MRI sub-study. **[b]** All patients in the overall study pooled, regardless of treatment arm. **[c]** n=331. **[d]** In patients in the MRI sub-studies with valid SPARCC SIJ inflammation assessments at baseline, Week 16, and Week 52: nr-axSpA: PBO: n=51, BKZ: n=64; r-axSpA: PBO: n=39, BKZ: n=70. **[e]** In patients in the MRI sub-studies with valid SPARCC SSS assessments at baseline, Week 16, and Week 52: nr-axSpA: PBO: n=59; BKZ: n=69, r-axSpA: PBO: n=41; BKZ: n=75.

Figure 1 SPARCC SIJ inflammation scores up to Week 52 (OC)

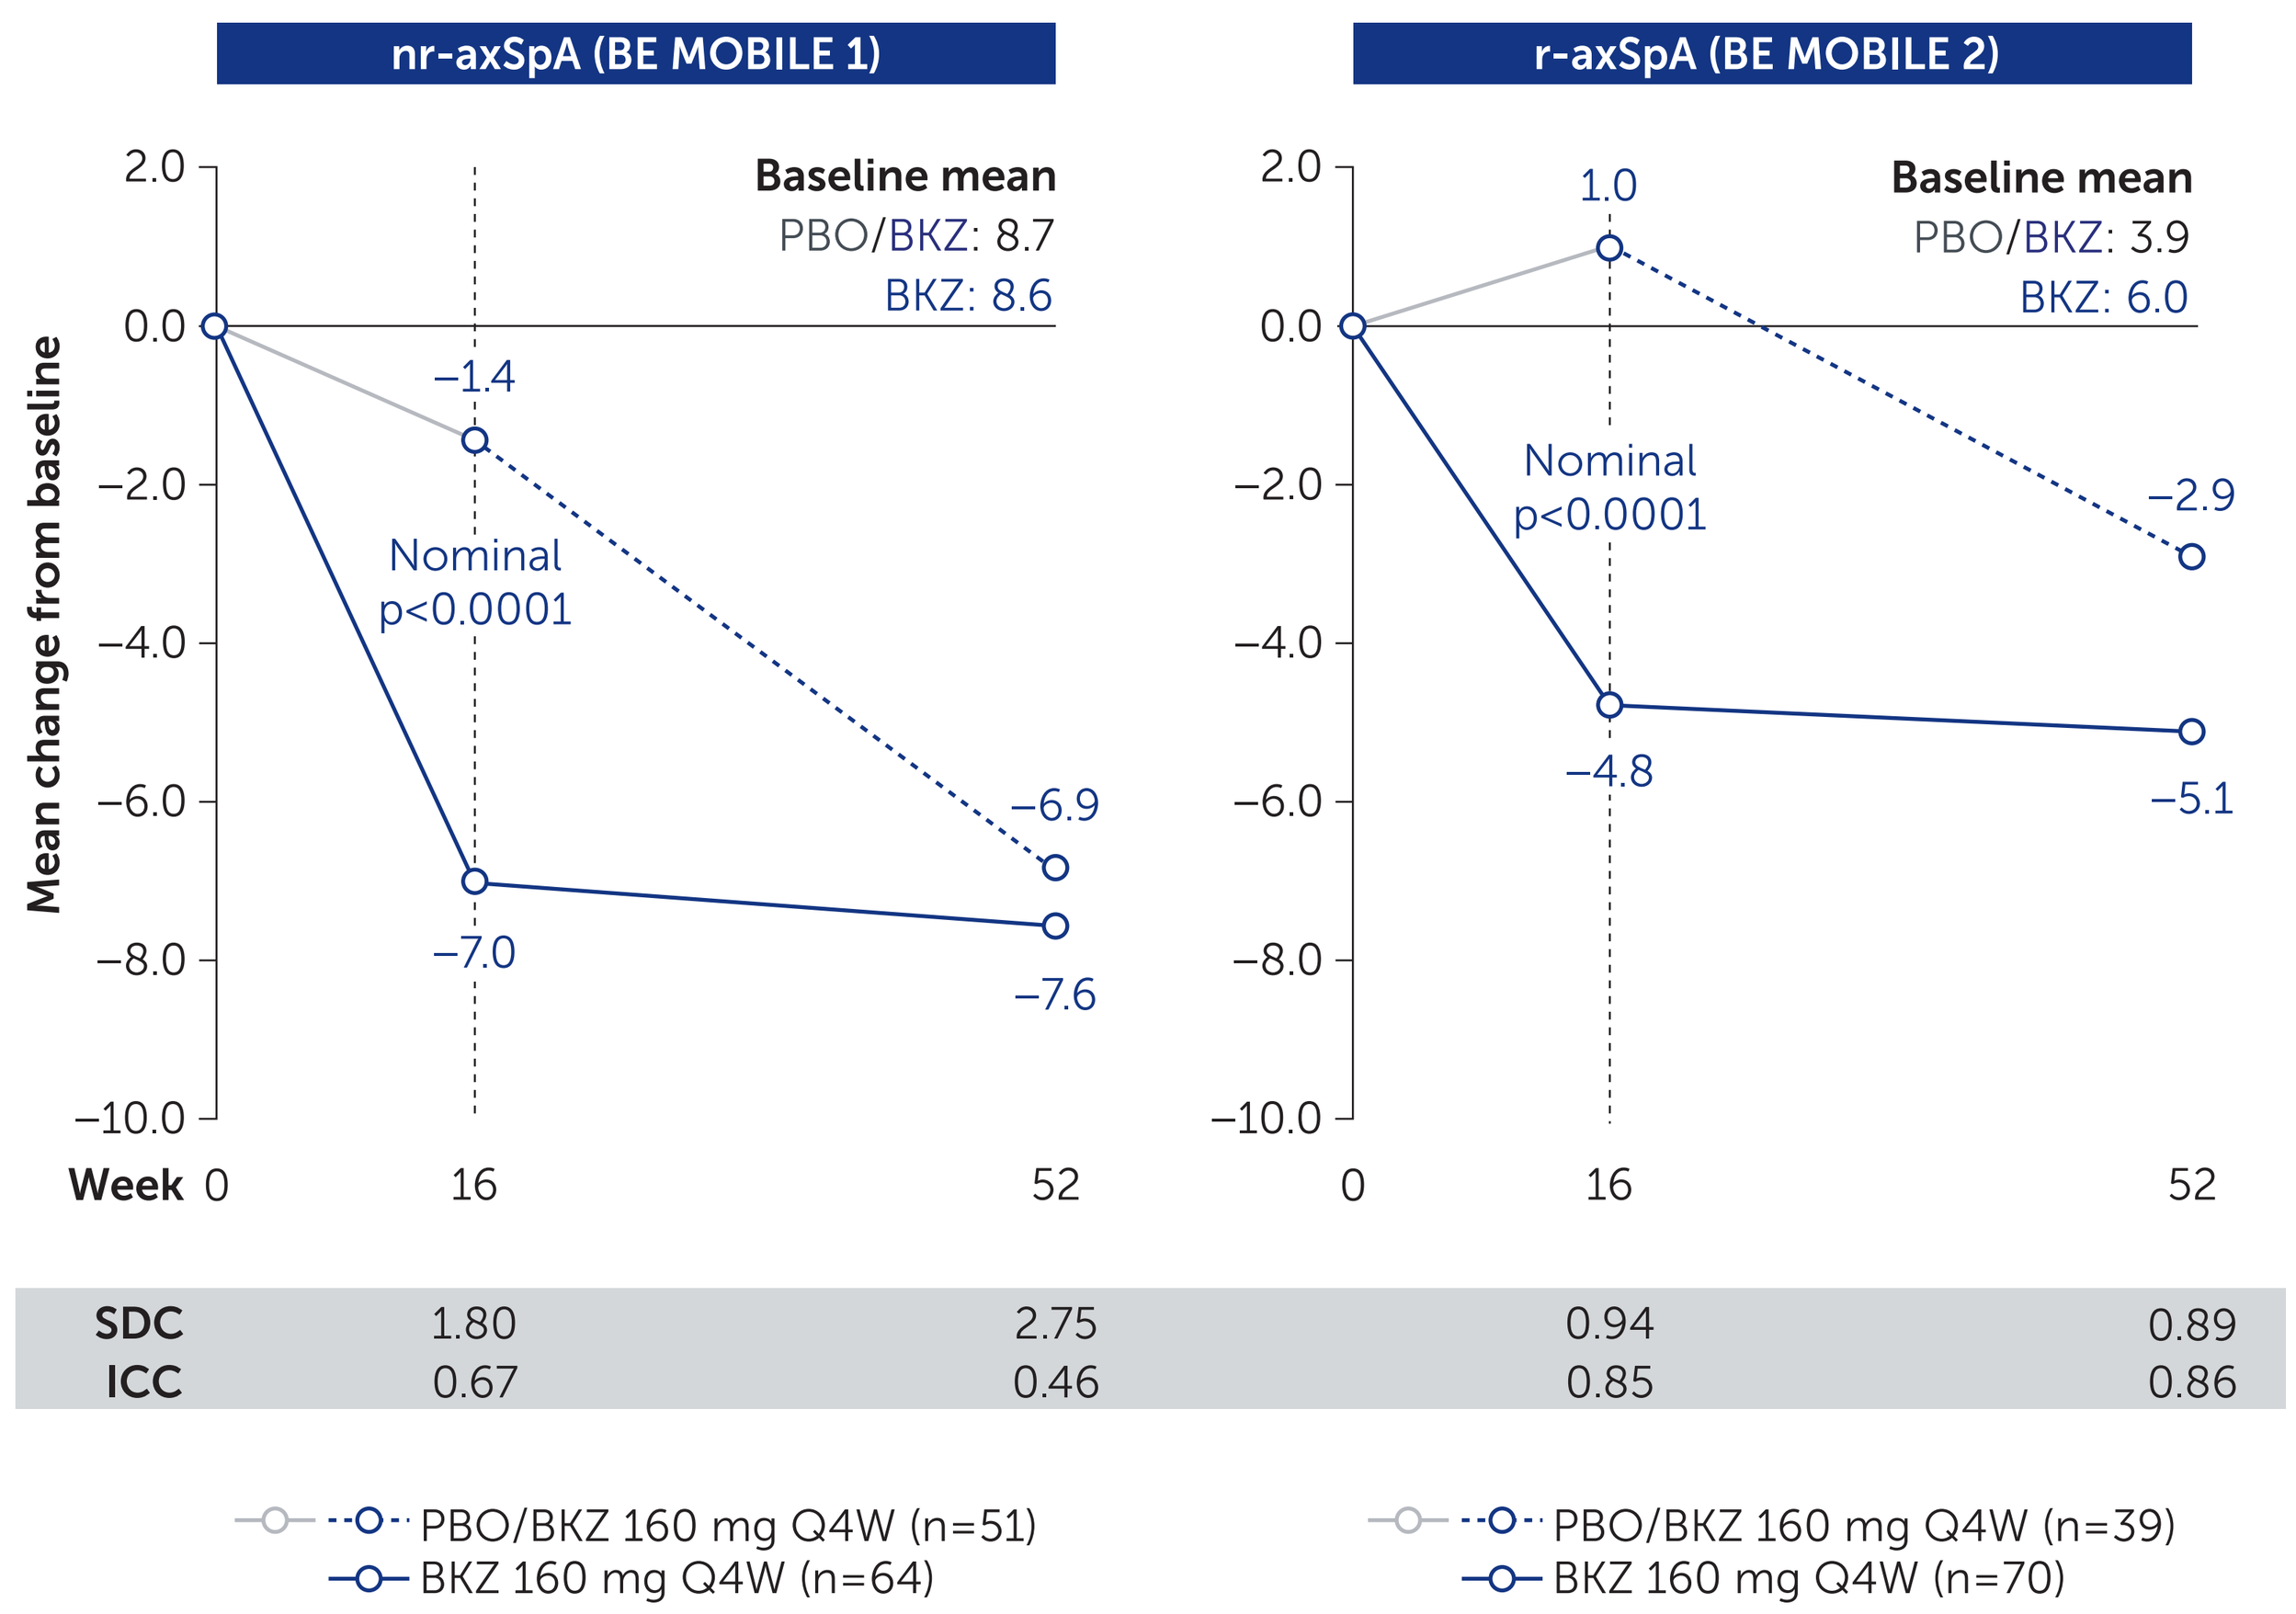

nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
Mean change from baseline
Baseline mean
PBO/BKZ: 8.7
BKZ: 8.6
PBO/BKZ: 3.9
BKZ: 6.0
Nominal p<0.0001
−1.4
−7.0
−6.9
−7.6
1.0
−4.8
−2.9
−5.1
Week 0 16 52
SDC 1.80 2.75 0.94 0.89
ICC 0.67 0.46 0.85 0.86
PBO/BKZ 160 mg Q4W (n=51)
BKZ 160 mg Q4W (n=64)
PBO/BKZ 160 mg Q4W (n=39)
BKZ 160 mg Q4W (n=70)

Randomized set. Includes only patients in the MRI sub-studies with valid SPARCC SIJ inflammation assessments at baseline, Week 16, and Week 52. SPARCC SIJ inflammation scores range from 0–72, with lower scores indicating less inflammation. SDC was calculated as 1.96 × standard error × √2, where standard error is the difference of the change scores between the two readers (per patient). The ICC assessed the inter-reader reliability using the two-way random effects mixed model for the absolute agreement between single scores from two independent readers. SDC and ICC are calculated based on change from baseline to the week displayed (i.e. baseline to Week 16, and baseline to Week 52, for the respective timepoints shown).

Figure 2 Achievement of MRI SIJ remission (OC)

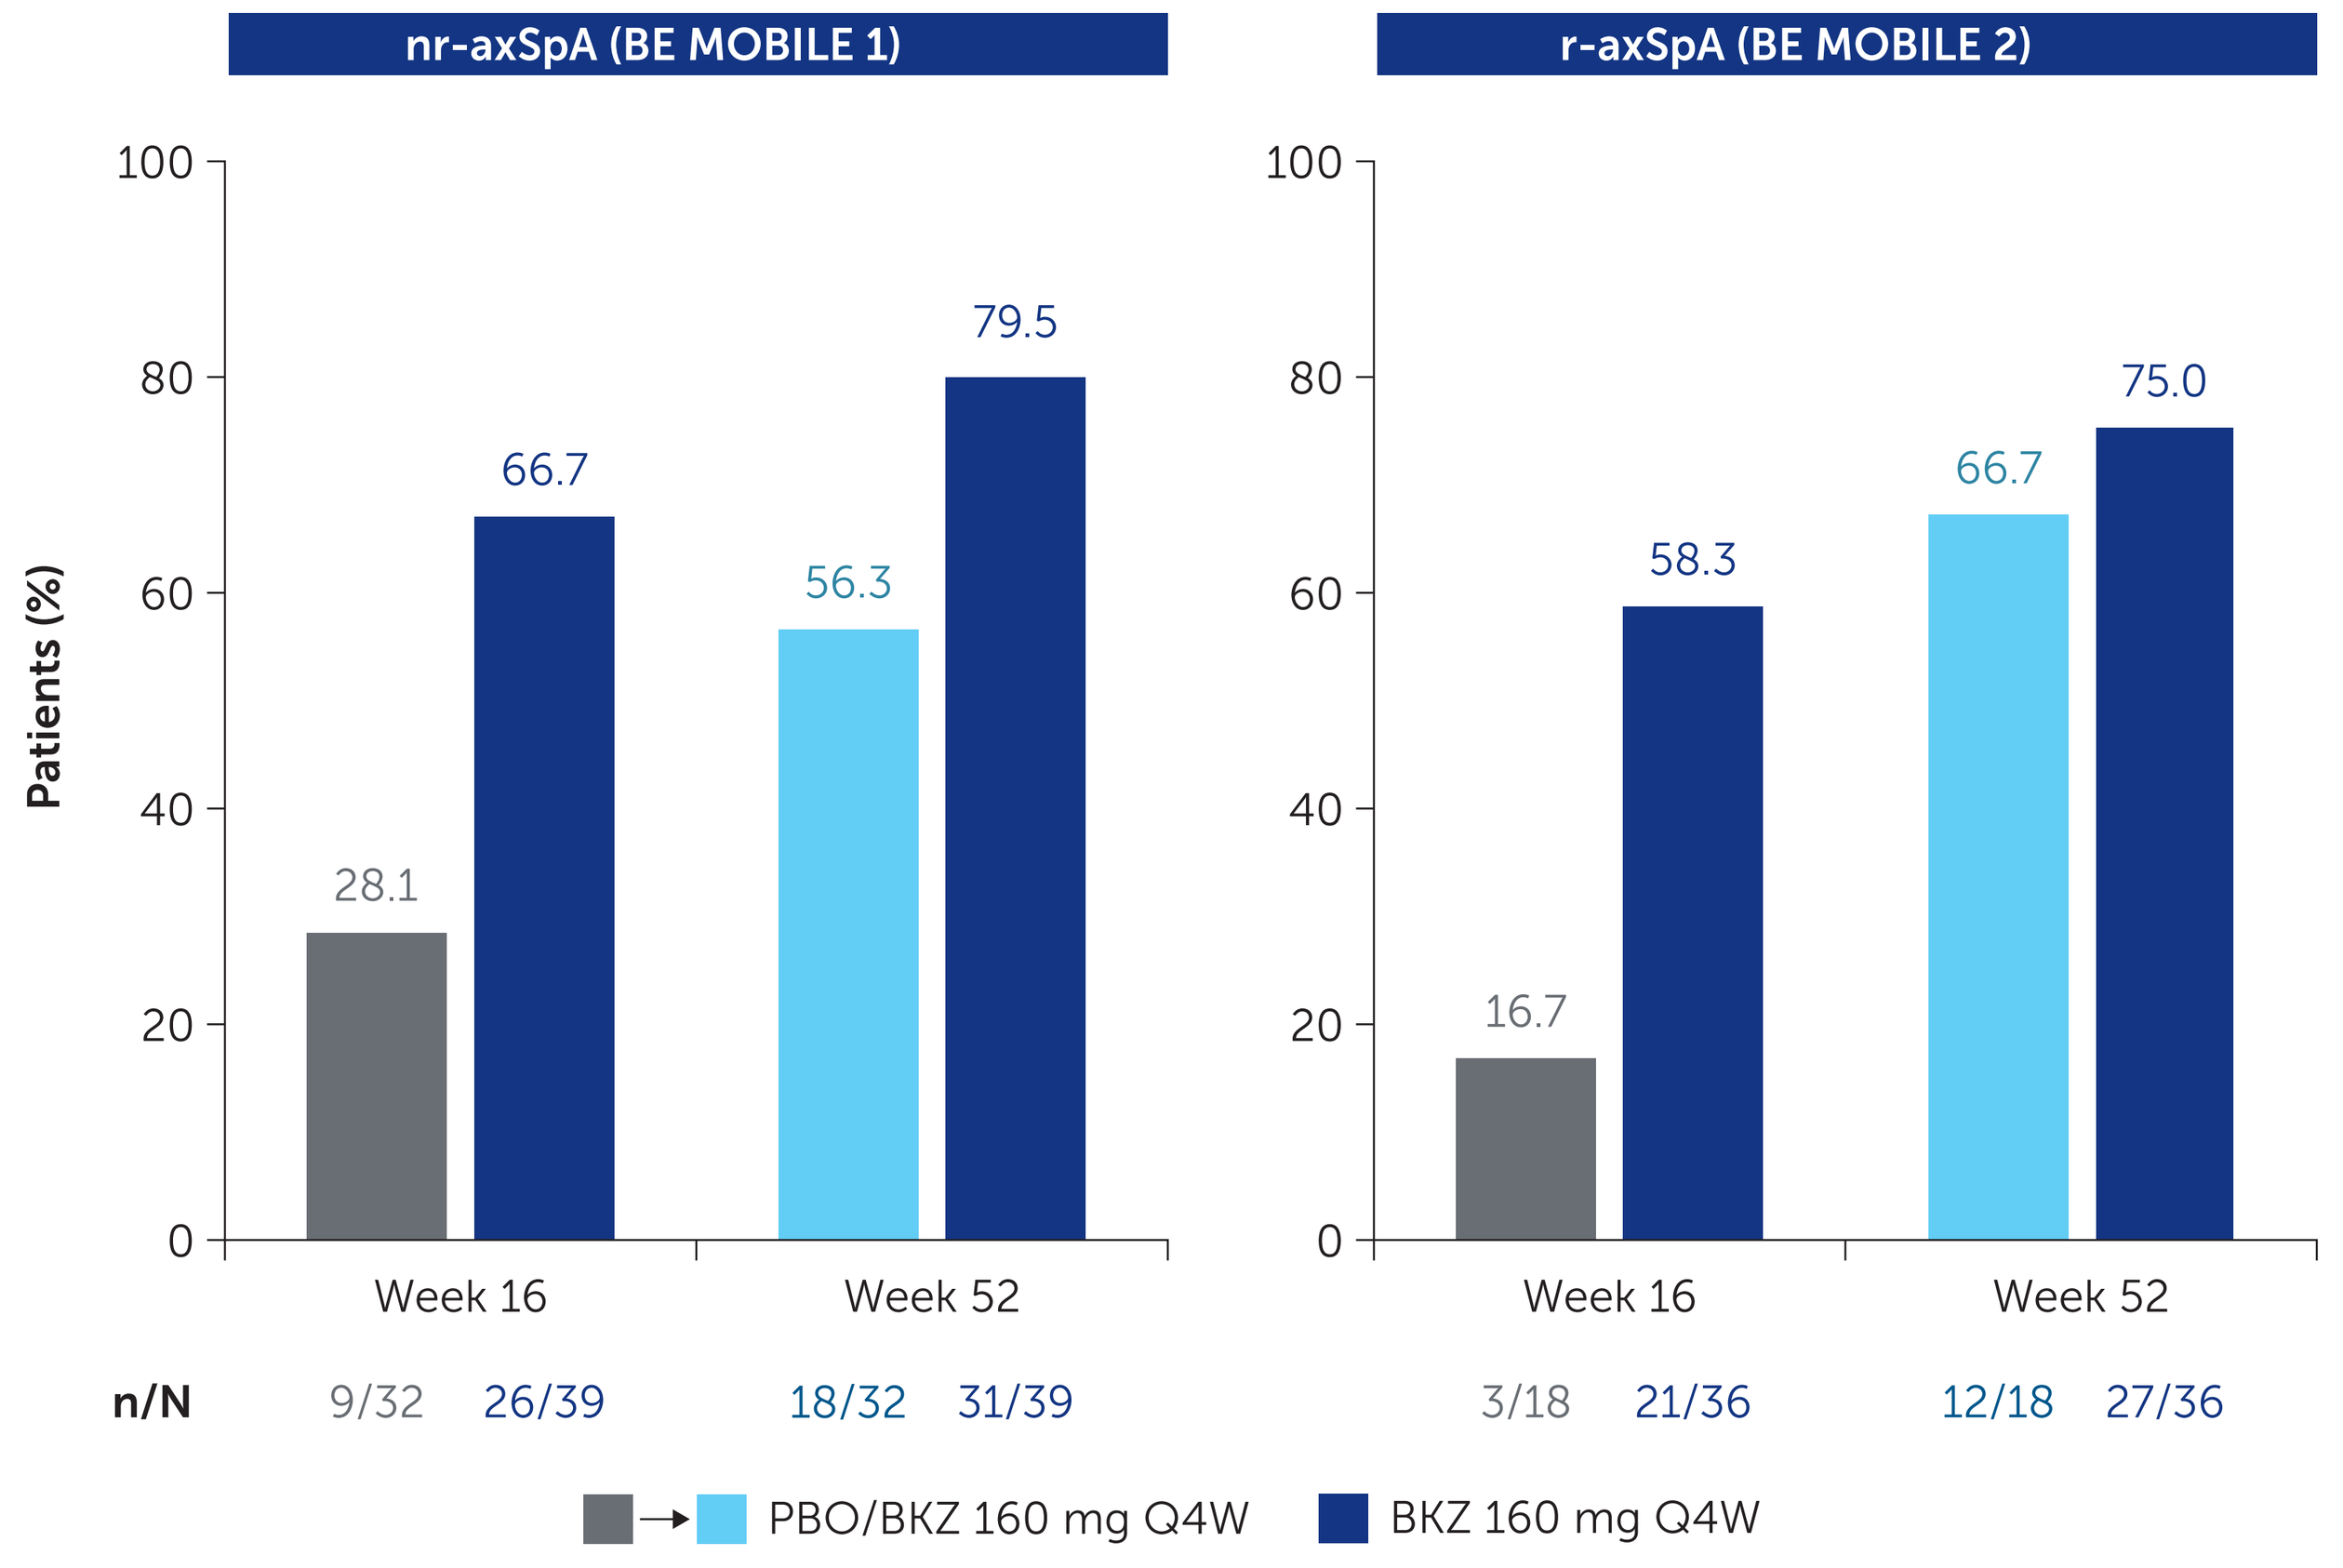

nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
Patients (%)
28.1
66.7
56.3
79.5
16.7
58.3
66.7
75.0
Week 16
Week 52
n/N 9/32 26/39 18/32 31/39 3/18 21/36 12/18 27/36
PBO/BKZ 160 mg Q4W
BKZ 160 mg Q4W

Randomized set; patients had existing inflammation at baseline (SPARCC SIJ inflammation score ≥2). Includes only patients in the MRI sub-studies with valid SPARCC SIJ inflammation assessments at baseline, Week 16, and Week 52.

Figure 3 Change from baseline in SPARCC SSS up to Week 52 (OC)

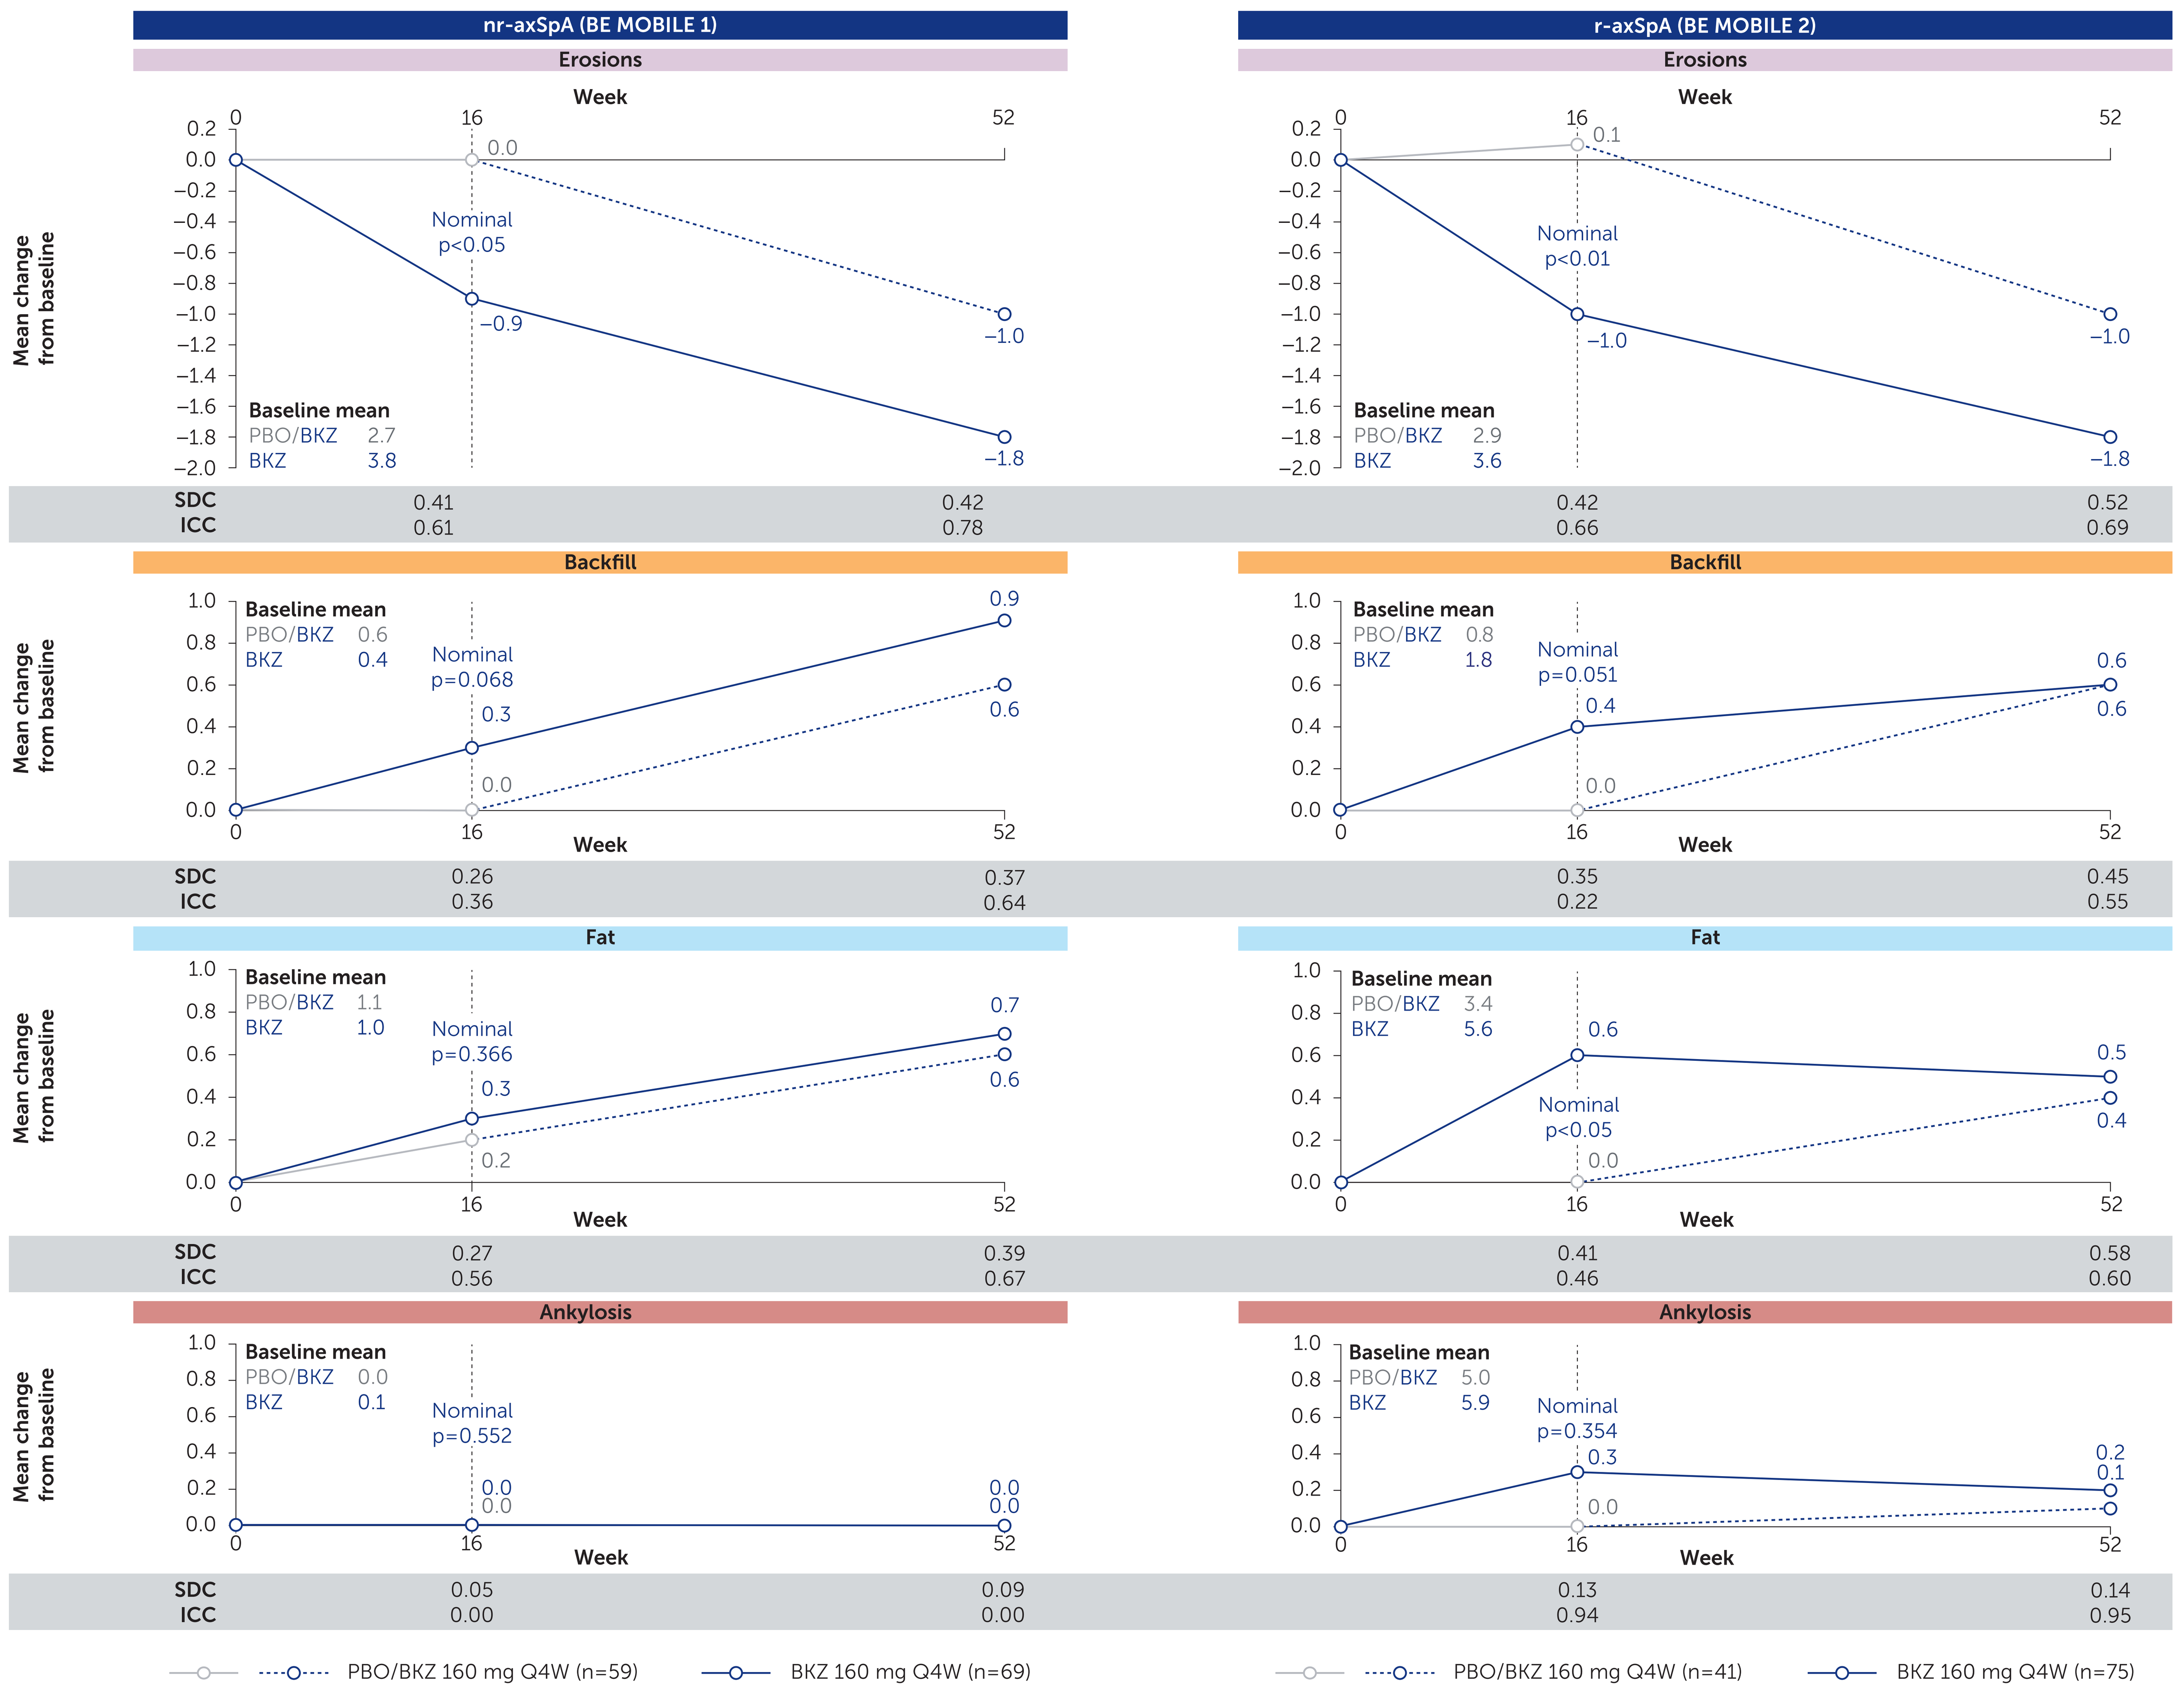

nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
Erosions
Backfill
Fat
Ankylosis
Mean change from baseline
Week
Baseline mean
PBO/BKZ 2.7
BKZ 3.8
Nominal p<0.05
0.0
−0.9
−1.0
−1.8
SDC 0.41 0.42
ICC 0.61 0.78
PBO/BKZ 2.9
BKZ 3.6
Nominal p<0.01
0.1
−1.0
−1.0
−1.8
SDC 0.42 0.52
ICC 0.66 0.69
PBO/BKZ 0.6
BKZ 0.4
Nominal p=0.068
0.3
0.0
0.9
0.6
SDC 0.26 0.37
ICC 0.36 0.64
PBO/BKZ 0.8
BKZ 1.8
Nominal p=0.051
0.4
0.0
0.6
0.6
SDC 0.35 0.45
ICC 0.22 0.55
PBO/BKZ 1.1
BKZ 1.0
Nominal p=0.366
0.3
0.2
0.7
0.6
SDC 0.27 0.39
ICC 0.56 0.67
PBO/BKZ 3.4
BKZ 5.6
0.6
Nominal p<0.05
0.0
0.5
0.4
SDC 0.41 0.58
ICC 0.46 0.60
PBO/BKZ 0.0
BKZ 0.1
Nominal p=0.552
0.0
0.0
0.0
0.0
SDC 0.05 0.09
ICC 0.00 0.00
PBO/BKZ 5.0
BKZ 5.9
Nominal p=0.354
0.3
0.0
0.2
0.1
SDC 0.13 0.14
ICC 0.94 0.95
PBO/BKZ 160 mg Q4W (n=59)
BKZ 160 mg Q4W (n=69)
PBO/BKZ 160 mg Q4W (n=41)
BKZ 160 mg Q4W (n=75)

Randomized set. Includes only patients in the MRI sub-studies with valid SPARCC SSS assessments at baseline, Week 16 and Week 52. SDC was calculated as 1.96 × standard error × √2, where standard error is the difference of the change scores between the two readers (per patient). The ICC assessed the interrater reliability using the two-way random effects mixed model for the absolute agreement between single scores from two independent readers. SDC and ICC are calculated from baseline to the week displayed (i.e. baseline to Week 16, and baseline to Week 52, for the respective timepoints shown).

ASDAS: Axial Spondyloarthritis Disease Activity Score; **axSpA:** axial spondyloarthritis; **BASDAI:** Bath Ankylosing Spondylitis Disease Activity Index; **BKZ:** bimekizumab; **BL:** baseline; **CV:** coefficient of variation; **hs-CRP:** high-sensitivity C-reactive protein; **ICC:** intra-class correlation; **Ig:** immunoglobulin; **IL:** interleukin; **MRI:** magnetic resonance imaging; **nr-axSpA:** non-radiographic axSpA; **OC:** observed case; **OLE:** open-label extension; **PBO:** placebo; **Q4W:** every 4 weeks; **r-axSpA:** radiographic axSpA; **SD:** standard deviation; **SDC:** smallest detectable change; **SIJ:** sacroiliac joints; **SPARCC:** Spondyloarthritis Research Consortium of Canada; **SSS:** SIJ Structural Score; **TNFi:** tumor necrosis factor inhibitor.

[1]University of Alberta, Department of Medicine, Edmonton, AB, Canada; [2]Leiden University Medical Center, Department of Rheumatology, Leiden, The Netherlands; [3]Zuyderland Medical Center, Heerlen, The Netherlands; [4]University Health Network and University of Toronto, Division of Rheumatology, Department of Medicine, Toronto, ON, Canada; [5]Charité – Universitätsmedizin Berlin, Department of Gastroenterology, Infectious Diseases and Rheumatology, Berlin, Germany; [6]German Rheumatism Research Centre, Department of Epidemiology, Berlin, Germany; [7]Ruhr-University Bochum, Rheumazentrum Ruhrgebiet Herne, Germany; [8]University of Alberta, Department of Radiology & Diagnostic Imaging, Edmonton, AB, Canada; [9]UCB, Monheim am Rhein, Germany; [10]UCB, Slough, United Kingdom; [11]UCB, Brussels, Belgium; [12]University of Copenhagen, Department of Clinical Medicine, Copenhagen, Denmark; [13]Copenhagen Center for Arthritis Research, Center for Rheumatology, Rigshospitalet, Glostrup, Denmark.

References: [1]Navarro-Compán V. Ann Rheum Dis 2021;80:1511–21; [2]Maksymowych WP. Arthritis Rheum 2009;60:93–102; [3]Baraliakos X. Ann Rheum Dis 2024;83:199–213; [4]Baraliakos X. Presented at EULAR 2024. POS0806; [5]Boel A. Ann Rheum Dis 2019;78:1545–9; [6]van der Heijde D. Ann Rheum Dis 2024;83:547–9; [7]van der Heijde D. Ann Rheum Dis 2023;82(4):515–26.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **WPM**, **SR**, **DP**, **XB**, **RGL**, **UM**, **TV**, **CP**, **AM**, **NdP**, **MØ**; Drafting of the publication, or reviewing it critically for important intellectual content: **WPM**, **SR**, **DP**, **XB**, **RGL**, **UM**, **TV**, **CP**, **AM**, **NdP**, **MØ**; Final approval of the publication: **WPM**, **SR**, **DP**, **XB**, **RGL**, **UM**, **TV**, **CP**, **AM**, **NdP**, **MØ**. **Author Disclosures: WPM:** Honoraria/consulting fees from AbbVie, BMS, Boehringer Ingelheim, Celgene, Eli Lilly, Galapagos, Janssen, Novartis, Pfizer, and UCB; research grants from AbbVie, Galapagos, Pfizer, and UCB; educational grants from AbbVie, Janssen, Novartis, and Pfizer; Chief Medical Officer for CARE Arthritis. **SR:** Grants from AbbVie, Galapagos/Alfasigma, MSD, Novartis, Pfizer, and UCB; consulting fees from AbbVie, Eli Lilly, Galapagos/Alfasigma, Janssen, Novartis, Pfizer, Sanofi, and UCB. **DP:** Speaker for AbbVie, BMS, Eli Lilly, MSD, Novartis, Pfizer, and UCB; consultant for AbbVie, Biocad, Eli Lilly, Gilead, GSK, MSD, MoonLake Immunotherapeutics, Novartis, Pfizer, Samsung Bioepis, and UCB; grant/research support from AbbVie, Eli Lilly, MSD, Novartis, and Pfizer. **XB:** Speakers bureau from AbbVie, BMS, Chugai, Eli Lilly, Galapagos, MSD, Novartis, Pfizer, and UCB; paid instructor for AbbVie, BMS, Chugai, Eli Lilly, Galapagos, MSD, Novartis, Pfizer, and UCB; consultant for AbbVie, BMS, Chugai, Eli Lilly, Galapagos, Gilead, Novartis, Pfizer, and UCB; grant/research support from Abbvie, Celltrion, Janssen, and Novartis. **RGL:** Consultant for CARE Arthritis and Image Analysis Group. **UM, AM, NdP:** Employees of UCB. **TV:** Employee and shareholder of UCB. **CP:** Contractor for UCB and employee of Veramed. **MØ:** Research grants from Abbott, Pfizer, and Centocor; consulting fees from Abbott, Pfizer, Merck, Roche, and UCB; speakers bureau for Abbott, BMS, Merck, Mundipharma, Pfizer, and UCB. **Acknowledgements:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to these studies. The authors acknowledge Carmen Fleurinck, previous employee of UCB, for her work on these studies, Celia Menckeberg, PhD, UCB, Breda, The Netherlands for publication coordination, Isabel Raynaud, MBBS iBSc, and Evelyn Turner, BSc, Costello Medical, Cambridge, UK, for medical writing and editorial assistance, and the Costello Medical Creative team for graphic design assistance. These studies were funded by UCB. All costs associated with development of this presentation were funded by UCB.

To receive a copy of this poster, scan the QR code
Link expiration: March 30, 2025

Presented at NCRA 2025 | March 15–16 | Durham, NC

Previously presented at ACR Convergence 2024 | November 14–19 | Washington, D.C.